Immune checkpoint inhibition by anti-PD-1 or CD137 co-stimulation enhances cytotoxicity towards CD30+ tumors mediated by the bispecific tetravalent CD30/CD16A TandAb AFM13

Abstract No 2323

Xing Zhao[1], Narendiran Rajasekaran[2], Uwe Reusch[3], Jens-Peter Marschner[3], Martin Treder[3#], Holbrook Edwin Kohrt[2]

[1]Tissue Engineering and Stem Cells Research Center, Department of Immunology, Guiyang Medical University, Guiyang, China; [2]Center for Clinical Sciences Research Stanford, Stanford, CA 94305-5151; [3]Affimed GmbH, Im Neuenheimer Feld 582, 69120 Heidelberg, Germany

Abstract

AFM13 is an NK-cell-engaging CD30/CD16A bispecific tetravalent TandAb antibody currently in Phase 2 clinical development in Hodgkin lymphoma (HL) and CD30+ malignancies. Immune checkpoints inhibitors have demonstrated clinical efficacy in a variety of cancers, including HL. NK-cells are regulated by a number of check-points, prompting us to investigate the combination of AFM13 with several immuno-modulatory antibodies. In previous experiments we were able to demonstrate higher efficacy of AFM13 than several immuno-modulatory antibodies in monotherapy and strong synergy between AFM13 and an anti-PD-1 antibody *in vitro*, as well as *in vivo* in PDX models with human CD30+ HL tumors. In order to investigate the underlying immunological mechanisms we employed the same PDX model by implanting tumor fragments derived from surgical specimens of HL patients in immunodeficient mice. After establishing tumors, mice were reconstituted with autologous patient-derived PBMC and treated with AFM13 alone and in combination with anti-PD-1 weekly for a total of three weeks. Tumor size, tumor-infiltrating human lymphocytes, myeloid cells and intratumoral cytokines were evaluated on days 30, 44, and 58, i.e. 2, 16 and 30 days after treatment start. While monotherapy with AFM13 was reproducibly more potent than anti-PD-1, significant synergy was observed when both agents were combined. Analysis of the tumors on day 58 revealed a strong correlation between tumor growth inhibition and levels of tumor-infiltrating NK-cells, T-cells, myeloid cells and intratumoral cytokines such as IFN-γ. In contrast to anti-PD-1 monotherapy, which only induced T-cell infiltration, AFM13 monotherapy was able to induce infiltration of NK- and T-cells in the tumors, with the combination further enhancing infiltration of both. AFM13 induced stronger infiltration of macrophages than anti-PD-1, which was also increased by the combination of both agents, thereby further supporting crosstalk between innate and adaptive immunity. Furthermore, tumor analyses at earlier time-points (days 30 and 44) showed that the initial immune response is characterized by NK-cell infiltration and activation, as well as infiltration of macrophages, whilst the adaptive immune response by T-cells and activated dendritic cells was more pronounced on day 58. Combining AFM13 and anti-PD-1 augments infiltration and activation of all immune cell subpopulations.

In conclusion, our data show strong synergistic antitumor efficacy when AFM13 is combined with anti-PD-1 checkpoint blockade in HL PDX models, mediated by tumor-infiltrating lymphocytes, macrophages and dendritic cells, and provide strong evidence for crosstalk between innate and adaptive immunity induced by AFM13-recruited human NK-cells.

AFM13

Mechanism of action of the CD30/CD16A TandAb

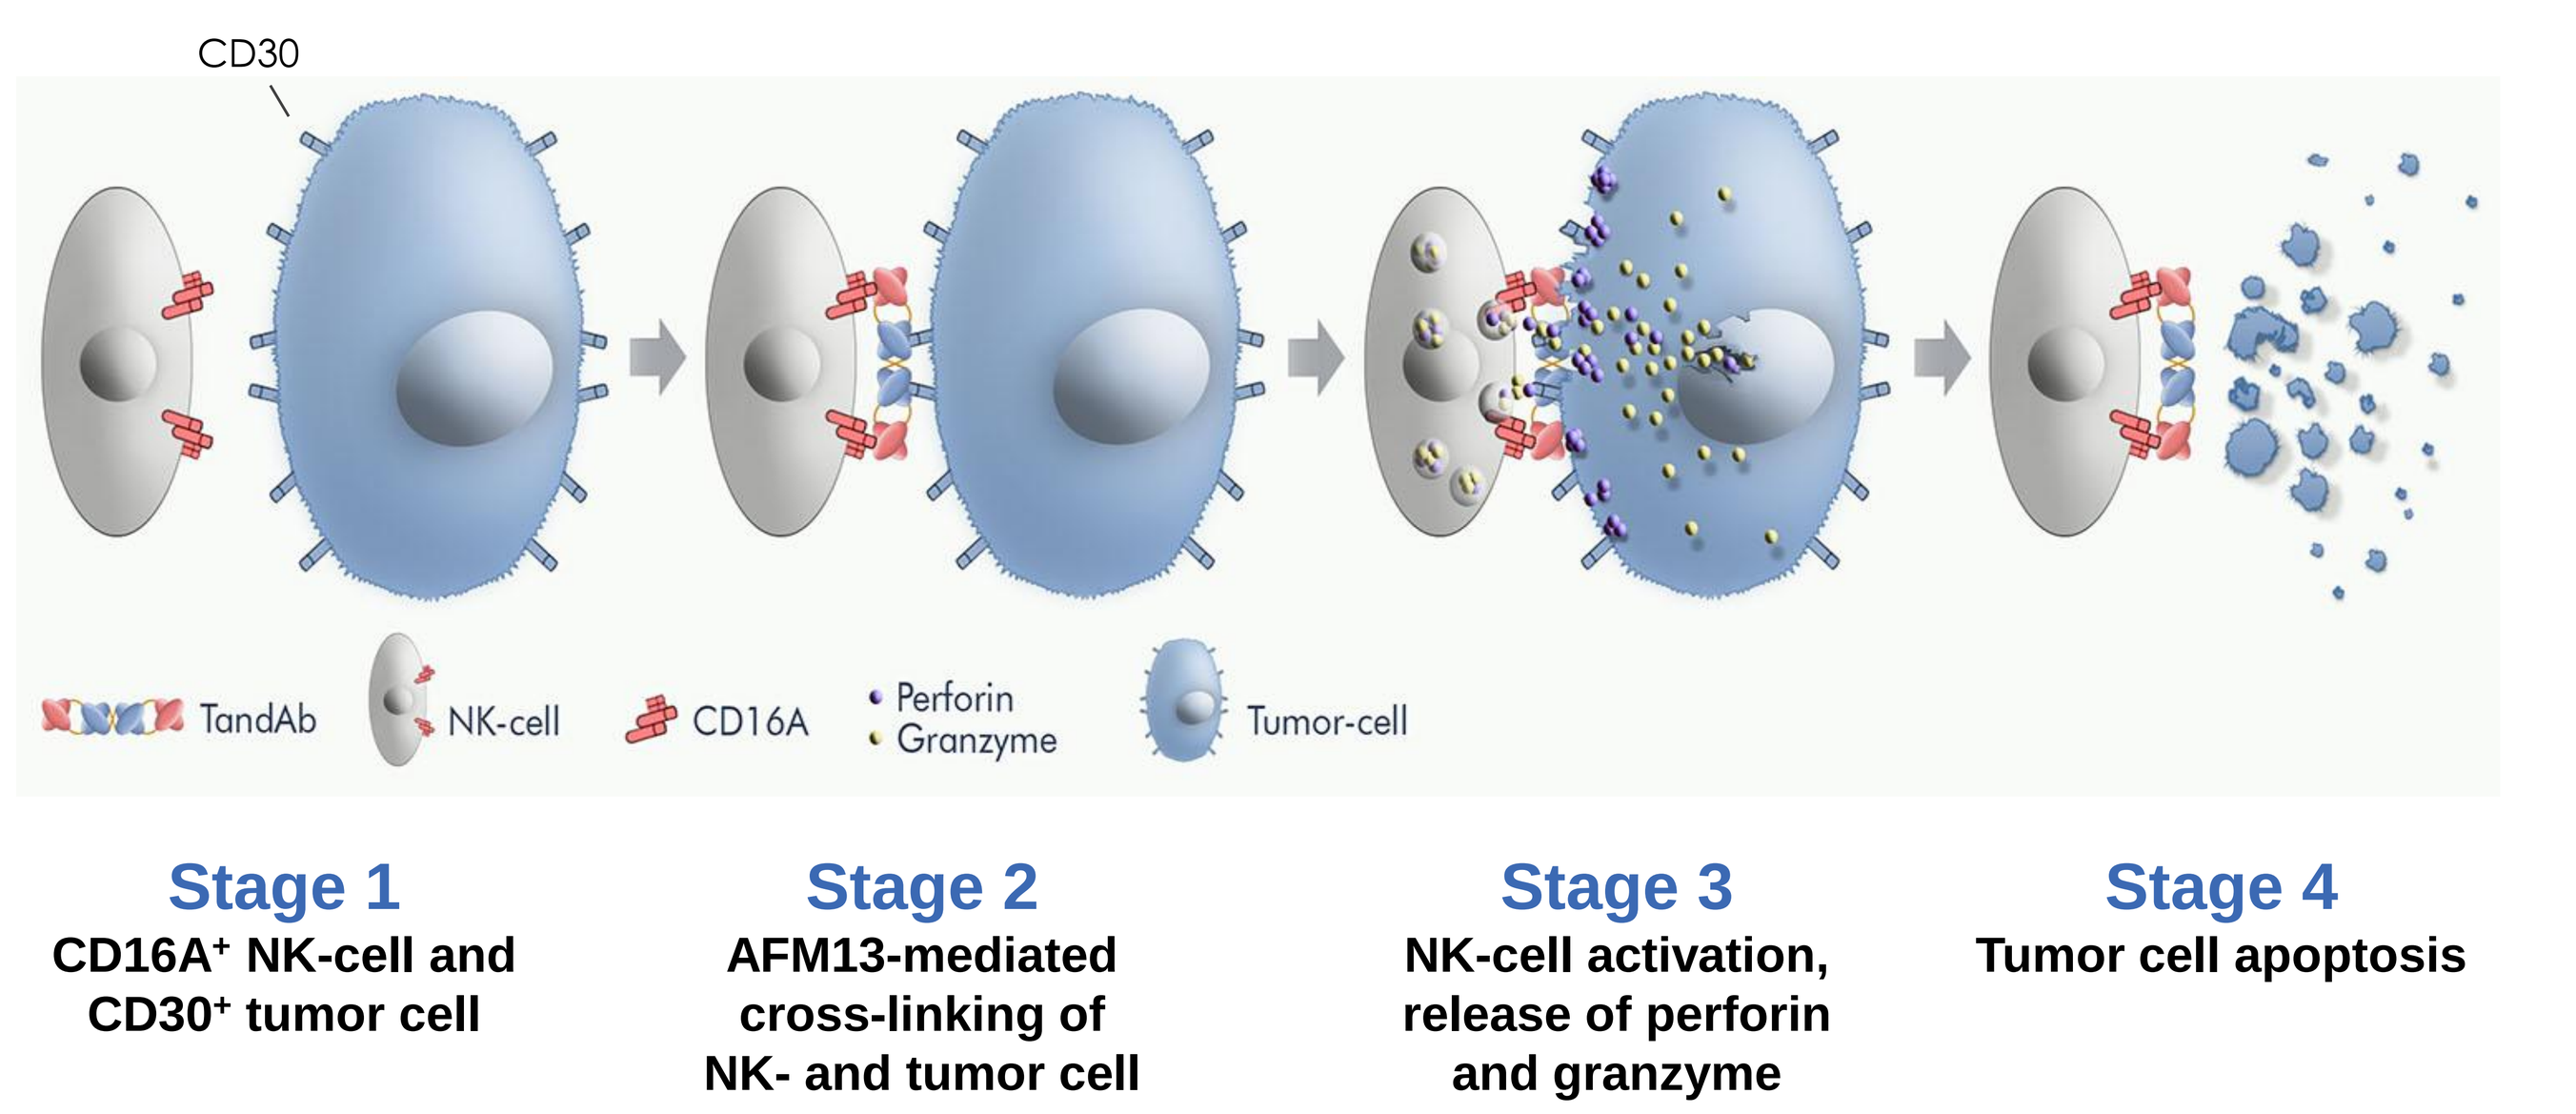
CD30
TandAb
NK-cell
CD16A
Perforin
Granzyme
Tumor-cell
Stage 1
CD16A+ NK-cell and CD30+ tumor cell
Stage 2
AFM13-mediated cross-linking of NK- and tumor cell
Stage 3
NK-cell activation, release of perforin and granzyme
Stage 4
Tumor cell apoptosis

Combination of AFM13 with checkpoint blockade or co-stimulation *in vivo*

In vivo **activity of AFM13 in combinations with CPIs in a Hodgkin lymphoma PDX model**

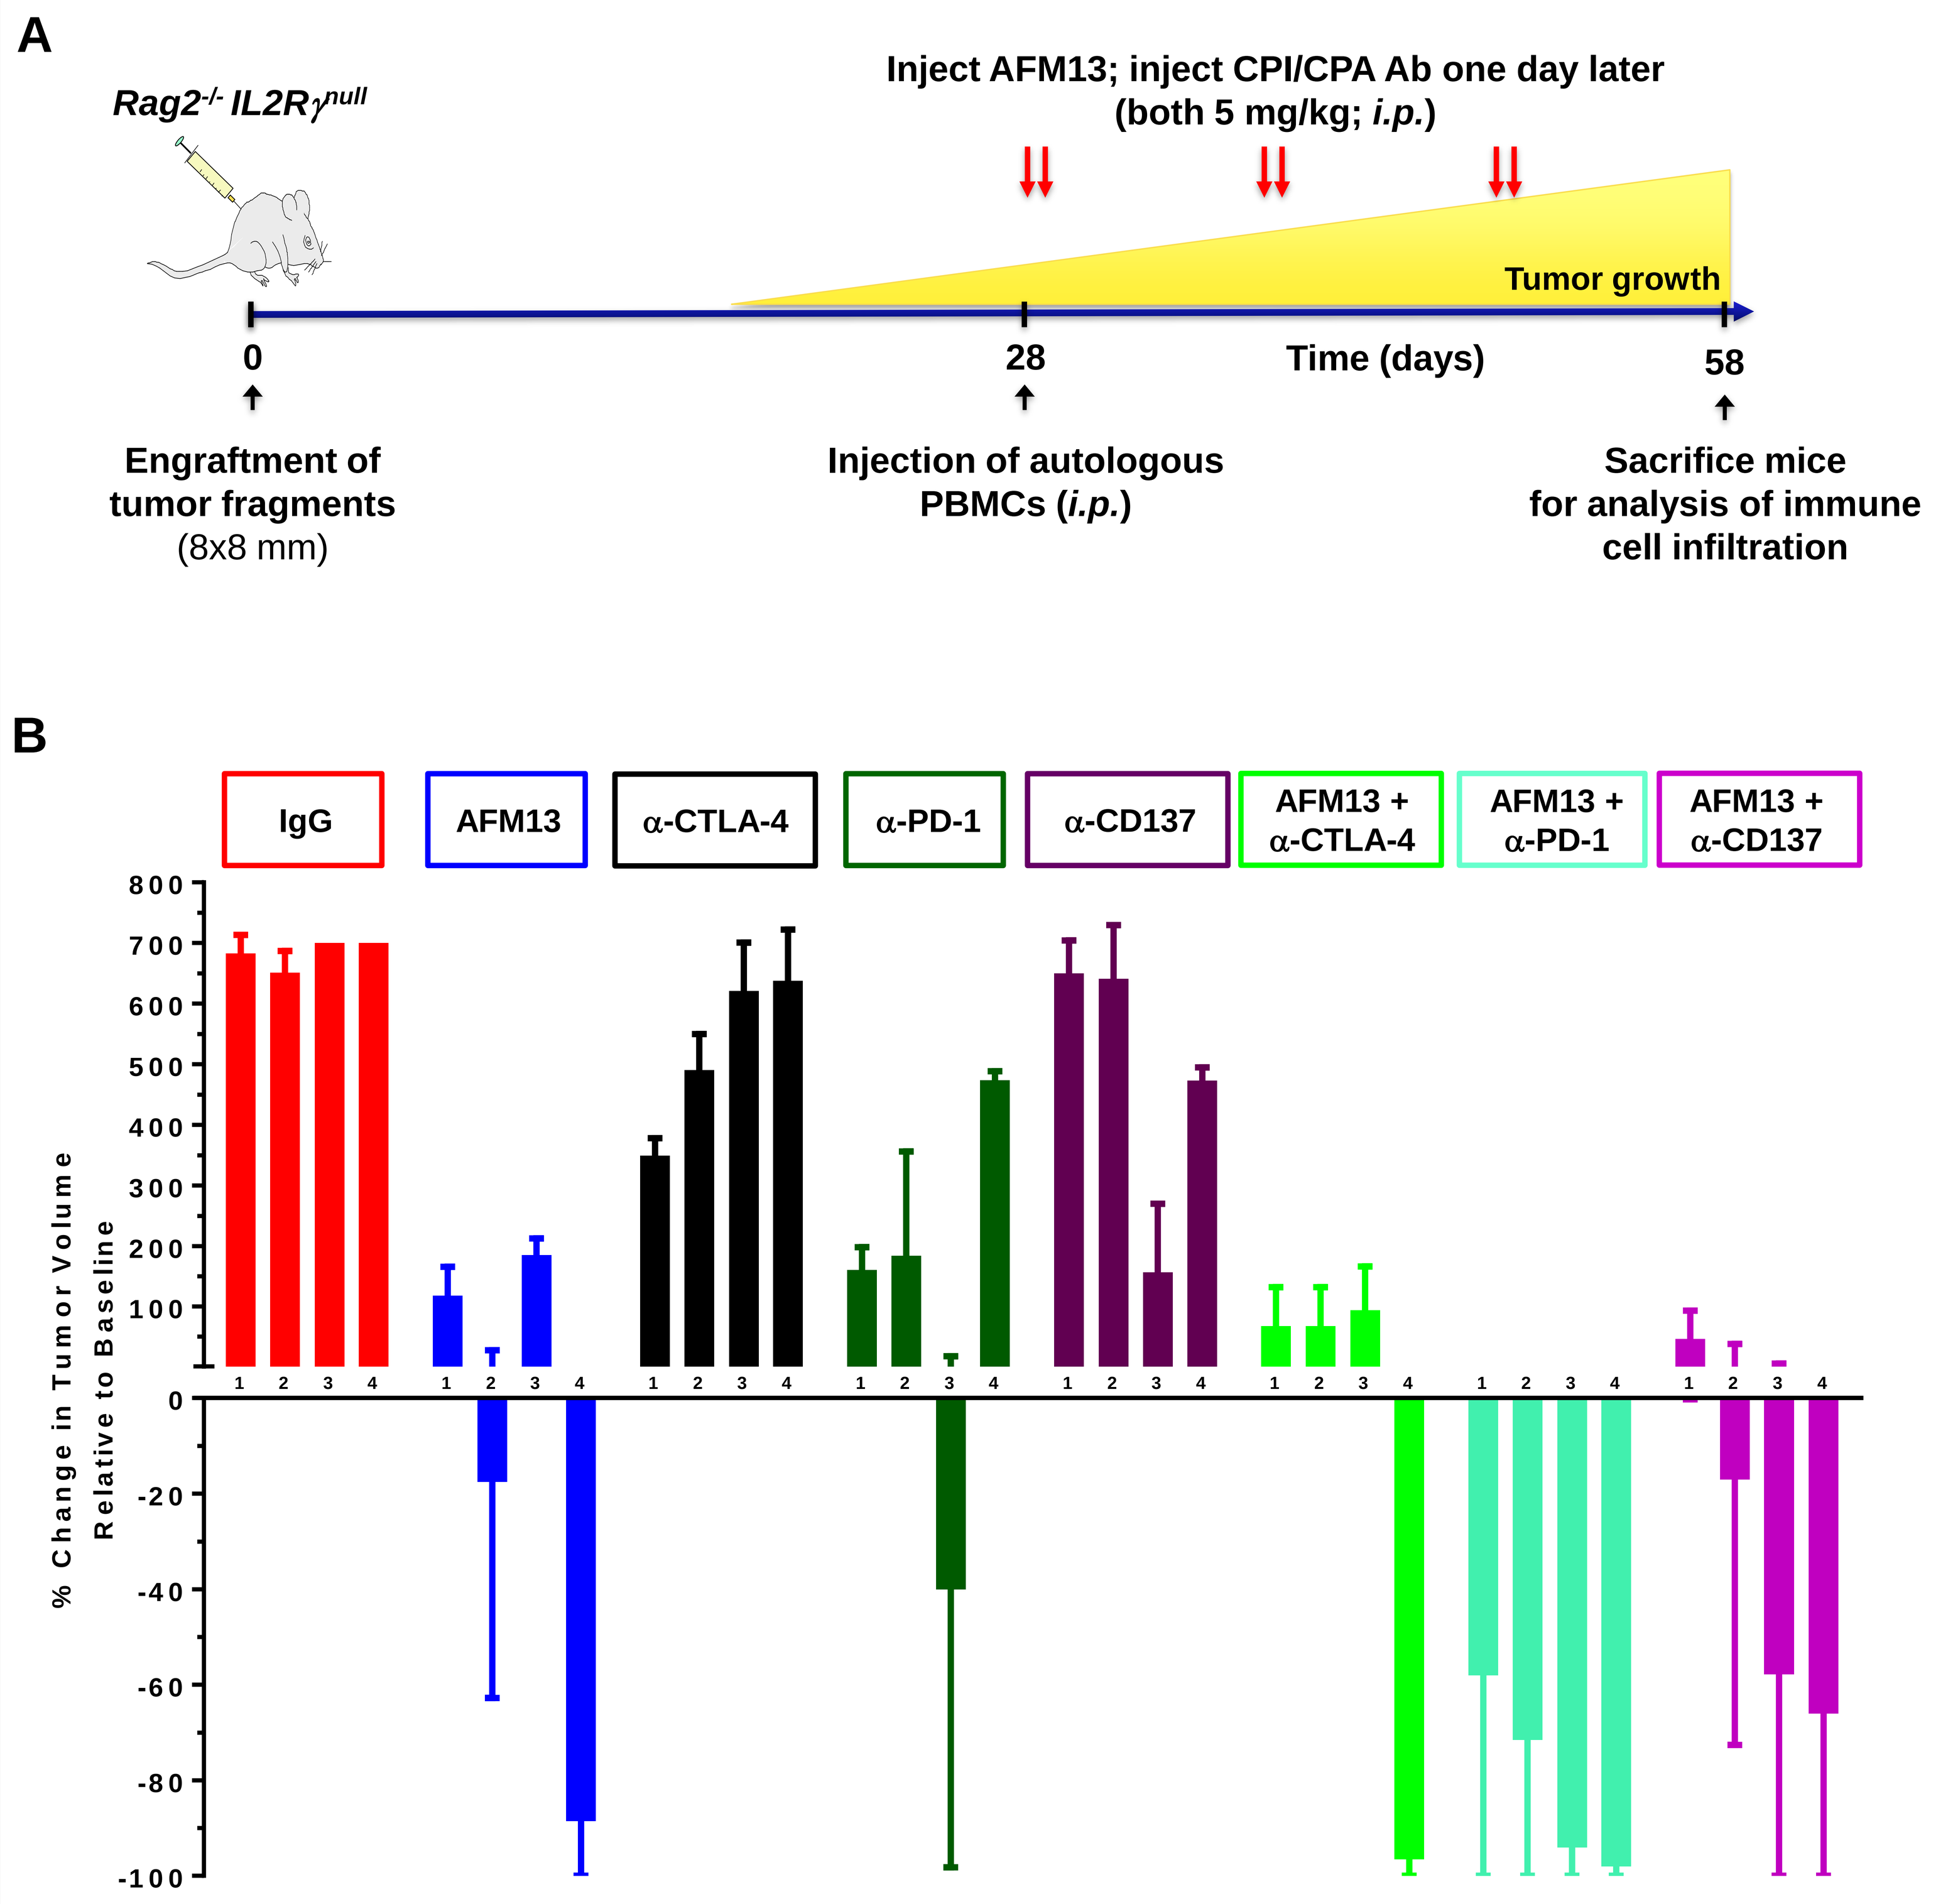
A
Rag2-/- IL2Rγnull
Inject AFM13; inject CPI/CPA Ab one day later
(both 5 mg/kg; i.p.)
Tumor growth
0
28
Time (days)
58
Engraftment of tumor fragments (8x8 mm)
Injection of autologous PBMCs (i.p.)
Sacrifice mice for analysis of immune cell infiltration
B
IgG
AFM13
α-CTLA-4
α-PD-1
α-CD137
AFM13 + α-CTLA-4
AFM13 + α-PD-1
AFM13 + α-CD137
% Change in Tumor Volume Relative to Baseline

Impact of combined checkpoint inhibition or CD137 co-stimulation on AFM13-induced reduction of tumor growth in a patient-derived xenograft (PDX) model of Hodgkin lymphoma (HL). A) HL tumor fragments were grafted onto Rag2-/-- IL2Rγnull mice (n~100) and observed for engraftment. 80 mice with engraftment of similar size (0.5 cm²) were randomized into 8 groups on day 28, followed by i.p. infusion of autologous PBMCs (2x10⁶ PBMCs/mouse). Antibody treatment was initiated on days 28 (1st Ab) and 29 (2nd Ab) and repeated weekly for a total of three combined injections. Tumor size was monitored for 58 days. B) Change of tumor volumes on day 58 relative to original tumor in four independent PDX studies.

Key results

- **Combining AFM13 and anti-PD-1 strongly enhanced NK-cell-mediated CD30+ target cell lysis *in vitro***
- **AFM13-induced tumor lysis was substantially enhanced by PD-1 blockade in *in vivo* PDX models of Hodgkin lymphoma**
- **AFM13-mediated anti-tumor efficacy results from innate to adaptive immune crosstalk and involves not only NK-cells and T-cells, but also macrophages and dendritic cells, thus forming an integrated immune response**

AFM13-mediated immune crosstalk not only involves NK- and T-cells, but also macrophages and dendritic cells in *in vivo* PDX models of Hodgkin lymphoma

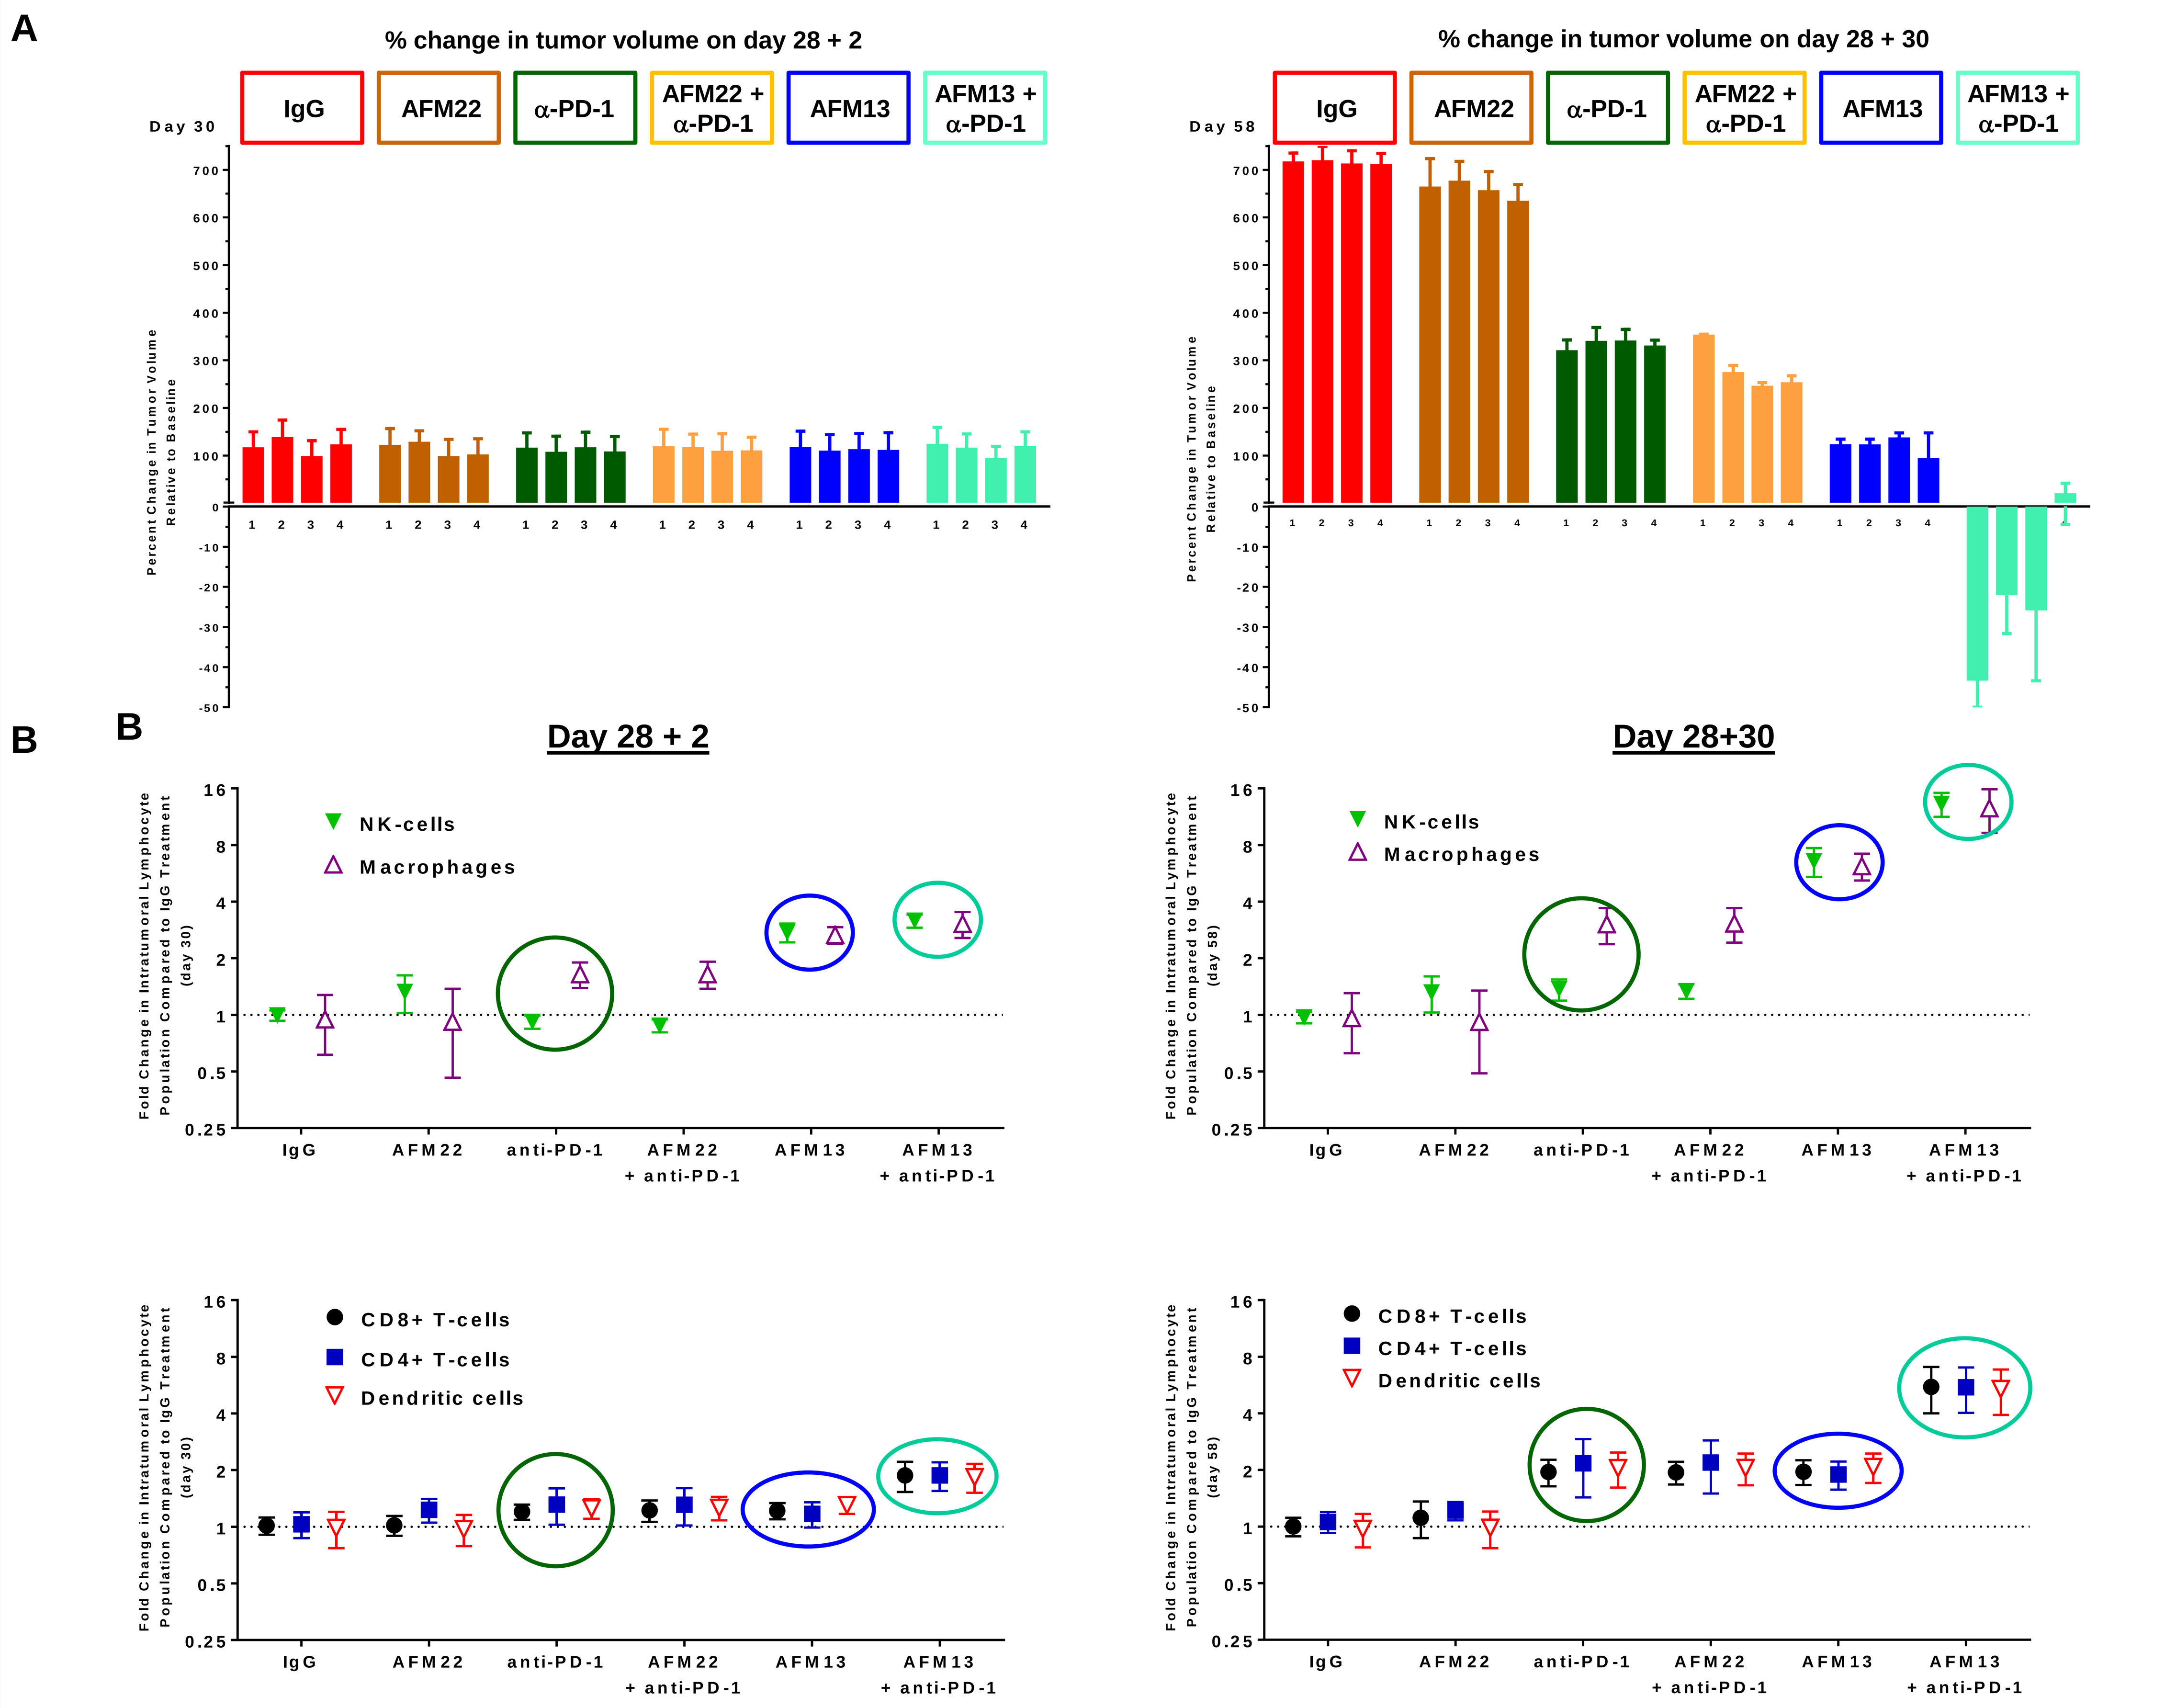
A
% change in tumor volume on day 28 + 2
% change in tumor volume on day 28 + 30
Day 30
Day 58
IgG
AFM22
α-PD-1
AFM22 + α-PD-1
AFM13
AFM13 + α-PD-1
Percent Change in Tumor Volume Relative to Baseline
B
Day 28 + 2
Day 28+30
NK-cells
Macrophages
CD8+ T-cells
CD4+ T-cells
Dendritic cells
Fold Change in Intratumoral Lymphocyte Population Compared to IgG Treatment
IgG
AFM22
anti-PD-1
AFM22 + anti-PD-1
AFM13
AFM13 + anti-PD-1

A
- AFM13 reduced tumor growth in all four PDX experiments (1-4) with higher efficacy than anti-PD-1 monotherapy.
- Combination of both AFM13 and anti-PD-1 synergized tumor growth control, whereas an unrelated IgG or CD16A TandAb (AFM22) did not reduce tumor growth.

B
- Treatment with AFM13 resulted in strong innate immune cell (NK-cells and macrophages) infiltration into tumors at early time points, and, after 58 days, also of adaptive immune cells such as T-cells.
- Combining AFM13 with anti-PD-1 potentiates the infiltration of all innate and adaptive immune cell populations including NK-cells, macrophages, CD4+, CD8+ T-cells into tumors.

C
- Treatment with AFM13 or anti-PD-1 enhanced the levels of intratumoral cytokines (e.g. IFN-γ) and the combination of both augmented intratumoral cytokine concentrations.

C **Cytokine assessment on day 58**

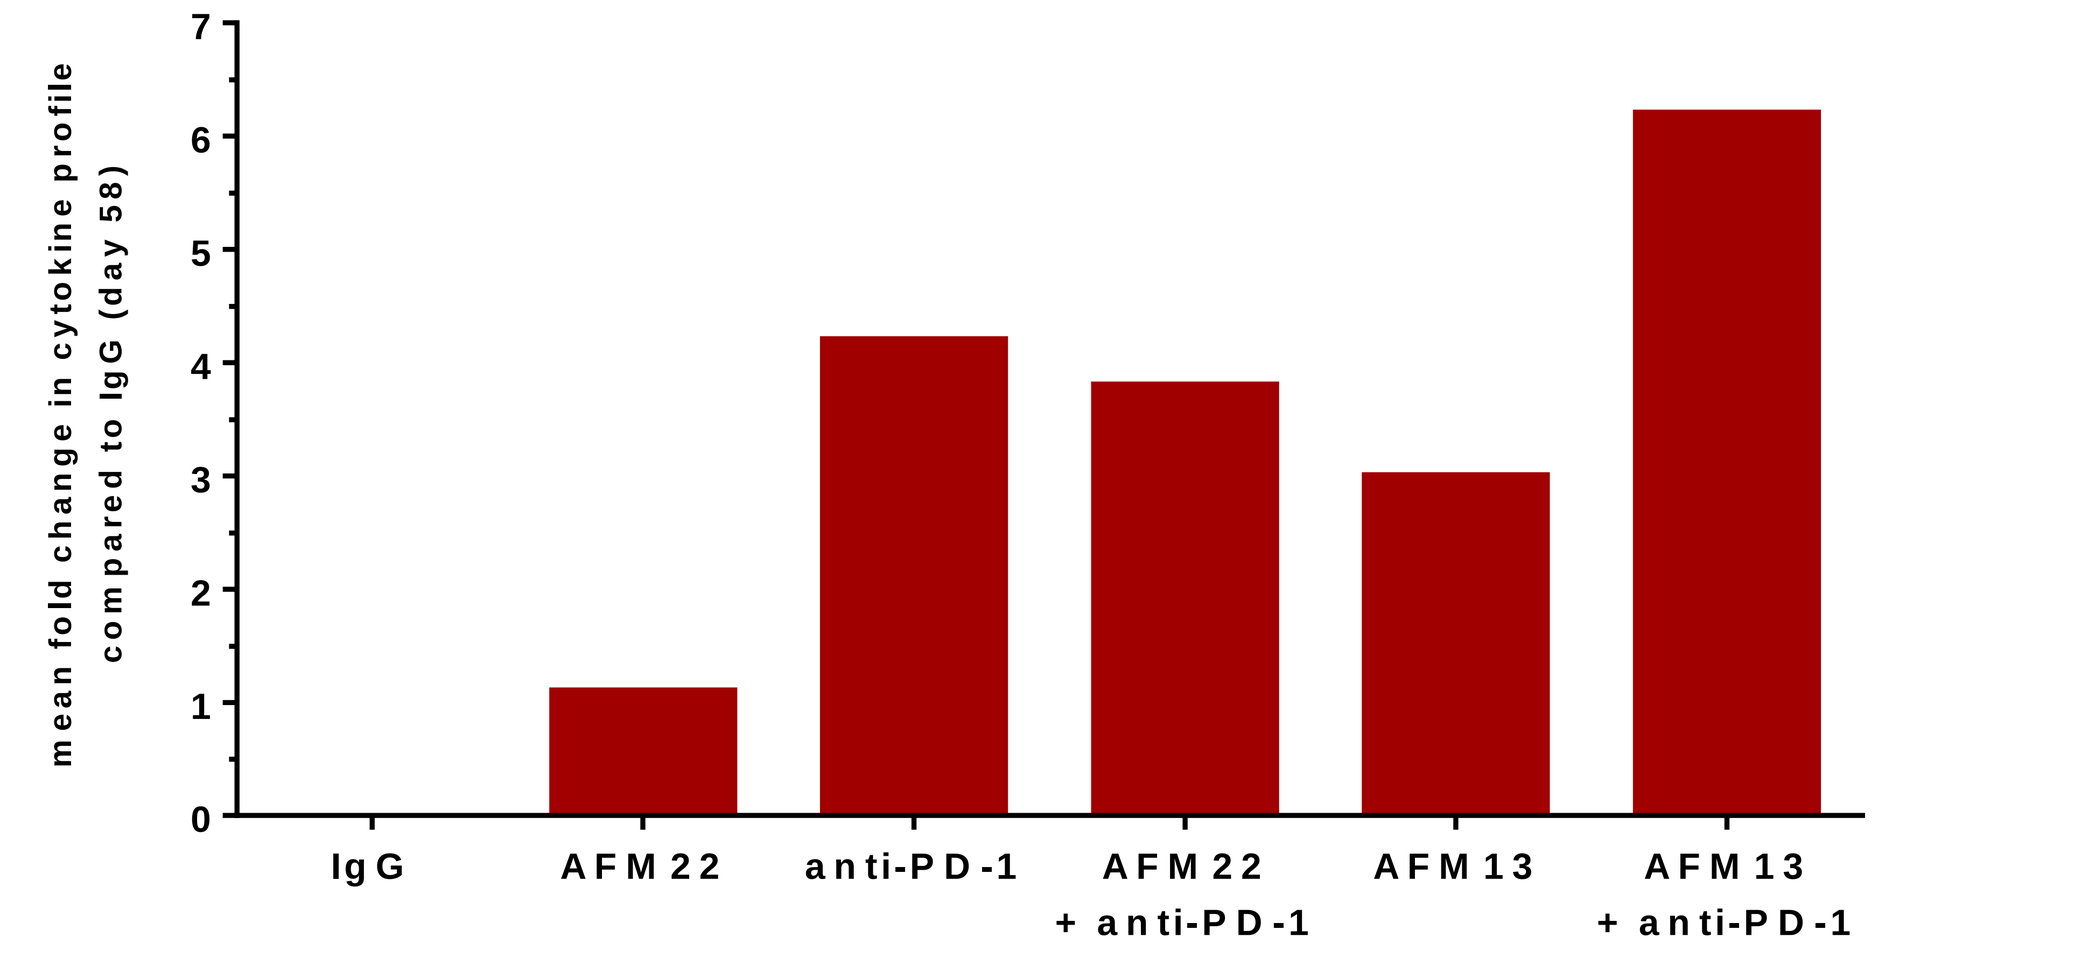
mean fold change in cytokine profile compared to IgG (day 58)
IgG
AFM22
anti-PD-1
AFM22 + anti-PD-1
AFM13
AFM13 + anti-PD-1

Corresponding author

References: 1. Reusch, U. *et al*. A novel tetravalent bispecific TandAb (CD30/CD16A) efficiently recruits NK cells for the lysis of CD30+ tumor cells. Mabs 6(3): 728–739 (2014).
2. Rothe, A. *et al*. A phase I study of the bispecific anti-CD30/CD16A antibody construct AFM13 in patients with relapsed or refractory Hodgkin lymphoma. Blood 125, 4024–4032 (2015).