Abstract No

580

T- or NK-cell recruiting anti-EGFRvIII TandAbs are highly specific and potent therapeutic antibody candidates for the treatment of EGFRvIII⁺ tumors

Kristina Ellwanger, Uwe Reusch, Ivica Fucek, Michael Weichel, Thorsten Gantke, Stefan Knackmuss, Michael Tesar, Martin Treder

Affimed GmbH, Im Neuenheimer Feld 582, 69120 Heidelberg, Germany

Introduction

EGFRvIII is the most prevalent tumor-specific variant of the wild-type EGFR and represents an attractive target in various solid tumors such as GBM, HNSCC, NSCLC and others. Despite the high medical need and the tumor specificity of this target in these indications very little therapeutic progress has been made with EGFRvIII. Due to its exceptional tumor exclusivity it is an ideal candidate for both T- and NK-cell engagement. We therefore developed tetravalent, bispecific EGFRvIII antibodies (TandAbs) to recruit either cytotoxic T- or NK-cells, aimed at the elimination of EGFRvIII⁺ cancer cells while sparing normal tissue that ubiquitously expresses EGFR. Selected lead candidates AFM21 (EGFRvIII/CD3) and AFM22 (EGFRvIII/CD16A) exhibited exquisite specificity towards EGFRvIII in Western Blot, SPR, ELISA and flow cytometry. No binding was observed to EGFR. Both displayed potent cytotoxicity in the low picomolar range and, most importantly, did not elicit T- or NK-cell activation or activation of immune cell proliferation, thus suggesting an excellent safety profile.
In vivo efficacy for AFM21 was demonstrated in a mouse xenograft model. The clinical relevance of EGFRvIII as a tumor marker and the use of our anti-EGFRvIII domains as companion diagnostics were evaluated by IHC on GBM, HNSCC, NSCLC and others.
In summary, these *in vitro* and *in vivo* studies qualify AFM21 and AFM22 as highly attractive therapeutic antibody candidates and present themselves as promising strategies for selective immunotherapy for the treatment of EGFRvIII⁺ tumors. The highly specific tumor expression of EGFRvIII and the absence of off-target activity of our TandAbs provide for an excellent safety profile reducing the risks of side effects associated with other EGFR-driven therapies.

EGFRvIII is associated with oncogenic transformation

- Most frequent mutant variant of EGFR
- In frame deletion of exons 2-7
- Truncation of 267aa of ECD
- Novel glycine residue at the junction
- Highly tumor-specific epitope
- Signaling is ligand-independent and constitutively active
- Contributes to enhanced tumorigenicity and resistance to conventional anti-tumor therapy

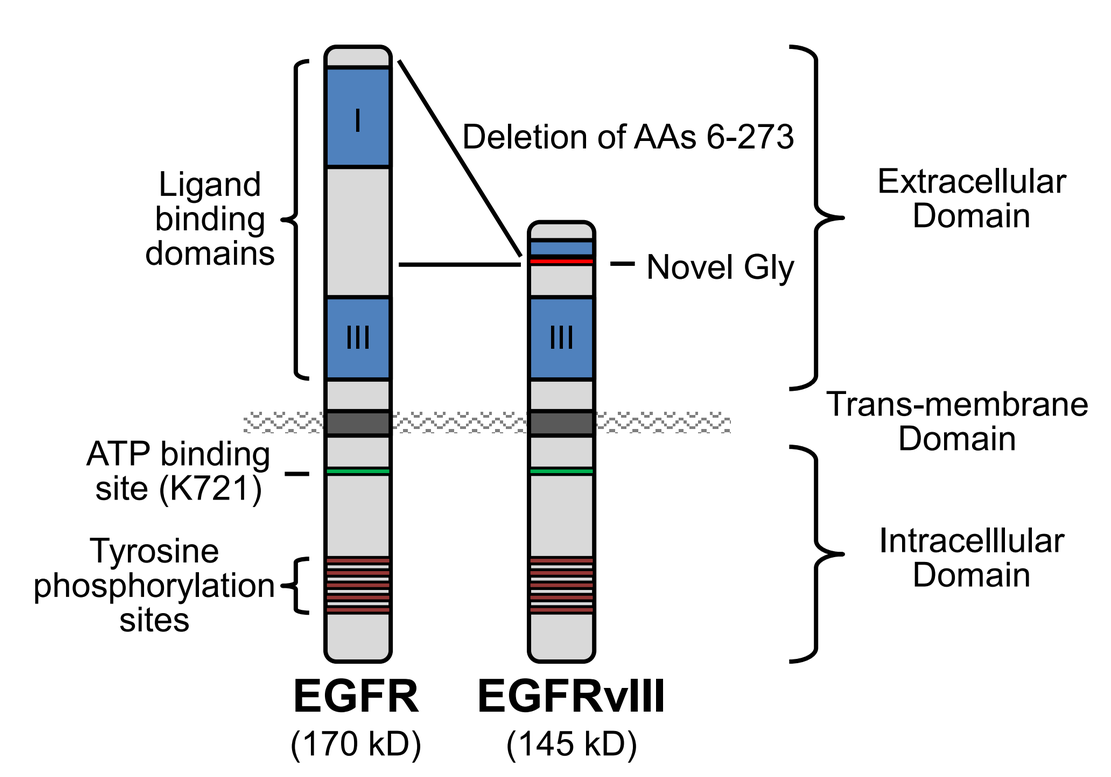

Deletion of AAs 6-273
Ligand binding domains
Novel Gly
Extracellular Domain
Trans-membrane Domain
ATP binding site (K721)
Tyrosine phosphorylation sites
Intracelllular Domain
EGFR (170 kD)
EGFRvIII (145 kD)

EGFRvIII-specific scFv were isolated, affinity matured and formatted into TandAbs

De novo screening (scFv phage display)

EGFRvIII-specific Fv sequence (Li3G30) identified, characterized and used for TandAb generation

Affinity maturation of scFv (AbAccel)

7/11 with 10- to >100-fold improved K_D, 3/11 with 4- to 6-fold improved k_{off}, all with maintained EGFRvIII-specificity

6 affinity matured Fv sequences used for TandAb construction

>40 EGFRvIII/CD3 and EGFRvIII/CD16A TandAb candidates were generated and tested

1 development candidate EGFRvIII/CD3 AFM21

1 development candidate EGFRvIII/CD16A AFM22

Binding selectivity (Biacore)

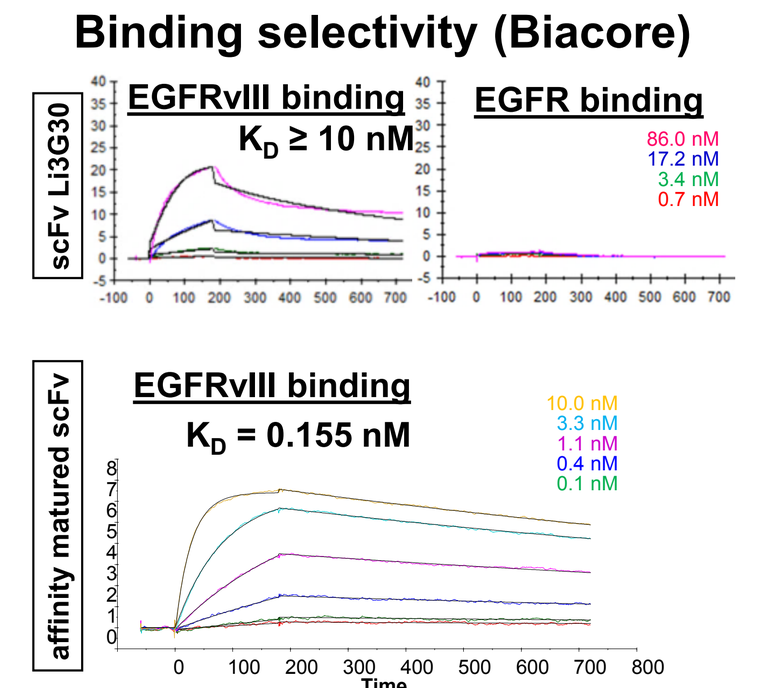

scFv Li3G30
EGFRvIII binding
$K_D \geq 10$ nM
EGFR binding
affinity matured scFv
EGFRvIII binding
$K_D = 0.155$ nM
Time

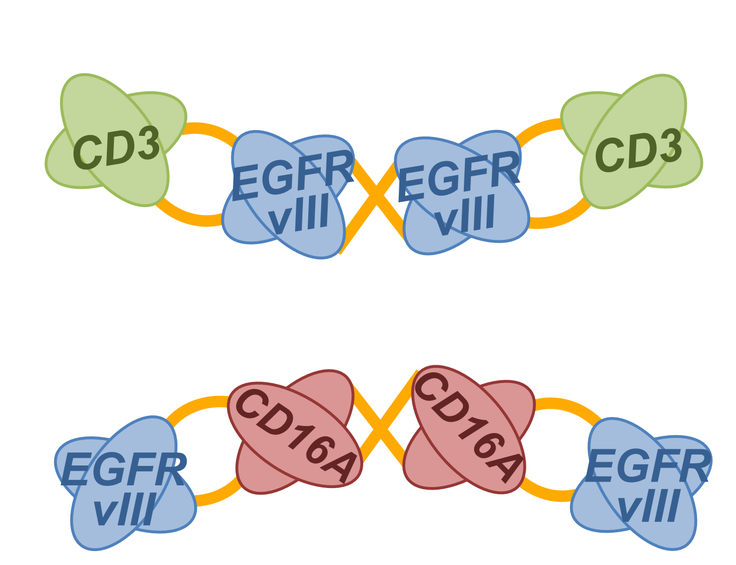

CD3
EGFR vIII
CD16A

EGFRvIII-specific T-cell and NK-cell recruiting TandAbs were characterized for EGFRvIII antigen binding

- Bivalently binding Diabodies and TandAb variants with effector domains binding to human and cynomolgus CD3 or CD16A were constructed, expressed and characterized.
- Apparent affinities to EGFRvIII in the bivalently binding TandAb format were further improved relative to the monovalently binding scFv. Reduced dissociation rates were achieved by both, affinity maturation and formatting into TandAbs or Diabodies.

EGFRvIII binding domain		EGFRvIII binding (SPR)							
		scFv		T-cell TandAb			NK-cell TandAb		
		K_D (nM)	improve-ment	K_D (nM)	k_{on} (1/Ms)	k_{off} (1/s)	K_D (nM)	k_{on} (1/Ms)	k_{off} (1/s)
parental	Li3G30	17.5	1	0.567	8.70E+5	4.93E-4	n.a.	n.a.	n.a.
Affinity matured	A3	0.25	70	0.006	3.43E+6	2.08E-5	0.035	4.08E+6	1.43E-4
	A4	0.16	113	0.011	8.22E+6	8.79E-5	0.016	1.09E+7	1.76E-4
	AFM21/AFM22	0.85	21	0.043	2.31E+6	9.81E-5	0.080	2.39E+6	1.92E-4

Highly specific binding

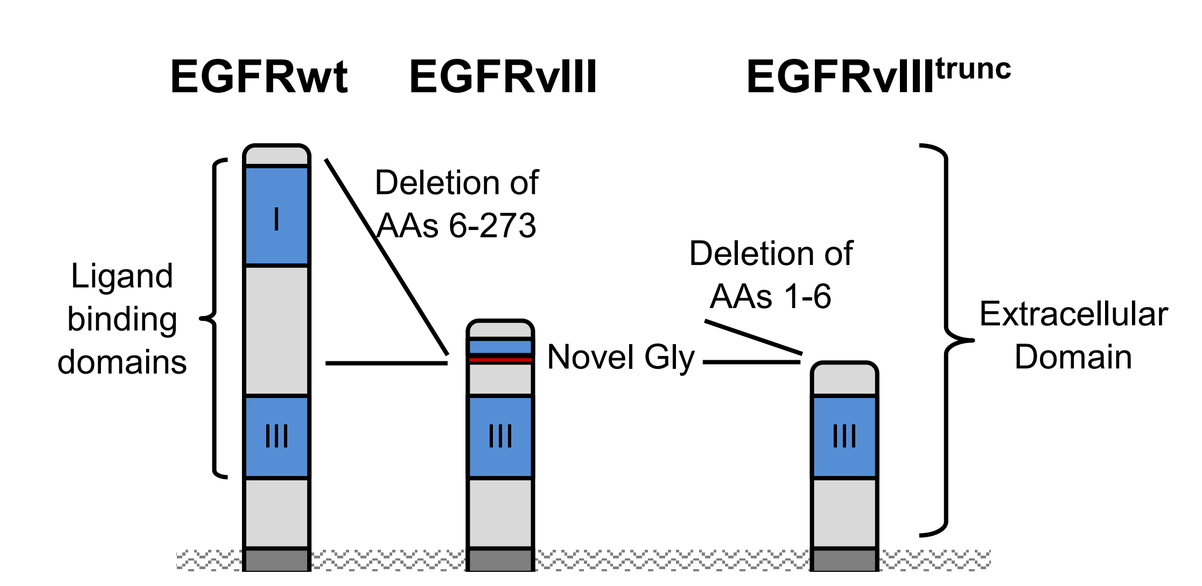

EGFRwt
EGFRvIII
EGFRvIII^{trunc}
Deletion of AAs 6-273
Deletion of AAs 1-6
Ligand binding domains
Novel Gly
Extracellular Domain

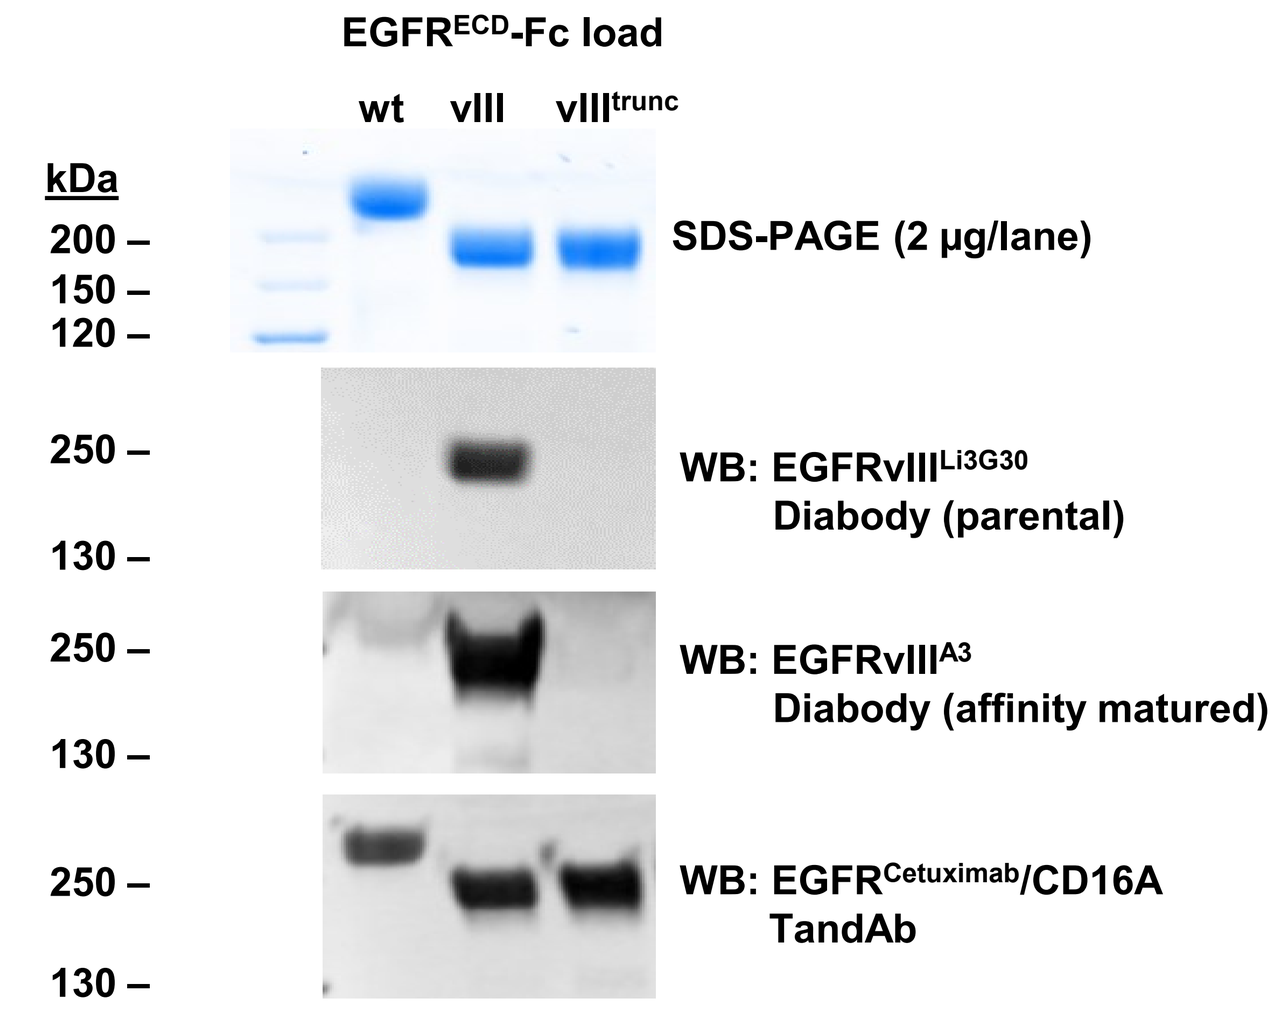

EGFR^{ECD}-Fc load
wt vIII vIII^{trunc}
kDa
SDS-PAGE (2 µg/lane)
WB: EGFRvIII^{Li3G30} Diabody (parental)
WB: EGFRvIII^{A3} Diabody (affinity matured)
WB: EGFR^{Cetuximab}/CD16A TandAb

- Truncation of 6 N-terminal amino acids in the extracellular domain of EGFRvIII abolishes binding of our EGFRvIII-specific antibodies.
- Selective binding to the unique N-terminal EGFRvIII epitope was maintained during affinity maturation and confirmed by SDS-PAGE, Western Blot and ELISA with recombinant antigen variants; no binding to EGFR was observed with our antibodies.
- Peptide mapping analysis incl. different amino acid substituents demonstrated binding to a N-terminal epitope containing the EGFRvIII-specific novel glycin for our EGFRvIII specific diabody, whereas reference antibody L8A4 showed a different pattern (data not shown).

EGFRvIII target expression as determined by immunohistochemistry

Immunohistochemistry with EGFRvIII^{Li3G30} Diabody (0.5 µg/ml) on cryo sections

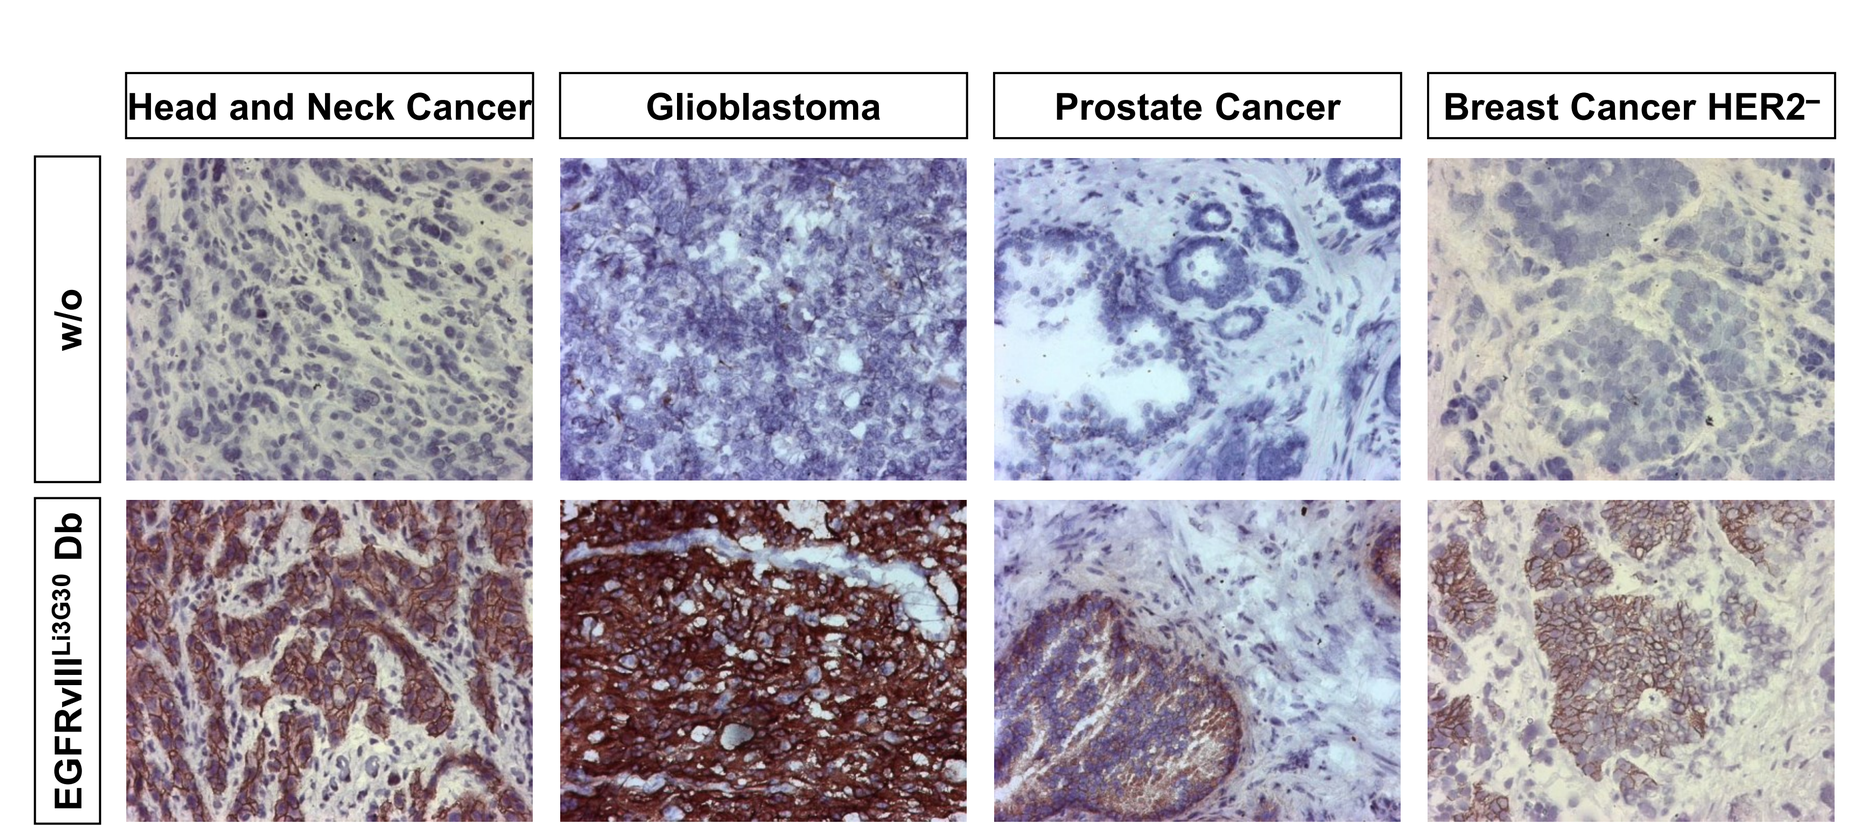

Head and Neck Cancer
Glioblastoma
Prostate Cancer
Breast Cancer HER2⁻
w/o
EGFRvIII^{Li3G30} Db

- Demonstrated EGFRvIII expression in several solid tumors
- No expression in healthy tissues (data not shown)

EGFRvIII-targeting TandAbs are well expressed and stable

- EGFRvIII-targeting TandAbs are well expressed with research grade expression titers
- Melting point Analysis by DSF:

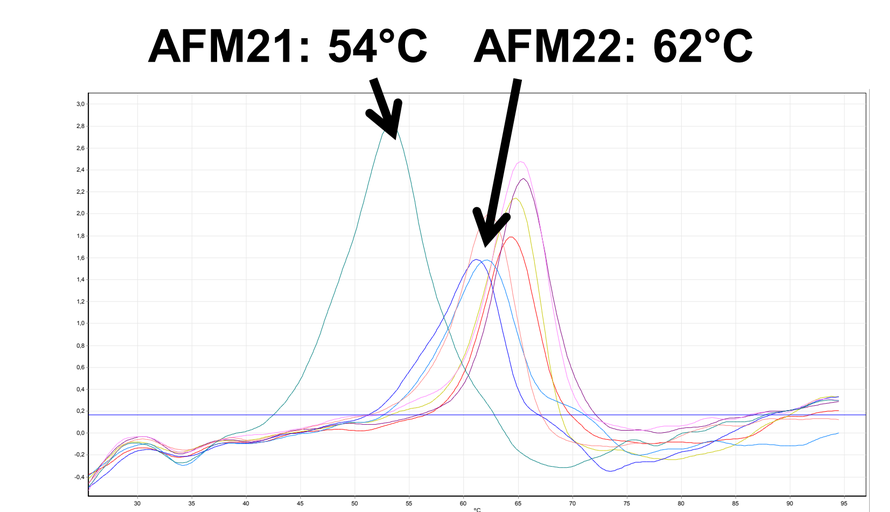

AFM21: 54°C
AFM22: 62°C

Research grade expression of AFM21 and AFM22 TandAb candidates

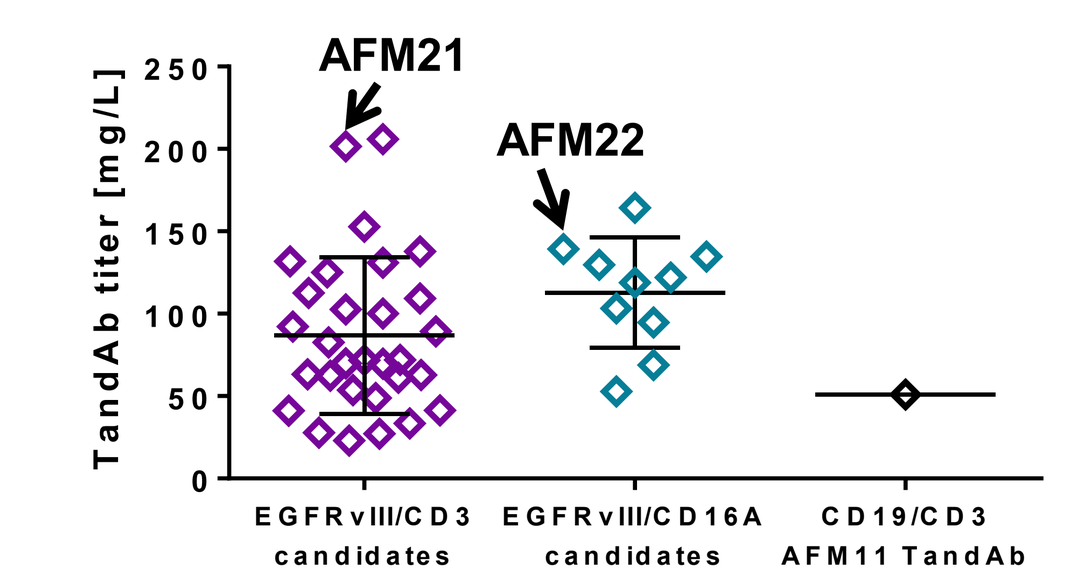

AFM21
AFM22
TandAb titer [mg/L]
EGFRvIII/CD3 candidates
EGFRvIII/CD16A candidates
CD19/CD3 AFM11 TandAb

- EGFRvIII-targeting TandAbs show good thermostability and serum stability

Thermostability analysis by SE-HPLC

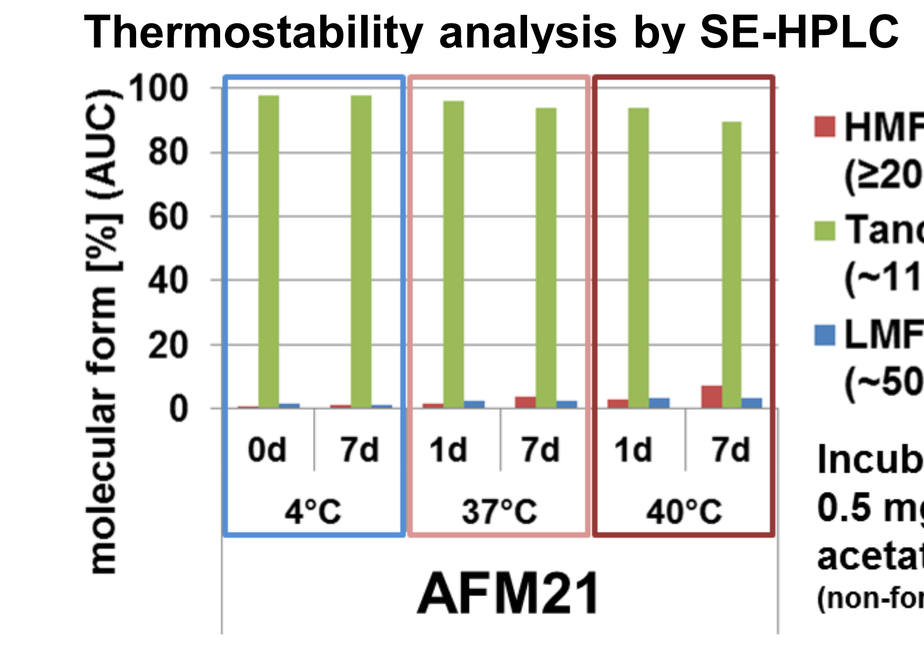

molecular form [%] (AUC)
0d 7d 1d 7d 1d 7d
4°C 37°C 40°C
AFM21
HMF (≥200 kDa)
TandAb (~110 kDa)
LMF (~50 kDa)
Incubation at 0.5 mg/ml in acetate buffer (non-formulated)

Stability of AFM22 after incubation in human serum (antigen binding activity in ELISA)

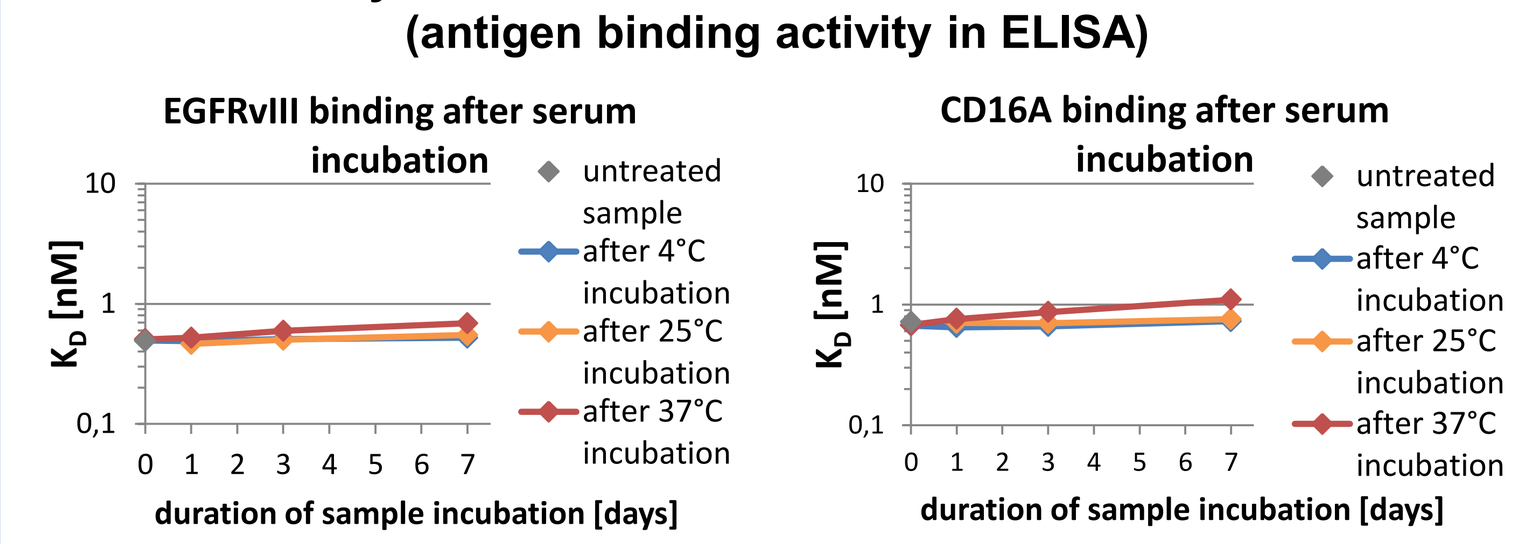

EGFRvIII binding after serum incubation
CD16A binding after serum incubation
K_D [nM]
untreated sample
after 4°C incubation
after 25°C incubation
after 37°C incubation
duration of sample incubation [days]

Freeze-thaw stability of AFM21

(3x -80°C/25°C)

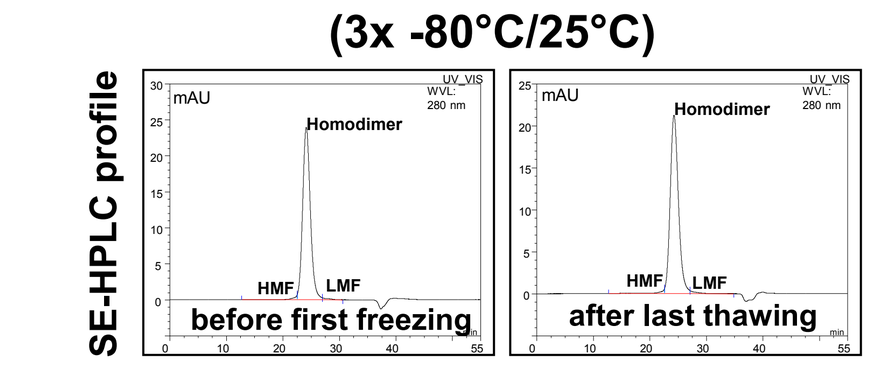

SE-HPLC profile
before first freezing
after last thawing

AFM21 and AFM22 bind with high affinity and specificity to target and effector cells

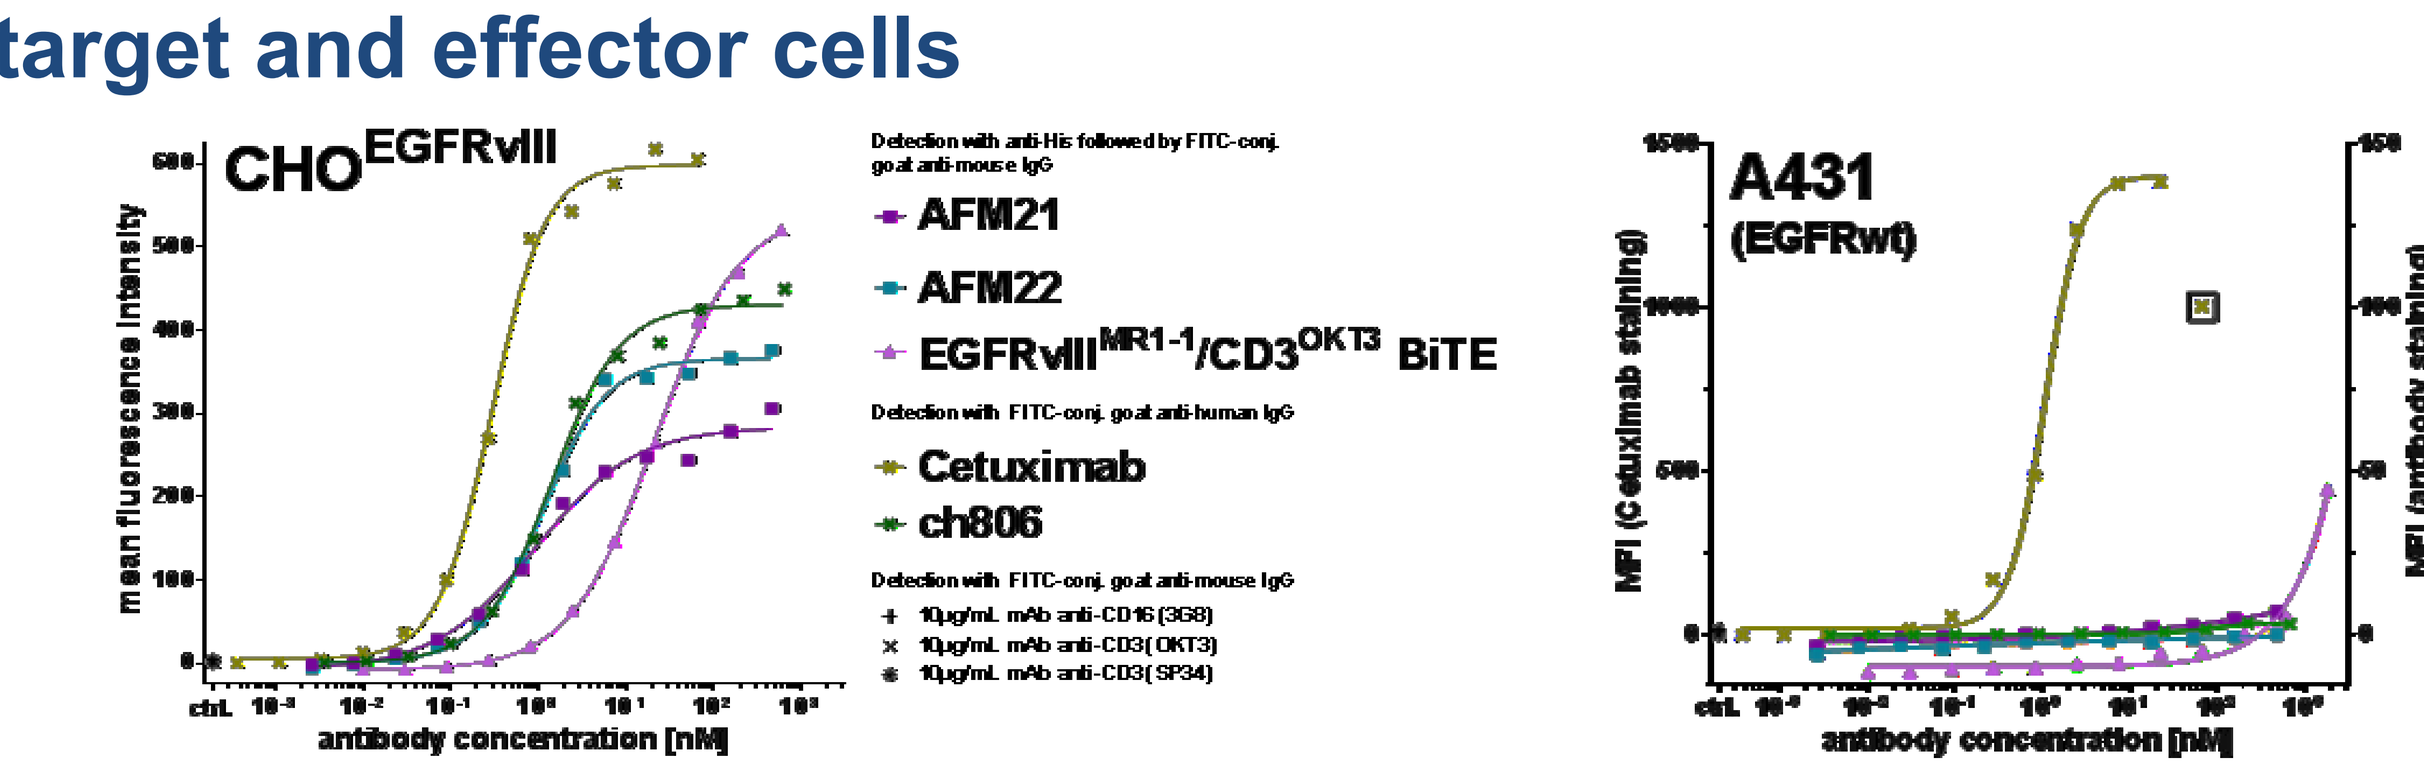

CHO^{EGFRvIII}
A431 (EGFRwt)
AFM21
AFM22
EGFRvIII^{MR1-1}/CD3^{OKT3} BiTE
Cetuximab
ch806
antibody concentration [nM]

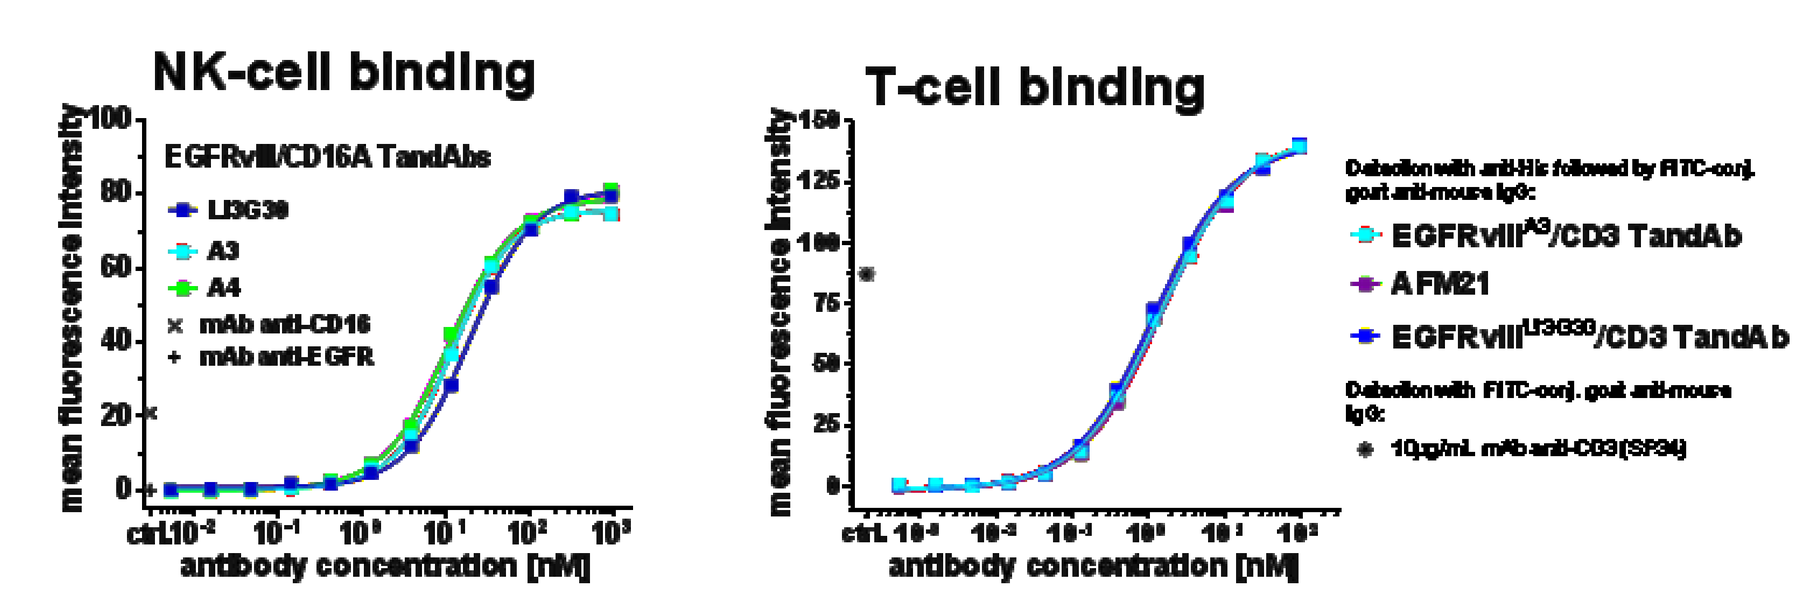

NK-cell binding
T-cell binding
EGFRvIII/CD16A TandAbs
Li3G30
A3
A4
AFM21
antibody concentration [nM]

AFM21 or AFM22 showed no off-target activity

PBMC Proliferation assay (BrdU)

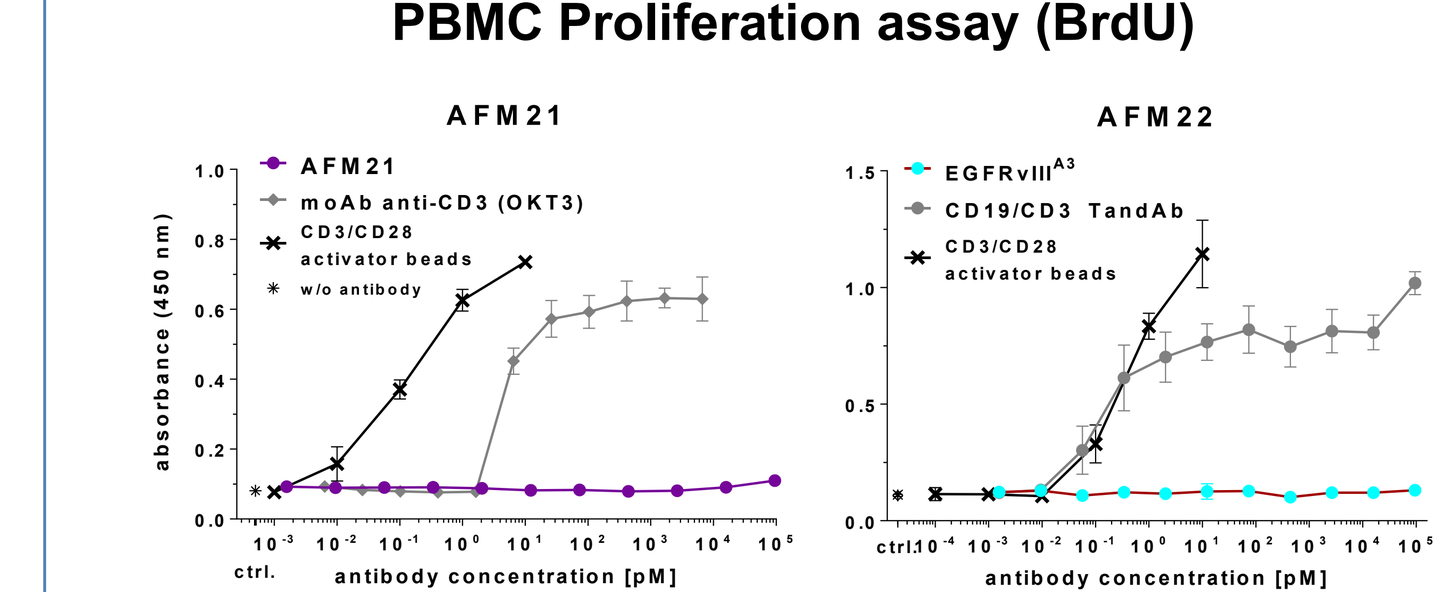

AFM21
AFM22
moAb anti-CD3 (OKT3)
CD3/CD28 activator beads
w/o antibody
EGFRvIII^{A3}
CD19/CD3 TandAb
absorbance (450 nm)
antibody concentration [pM]

Primary human PBMC (4x10⁵/well) were incubated with EGFRvIII/CD3 or EGFRvIII/CD16A TandAb candidates or control antibodies for 5 days. Proliferation was analysed by BrdU incorporation assay.

Primary PBMC cells were cultured for 24 h +/- AFM21 or AFM22 +/- DK-MG target cells at an E:T ratio of 25:1 before PBMC cells were analyzed for expression of activation markers by flow cytometry. As a positive control cells were cultured in the presence of CD3/CD28 activator beads.

Target-dependent induction of activation markers

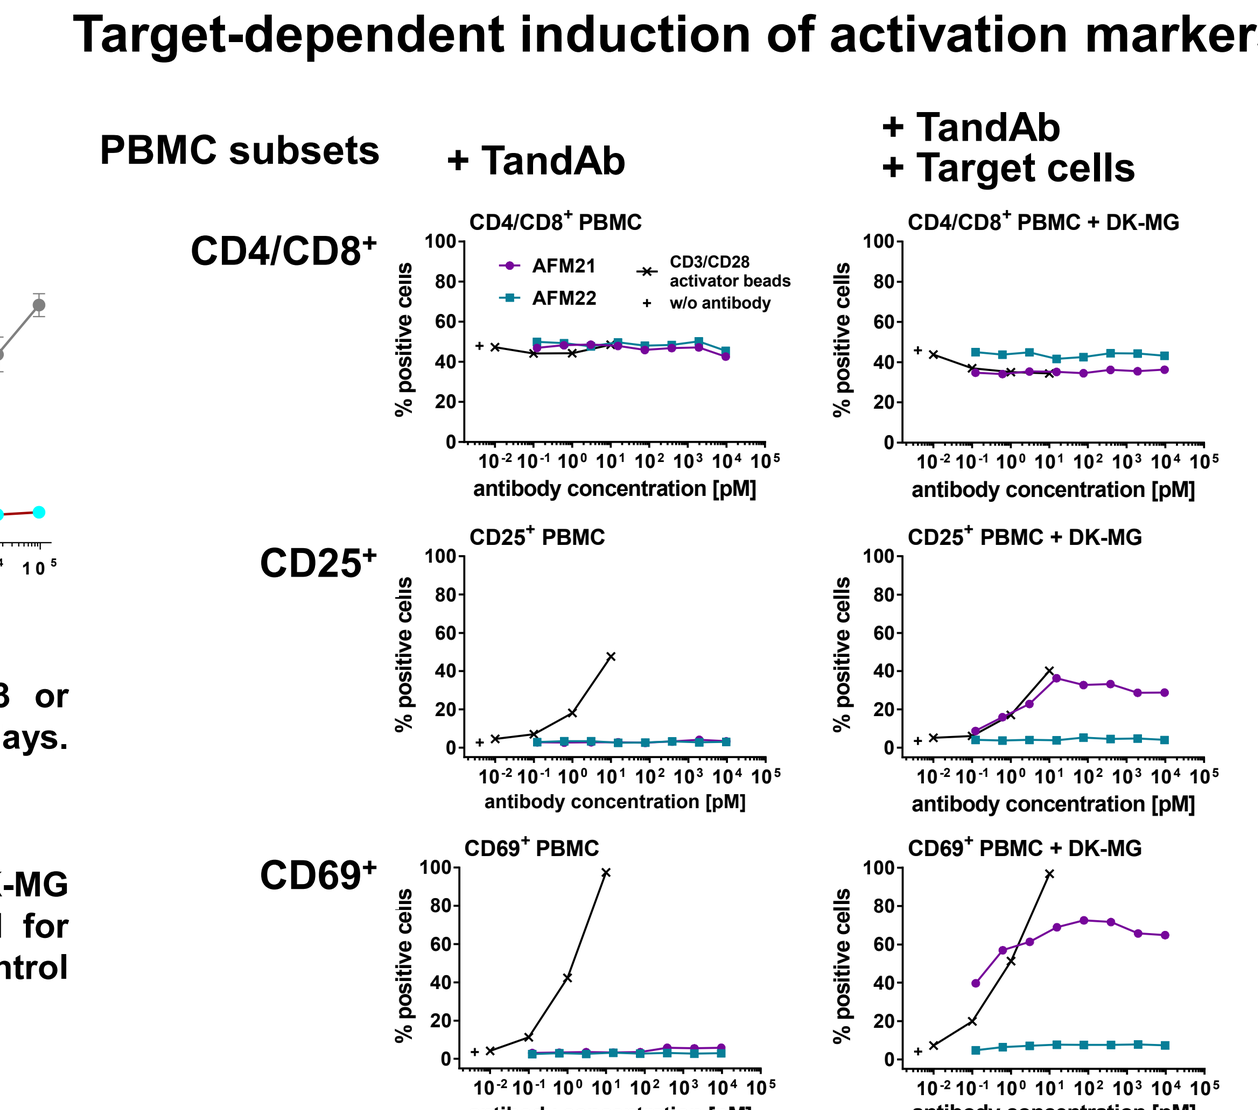

PBMC subsets
+ TandAb
+ TandAb + Target cells
CD4/CD8⁺
CD25⁺
CD69⁺
% positive cells
antibody concentration [pM]

- No induction of T cell or PBMC proliferation or activation markers by AFM21 or AFM22 TandAbs in the absence of target cells

AFM21 and AFM22 are highly potent and specific killers of EGFRvIII⁺ target cells

Cytotoxicity of EGFRvIII/CD3 TandAbs (T-cells as effector cells)

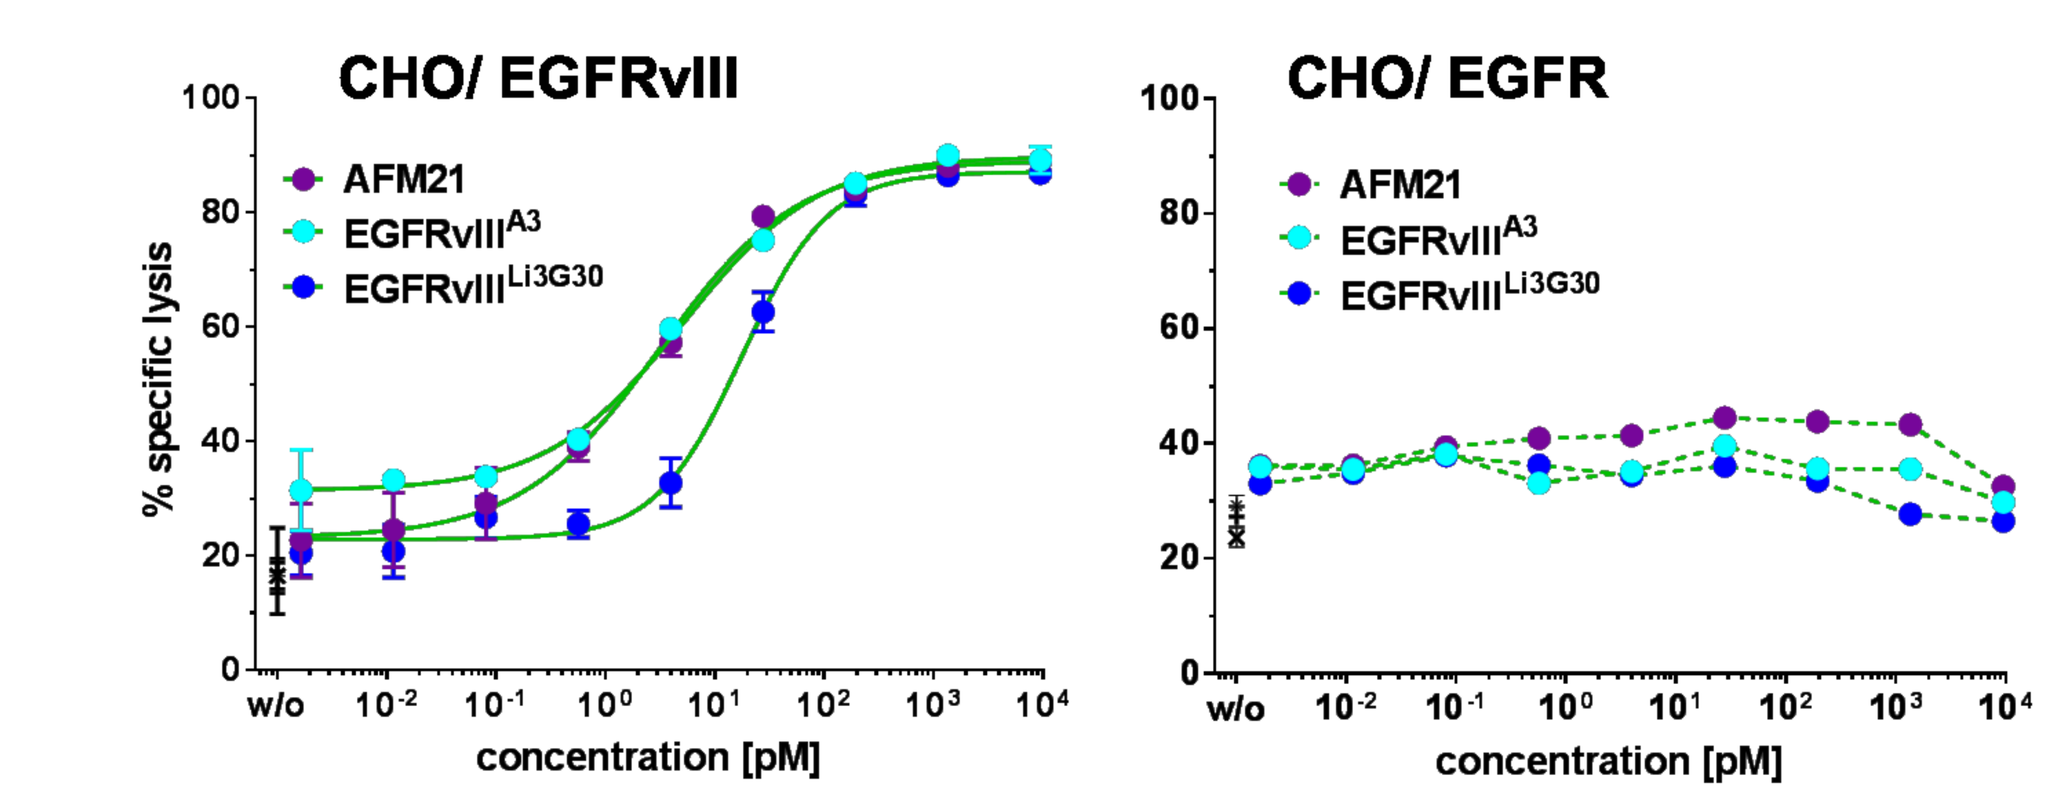

CHO/ EGFRvIII
CHO/ EGFR
AFM21
EGFRvIII^{A3}
EGFRvIII^{Li3G30}
% specific lysis
concentration [pM]

Cytotoxicity on DK-MG (PBMC as effector cells)

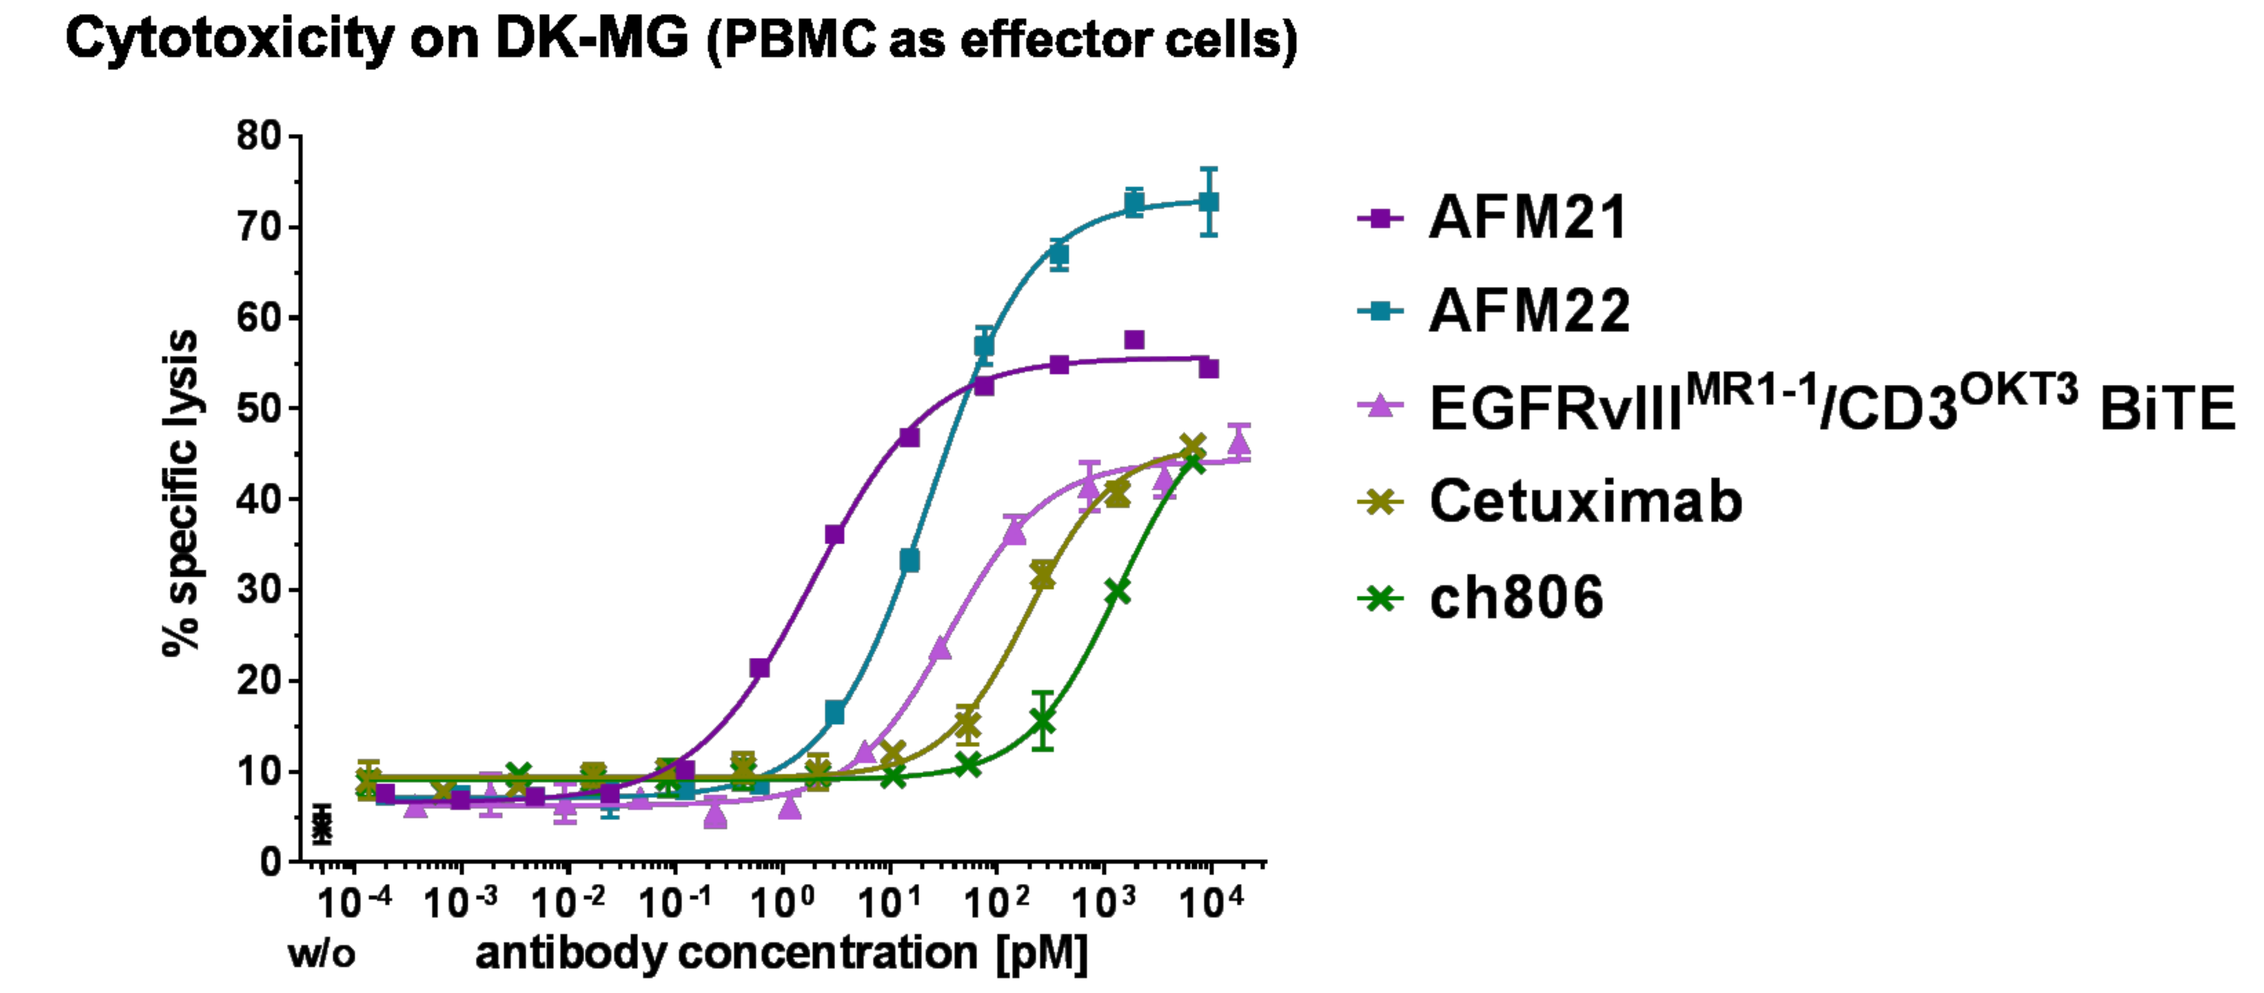

AFM21
AFM22
EGFRvIII^{MR1-1}/CD3^{OKT3} BiTE
Cetuximab
ch806
% specific lysis
antibody concentration [pM]

EGFRvIII binding domain		effector domain	Cytotoxic potency (EC_{50}); mean of n≥3
Low affinity	Li3G30	CD3	34.8 pM
		CD16A	>250 pM
Affinity matured	AFM21	CD3	4.4 pM
	AFM22	CD16A	6.8 pM

AFM21: Inhibition of tumor growth *in vivo*

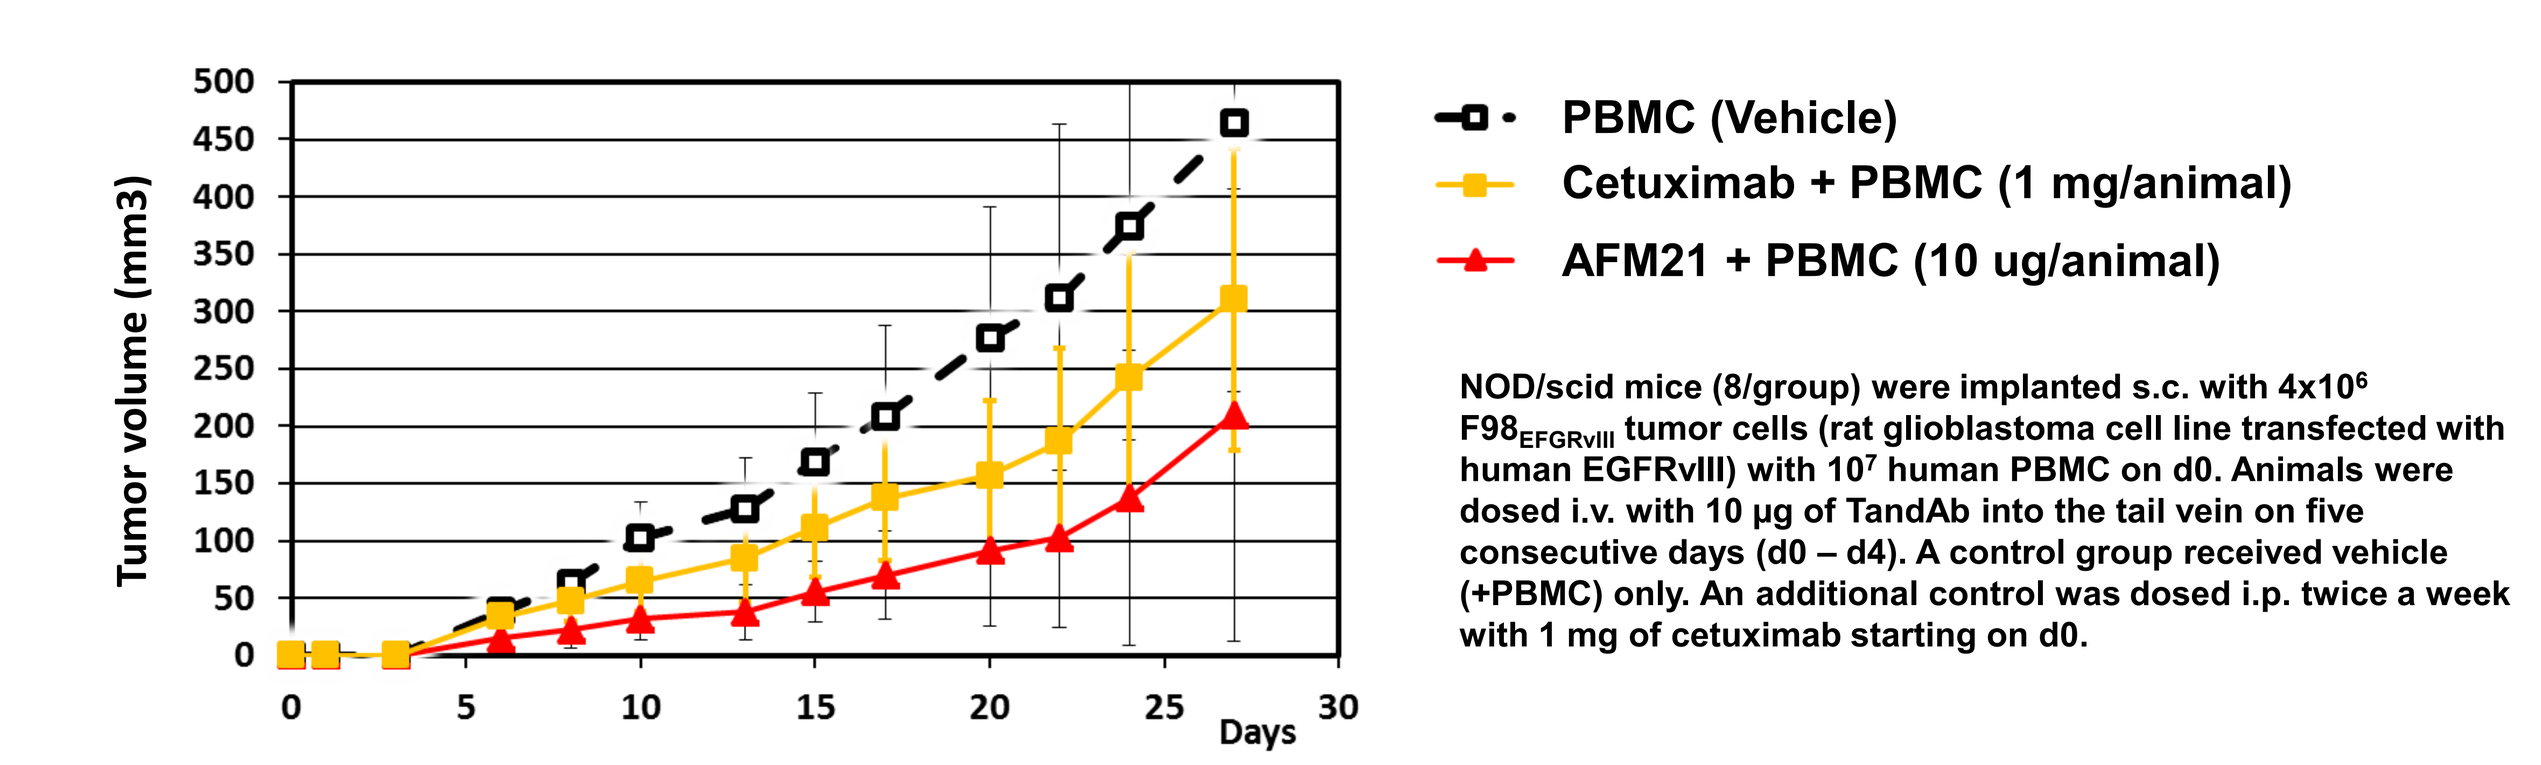

PBMC (Vehicle)
Cetuximab + PBMC (1 mg/animal)
AFM21 + PBMC (10 ug/animal)
Tumor volume (mm3)
Days

NOD/scid mice (8/group) were implanted s.c. with 4x10⁶ F98$_{EGFRvIII}$ tumor cells (rat glioblastoma cell line transfected with human EGFRvIII) with 10⁷ human PBMC on d0. Animals were dosed i.v. with 10 µg of TandAb into the tail vein on five consecutive days (d0 – d4). A control group received vehicle (+PBMC) only. An additional control was dosed i.p. twice a week with 1 mg of cetuximab starting on d0.

- AFM21 demonstrates anti-tumor efficacy and shows greater tumor growth inhibition than Cetuximab

Summary/Conclusion

- Fully characterized EGFRvIII-specific human scFv were formatted into TandAbs and AFM21 & AFM22 were selected as drug candidates
- AFM21 & AFM22 show excellent producibility, purity and stability
- Both drug candidates bind to the highly tumor-specific EGFRvIII epitope and this specificity was shown to be unique to our anti-EGFRvIII domains
- AFM21 & AFM22 showed superior cytotoxicity towards various EGFRvIII-expressing cells to other competing antibodies, with EC_{50} values in the range of 1 to 10 pM, and no off-target activity
- AFM21 mediates dose-dependent inhibition of EGFRvIII⁺ tumor growth in a mouse xenograft model
- In conclusion: AFM21 and AFM22 are excellent and superior therapeutic agents when compared to other therapeutic competitors such as ABT-806 or a BiTE-based approach against EGFRvIII-expressing tumors

©2016 Affimed GmbH, Im Neuenheimer Feld 582, 69120 Heidelberg, Germany; contact: info@affimed.com; T: +49 6221 65307 0