Preliminary results from the Phase 2 study of AFM24 in combination with atezolizumab in patients with *EGFR* wild-type (*EGFR*-WT) non-small cell lung cancer (NSCLC)

2522

Hye Ryun Kim[1], Omar Saavedra[2], Andrés Cervantes[3], Iwona Lugowska[4], Arjun Oberoi[2], Anthony B. El-Khoueiry[5], Jacob Thomas[5], Wojciech Rogowski[6], Juanita S. Lopez[7], Byoung Yong Shim[8], Eric Christenson[9], Valentina Boni[10], Marina Chiara Garassino[11], Ramon Yarza[7], Ulrike Gärtner[12], Sarah Lutkewitz[12], Daniel Schütz[12], Kerstin Pietzko[12], Michael Emig[12], Daniela Morales Espinosa[12]

[1]Division of Medical Oncology, Yonsei Cancer Center, Yonsei University College of Medicine, Seoul, Republic of Korea; [2]Vall d'Hebron Institute of Oncology, Barcelona, Spain; [3]Hospital Clínico Universitario de Valencia, Valencia, Spain; [4]Maria Sklodowska-Curie National Institute of Oncology, Early-Phase Clinical Trials Unit, Warsaw, Poland; [5]University of Southern California, Norris Comprehensive Cancer Center, Los Angeles, CA, US; [6]Janusz Korczak Provincial Specialist Hospital, Slupsk, Poland; [7]Institute of Cancer Research at the Royal Marsden, Sutton, UK; [8]St Vincent Hospital, The Catholic University of Korea, Seoul, Republic of Korea; [9]Johns Hopkins University Hospital, Baltimore, MD, US; [10]Phase 1 Clinical Trial Unit, NEXT Oncology – Hospital Universitario Quirón Salud, Madrid, Spain; [11]University of Chicago, Department of Medicine, IL, US; [12]Affimed GmbH, Mannheim, Germany

BACKGROUND

- AFM24 is a tetravalent, bispecific Innate Cell Engager (ICE) that binds CD16A on natural killer (NK) cells and macrophages, and EGFR overexpressed on solid tumor cells, redirecting and enhancing the innate and possibly the adaptive immune response towards solid tumors[1,2]
- Unlike 'classical' targeted therapies that inhibit EGFR signalling activity, such as tyrosine kinase inhibitors and monoclonal antibodies, the effect of AFM24 does not depend on EGFR downstream signalling, nor on the presence of driver or resistance mutations in EGFR-expressing solid tumors[2–4]
- Atezolizumab, a PD-L1 inhibitor, abrogates activation of the PD-1 immune checkpoint, potentiating the adaptive immune response[5]
- We hypothesize that the combination of an ICE and an immune checkpoint inhibitor may synergistically enhance the cancer-immunity cycle and provide a promising, alternative strategy to overcome resistance to existing therapies for EGFR-expressing tumors[6]

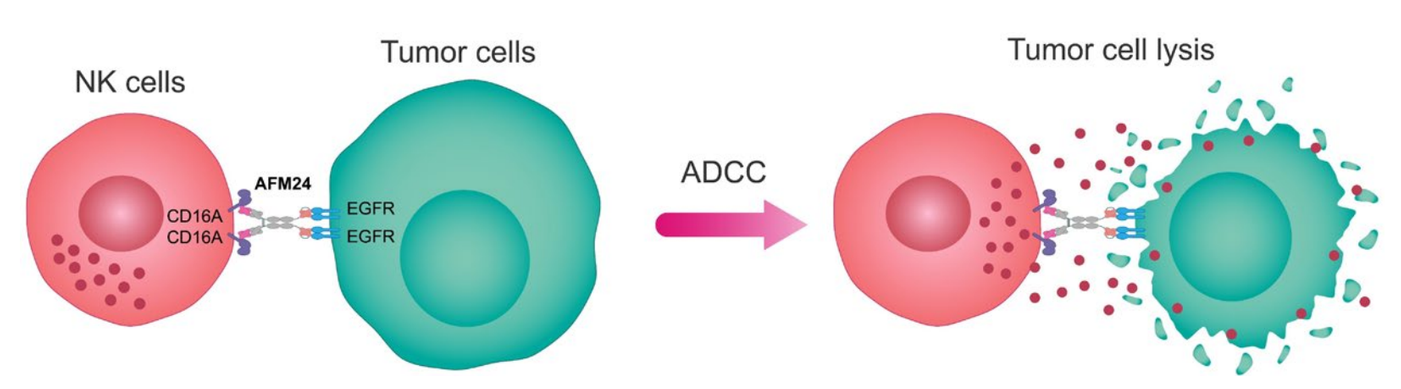
NK cells
Tumor cells
AFM24
CD16A
CD16A
EGFR
EGFR
ADCC
Tumor cell lysis

AFM24 mechanism of action

Simultaneous engagement of EGFR and CD16A with AFM24 facilitates potent, anti-EGFR-expressing tumor antibody-dependent cellular cytotoxicity (ADCC)

- This Phase 1/2a study (NCT05109442) is evaluating the combination of AFM24 with atezolizumab to target EGFR-expressing solid tumors; during the dose escalation phase, the recommended Phase 2 dose (RP2D) was established at 480 mg based on the well-tolerated safety profile and data from the AFM24 monotherapy trial[6]; here we present first data from the *EGFR*-WT NSCLC cohort

OBJECTIVE

To report the preliminary efficacy findings from the *EGFR*-WT NSCLC cohort during the dose expansion phase of the study

PHASE 2A STUDY DESIGN

Primary objective: Overall response rate by Investigator assessment (per RECIST v1.1)

Secondary objectives: Progression-free survival, duration of response, clinical benefit rate, safety, pharmacokinetics, immunogenicity

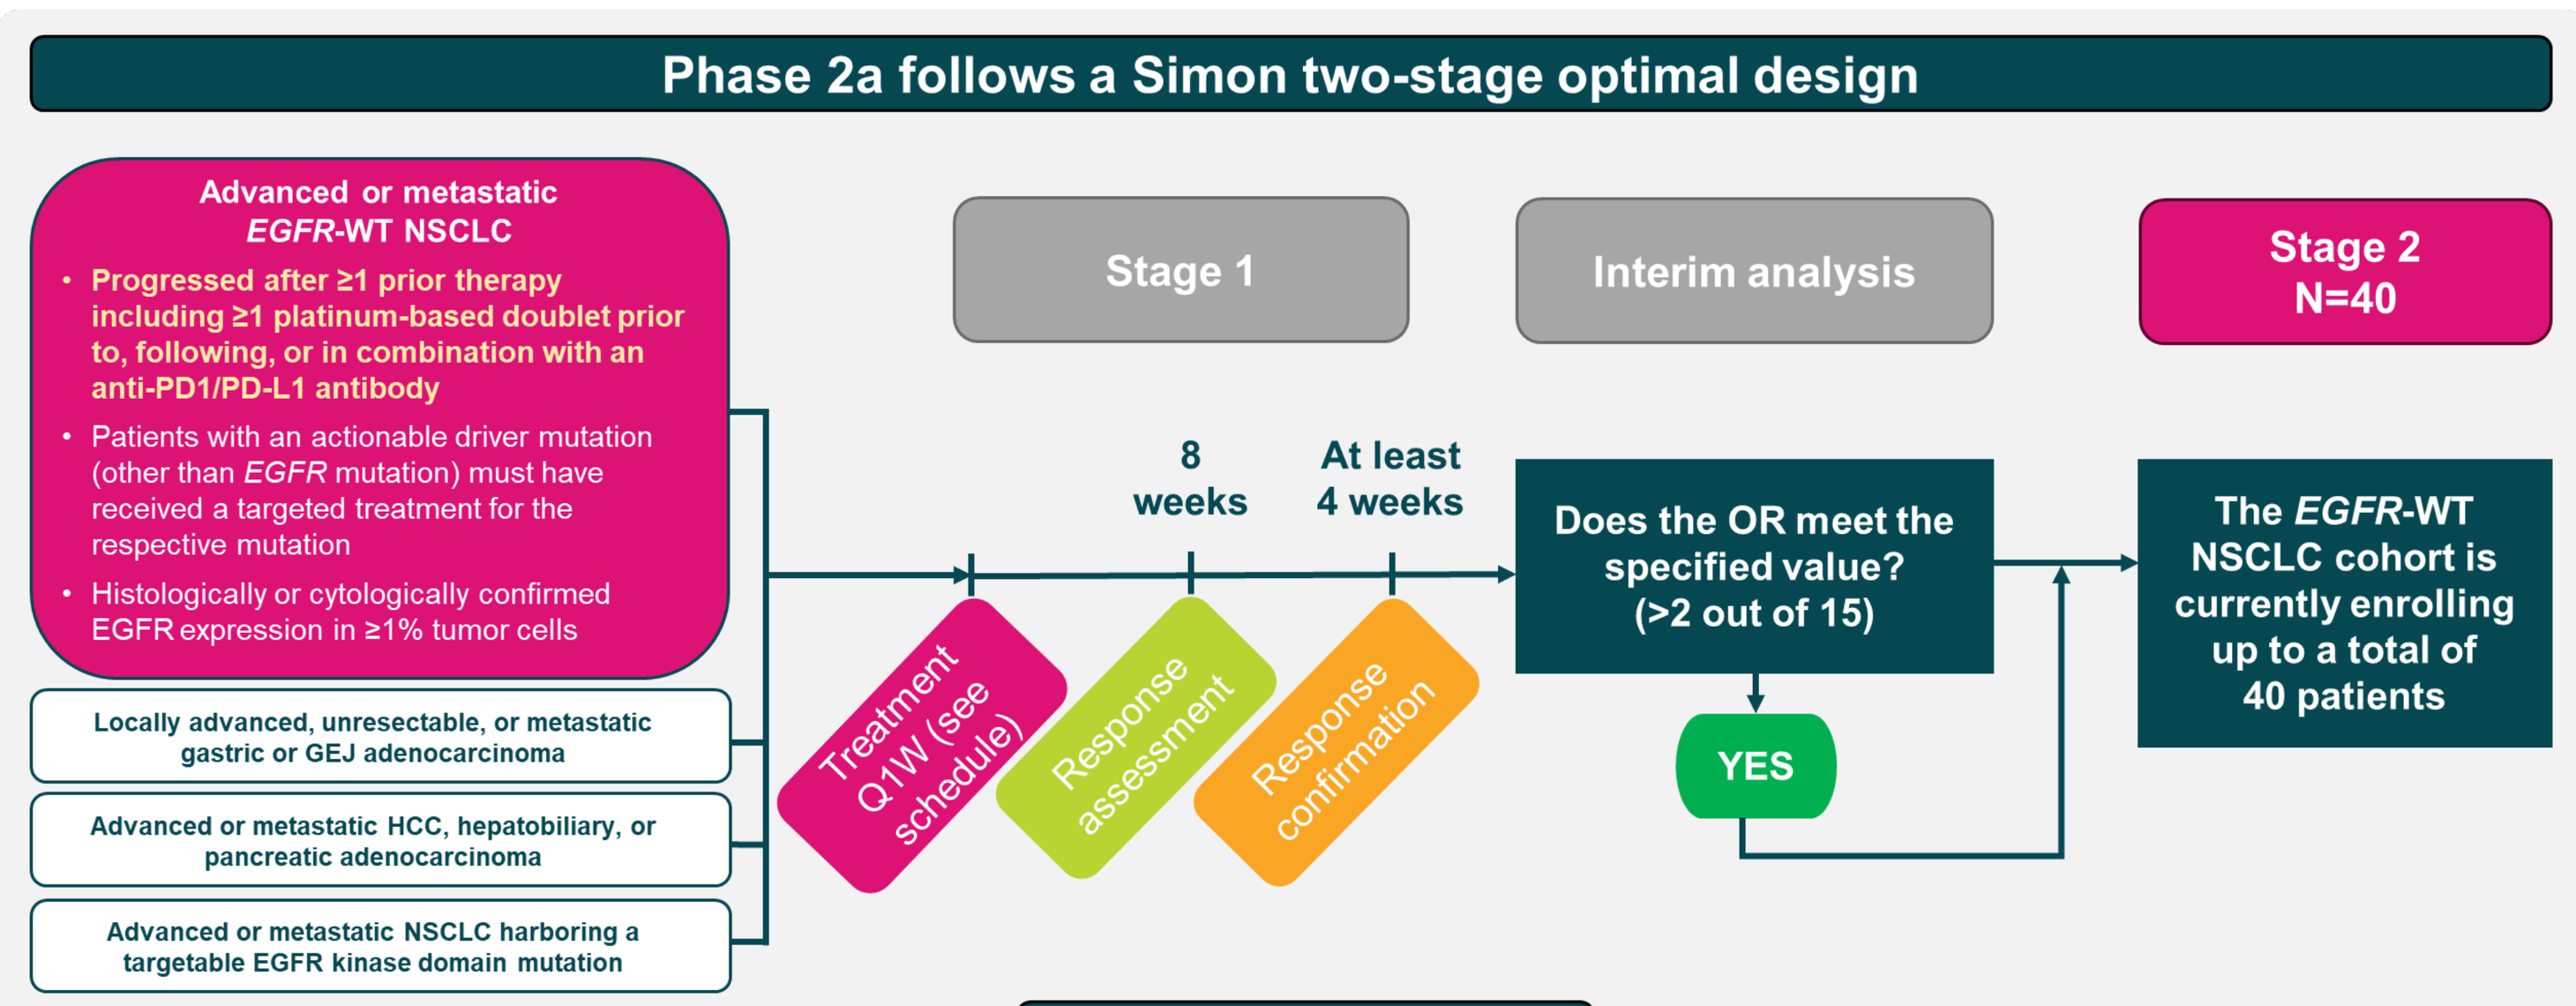
Phase 2a follows a Simon two-stage optimal design
Advanced or metastatic EGFR-WT NSCLC
Progressed after ≥1 prior therapy including ≥1 platinum-based doublet prior to, following, or in combination with an anti-PD1/PD-L1 antibody
Patients with an actionable driver mutation (other than EGFR mutation) must have received a targeted treatment for the respective mutation
Histologically or cytologically confirmed EGFR expression in ≥1% tumor cells
Locally advanced, unresectable, or metastatic gastric or GEJ adenocarcinoma
Advanced or metastatic HCC, hepatobiliary, or pancreatic adenocarcinoma
Advanced or metastatic NSCLC harboring a targetable EGFR kinase domain mutation
Stage 1
Interim analysis
Stage 2 N=40
8 weeks
At least 4 weeks
Treatment Q1W (see schedule)
Response assessment
Response confirmation
Does the OR meet the specified value? (>2 out of 15)
YES
The EGFR-WT NSCLC cohort is currently enrolling up to a total of 40 patients

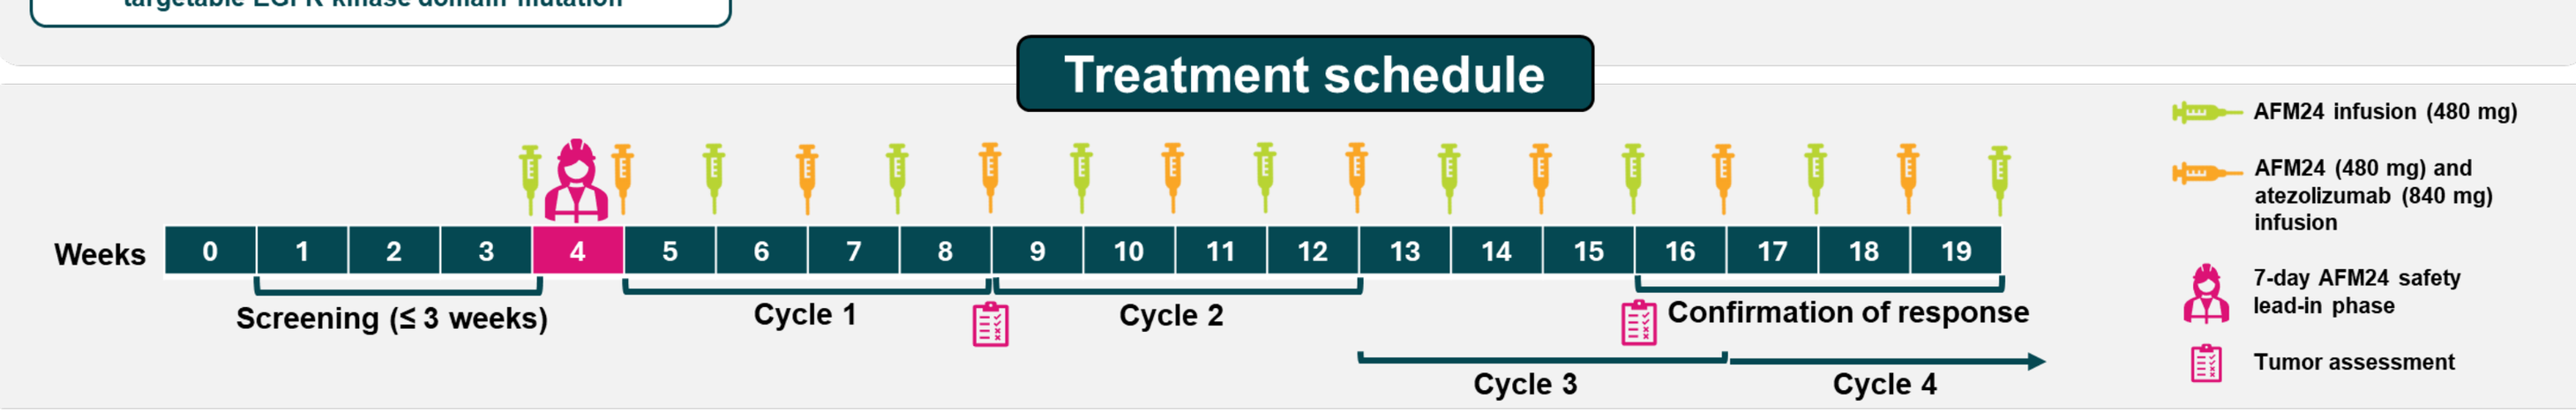
Treatment schedule
Weeks
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19
Screening (≤ 3 weeks)
Cycle 1
Cycle 2
Cycle 3
Cycle 4
Confirmation of response
AFM24 infusion (480 mg)
AFM24 (480 mg) and atezolizumab (840 mg) infusion
7-day AFM24 safety lead-in phase
Tumor assessment

AFM24 is administered IV Q1W, and atezolizumab IV Q2W in 28-day cycles for as long as clinical benefit is observed, or until disease progression, intolerable toxicity, Investigator discretion, or subject withdrawal of consent.
Tumor assessments are performed at initial screening, Cycles 2, 4, 6, 8, 10, 12 and every three cycles thereafter.

GEJ, gastro-esophageal junction; HCC, hepatocellular carcinoma; IV, intravenous; NSCLC, non-small cell lung cancer; OR, overall response; Q1W, once weekly; Q2W, once fortnightly; WT, wildtype.

RESULTS

Baseline characteristics

- As of March 18th 2024, 17 patients with *EGFR*-WT NSCLC had been treated with the combination, receiving a median (range) of 13 (2–35) doses of AFM24 over a duration of 17 (2–37) weeks

Table 1: Baseline characteristics of the *EGFR*-WT NSCLC cohort (n=17)

Age (years), n (%)	
Median (range)	**65.0 (40–75)**
18–64	8 (47.1)
≥65	9 (52.9)
Sex (male), n (%)	14 (82.4)
Ethnicity, n (%)	
Asian	9 (52.9)
White	8 (47.1)
Histopathological type at diagnosis, n (%)	
Adenocarcinoma	11 (64.7)
Squamous cell carcinoma	6 (35.3)
ECOG PS, n (%)	
0	2 (11.8)
1	15 (88.2)
Prior lines of therapy	
Median (range)	2 (1–5)

ECOG, Eastern Cooperative Oncology Group; NSCLC, non-small cell lung cancer; PS, performance score; WT, wild-type.

Safety

- A tolerable, well-managed safety profile was observed

Table 2: Summary of TEAEs, n patients, % (n=17)*

	AFM24-related	Atezolizumab-related	All
TEAE	13 (76.5)	9 (52.9)	17 (100.0)
TEAE ≥Grade 3	1 (5.9)	0 (0.0)	8 (47.1)
Serious TEAE	1 (5.9)	0 (0.0)	8 (47.1)
Fatal TEAE	0 (0.0)	0 (0.0)	2 (11.8)
TEAE leading to study drug discontinuation	0 (0.0)	0 (0.0)	0 (0.0)

*Each patient is counted a single time for each applicable specific adverse event with highest severity.
TEAE, treatment-emergent adverse event.

- Most AFM24 treatment-related adverse events (TRAEs) were mild/moderate
 - The most frequently reported AFM24 TRAEs were infusion-related reactions (IRRs); IRRs mainly occured at initial AFM24 infusion during the safety lead-in period (Cycle 1, Day −7) and resolved
- Three AFM24 TRAEs of ≥Grade 3 were observed in one patient
 - These included alanine aminotransferase and aspartate aminotransferase increase, and cytokine release syndrome (one event each)
- One serious AFM24 TRAE occurred (Grade 2 IRR)
- No Grade 4 or 5 AFM24 TRAEs were observed

Table 3: Summary of AFM24 TRAEs by Grade (≥5% patients), n patients, % (n=17)

	Grade 1/2	Grade ≥3	Any Grade
IRR	8 (47.1)	0 (0.0)	8 (47.1)
Fatigue	4 (23.5)	0 (0.0)	4 (23.5)
ALT increased	2 (11.8)	1 (5.9)	3 (17.6)
AST increased	2 (11.8)	1 (5.9)	3 (17.6)
Rash	2 (11.8)	0 (0.0)	2 (11.8)
Asthenia	2 (11.8)	0 (0.0)	2 (11.8)

ALT, alanine aminotransferase; AST, aspartate aminotransferase; IRR, infusion-related reaction; TRAE, treatment-related adverse event.

In evaluable patients (n=15), AFM24 combined with atezolizumab exhibited a disease control rate (DCR) of 73.3%

RECIST status and treatment durations (by CT per Investigator assessment)

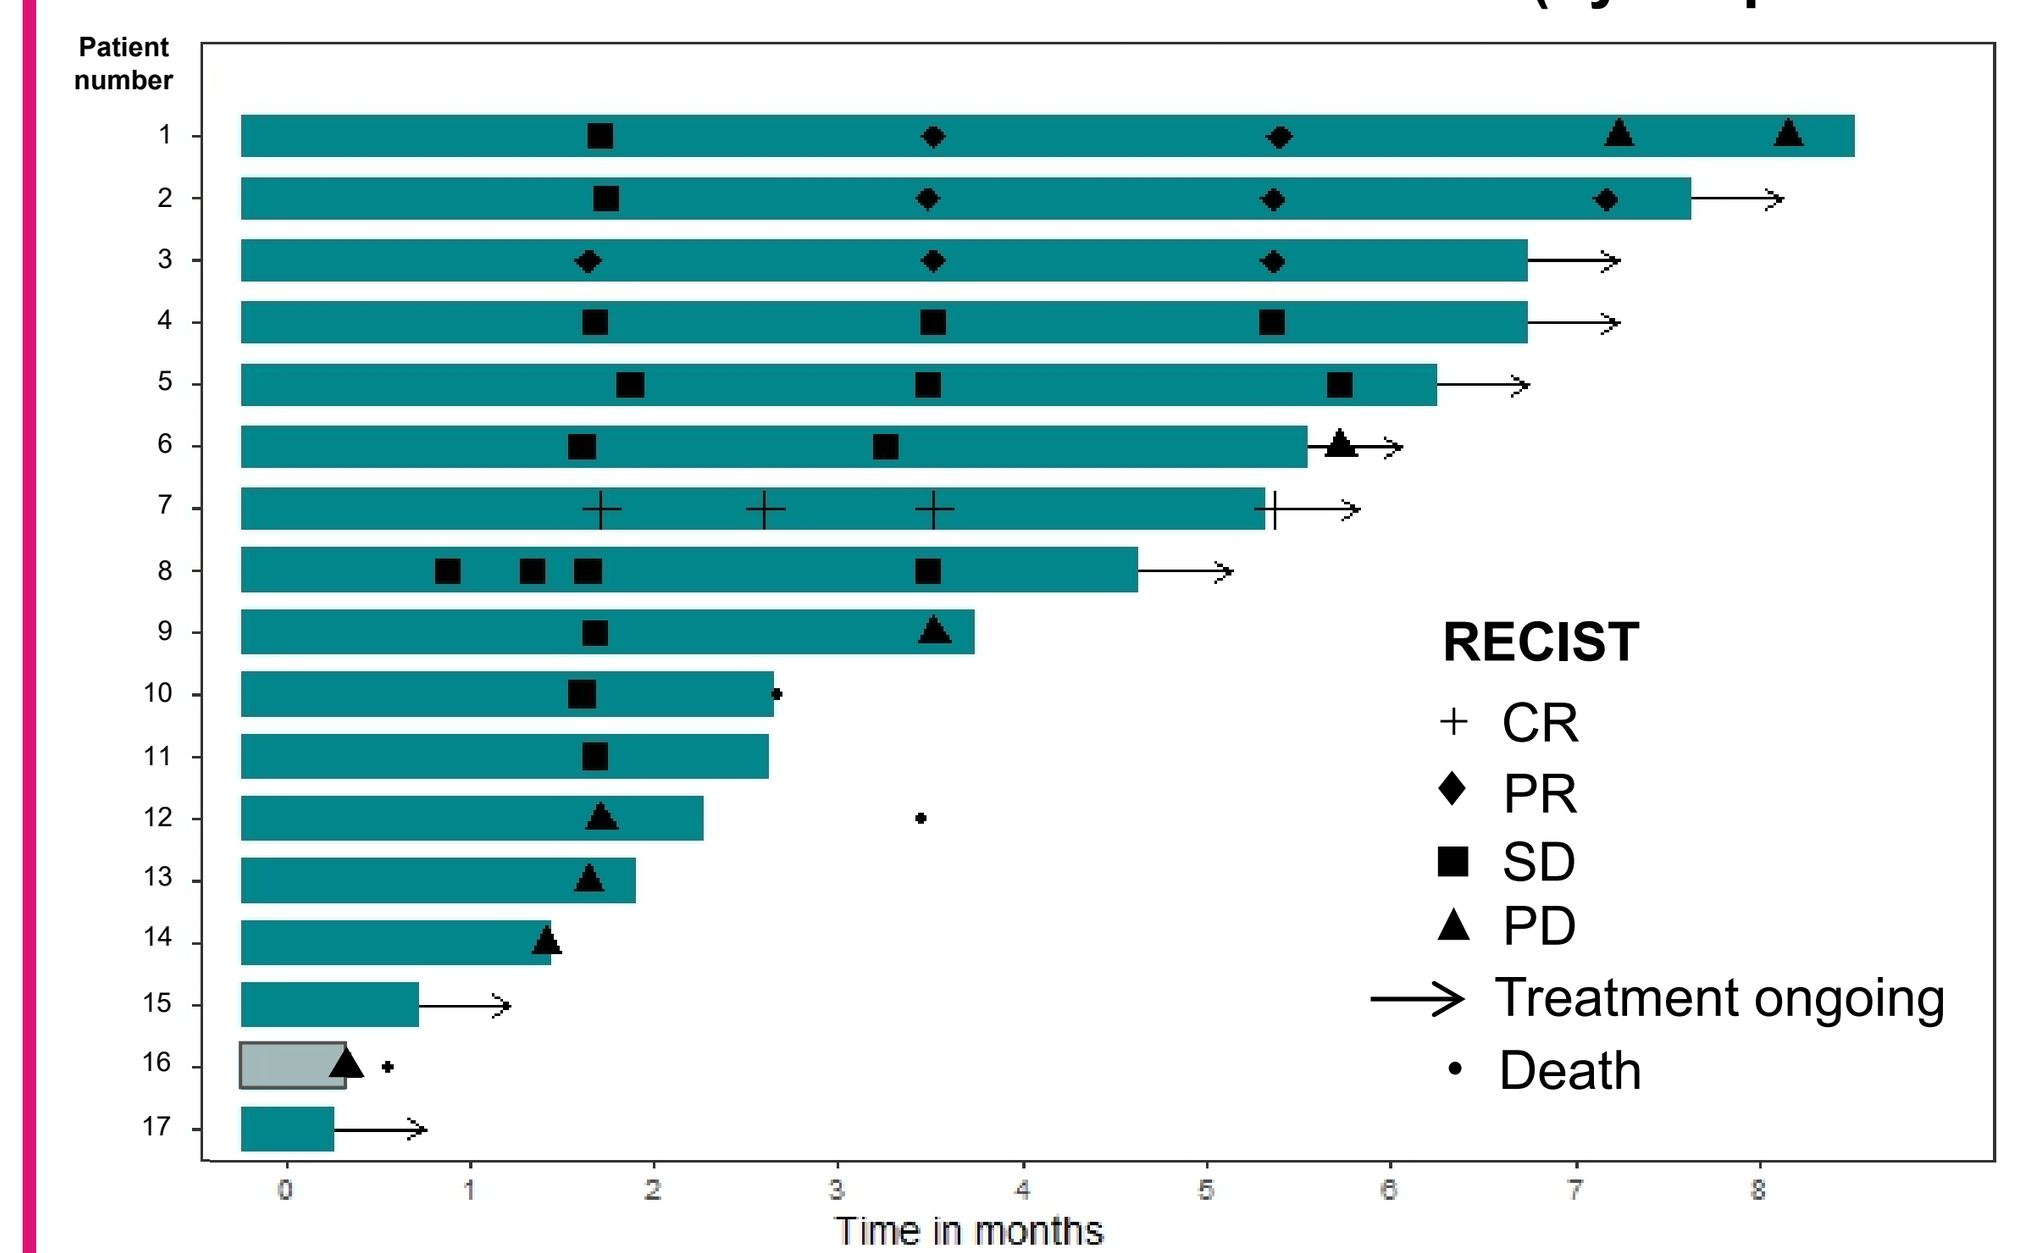
Patient number
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17
0 1 2 3 4 5 6 7 8
Time in months
RECIST
CR
PR
SD
PD
Treatment ongoing
Death

Each bar represents one patient. As of March 2024, 17 patients were included in the full analysis set (FAS). There were 15 patients that had at least one post-baseline CT scan available (i.e. response evaluable).

CI, confidence interval; CR, complete response; CT, computerized tomography; DCR, disease control rate; PD, progressive disease; PFS, progression-free survival; PR, partial response; SD, stable disease.

After >5 months on treatment, one patient had an ongoing, confirmed CR

Three patients had PRs; at >5 months on treatment, two are ongoing

Seven patients exhibited SD; of these patients, four exhibited SD for ≥3.5 months

At the data cut-off, the median (range) PFS with AFM24 and atezolizumab was 5.9 (1.84–inf) months; median follow-up time was 5.6 months

A patient exhibiting a PR experienced a 50% overall shrinkage of their target lesions with AFM24 and atezolizumab

74-year-old Asian male

Diagnosed June 2020

Metastatic lung squamous cell carcinoma
- Stage IV with one target lesion: Right upper lobe and two non-target lesions mediastinal lymph nodes and left adrenal gland

Treatment history

Jul 2020 – Sept 2020	Oct 2020 – Jun 2022	May 2023 – Jun 2023	August 2023 – Present
Tiragolumab/placebo + atezolizumab BOR = SD Discontinued due to PD	Cisplatin + pemetrexed + gemcitabine BOR = PR Discontinued due to toxicity	SRF388 BOR = PD	AFM24 + atezolizumab

CT scan images of the right upper lobe lesion of a patient exhibiting a confirmed PR on 480 mg AFM24 + atezolizumab

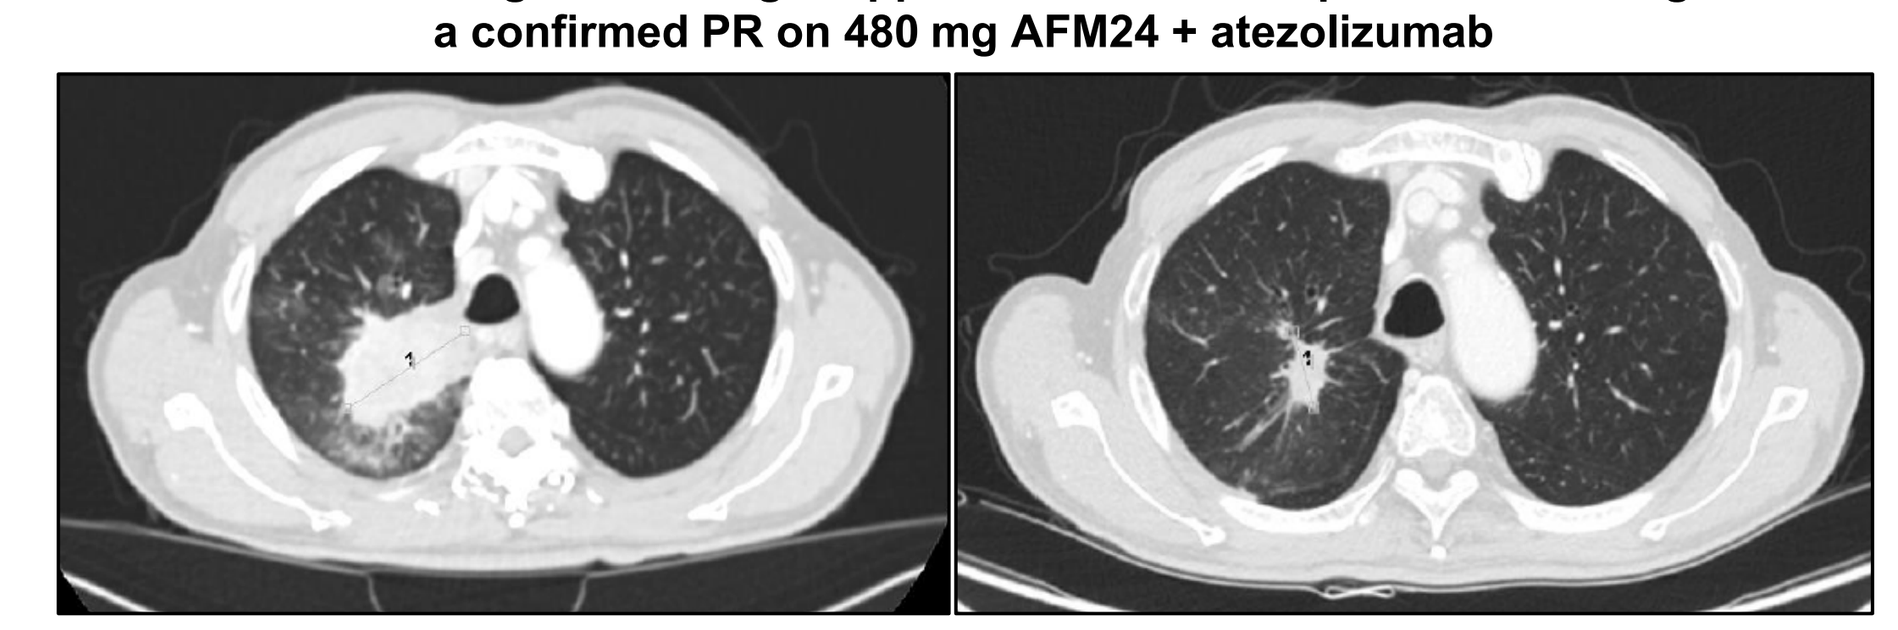

Tumor response by Investigator assessment per RECIST v1.1

Sum of diameters, target lesions in mm

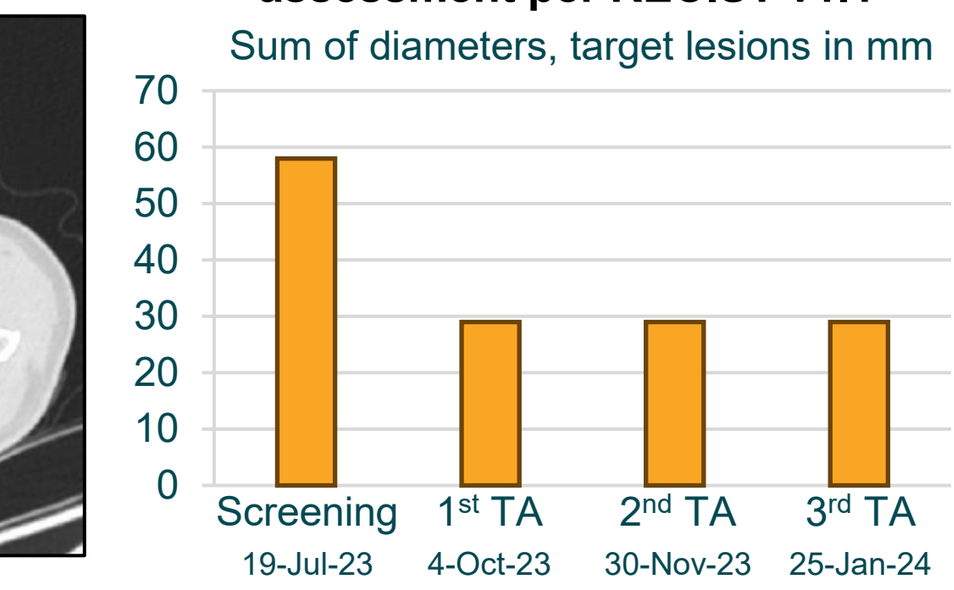
70
60
50
40
30
20
10
0
Screening 19-Jul-23
1st TA 4-Oct-23
2nd TA 30-Nov-23
3rd TA 25-Jan-24

Informed consent was obtained from the patient for the publication of these images. BOR, best objective response; CT, computerized tomography; PD, progressive disease; PR, partial response; SD, stable disease; TA, tumor assessment.

A reduction in tumor burden was observed in seven patients; several patients also exhibited deepening of responses to AFM24 in combination with atezolizumab

A **Best percentage change from baseline in sum of long diameter (by CT per Investigator assessment)**

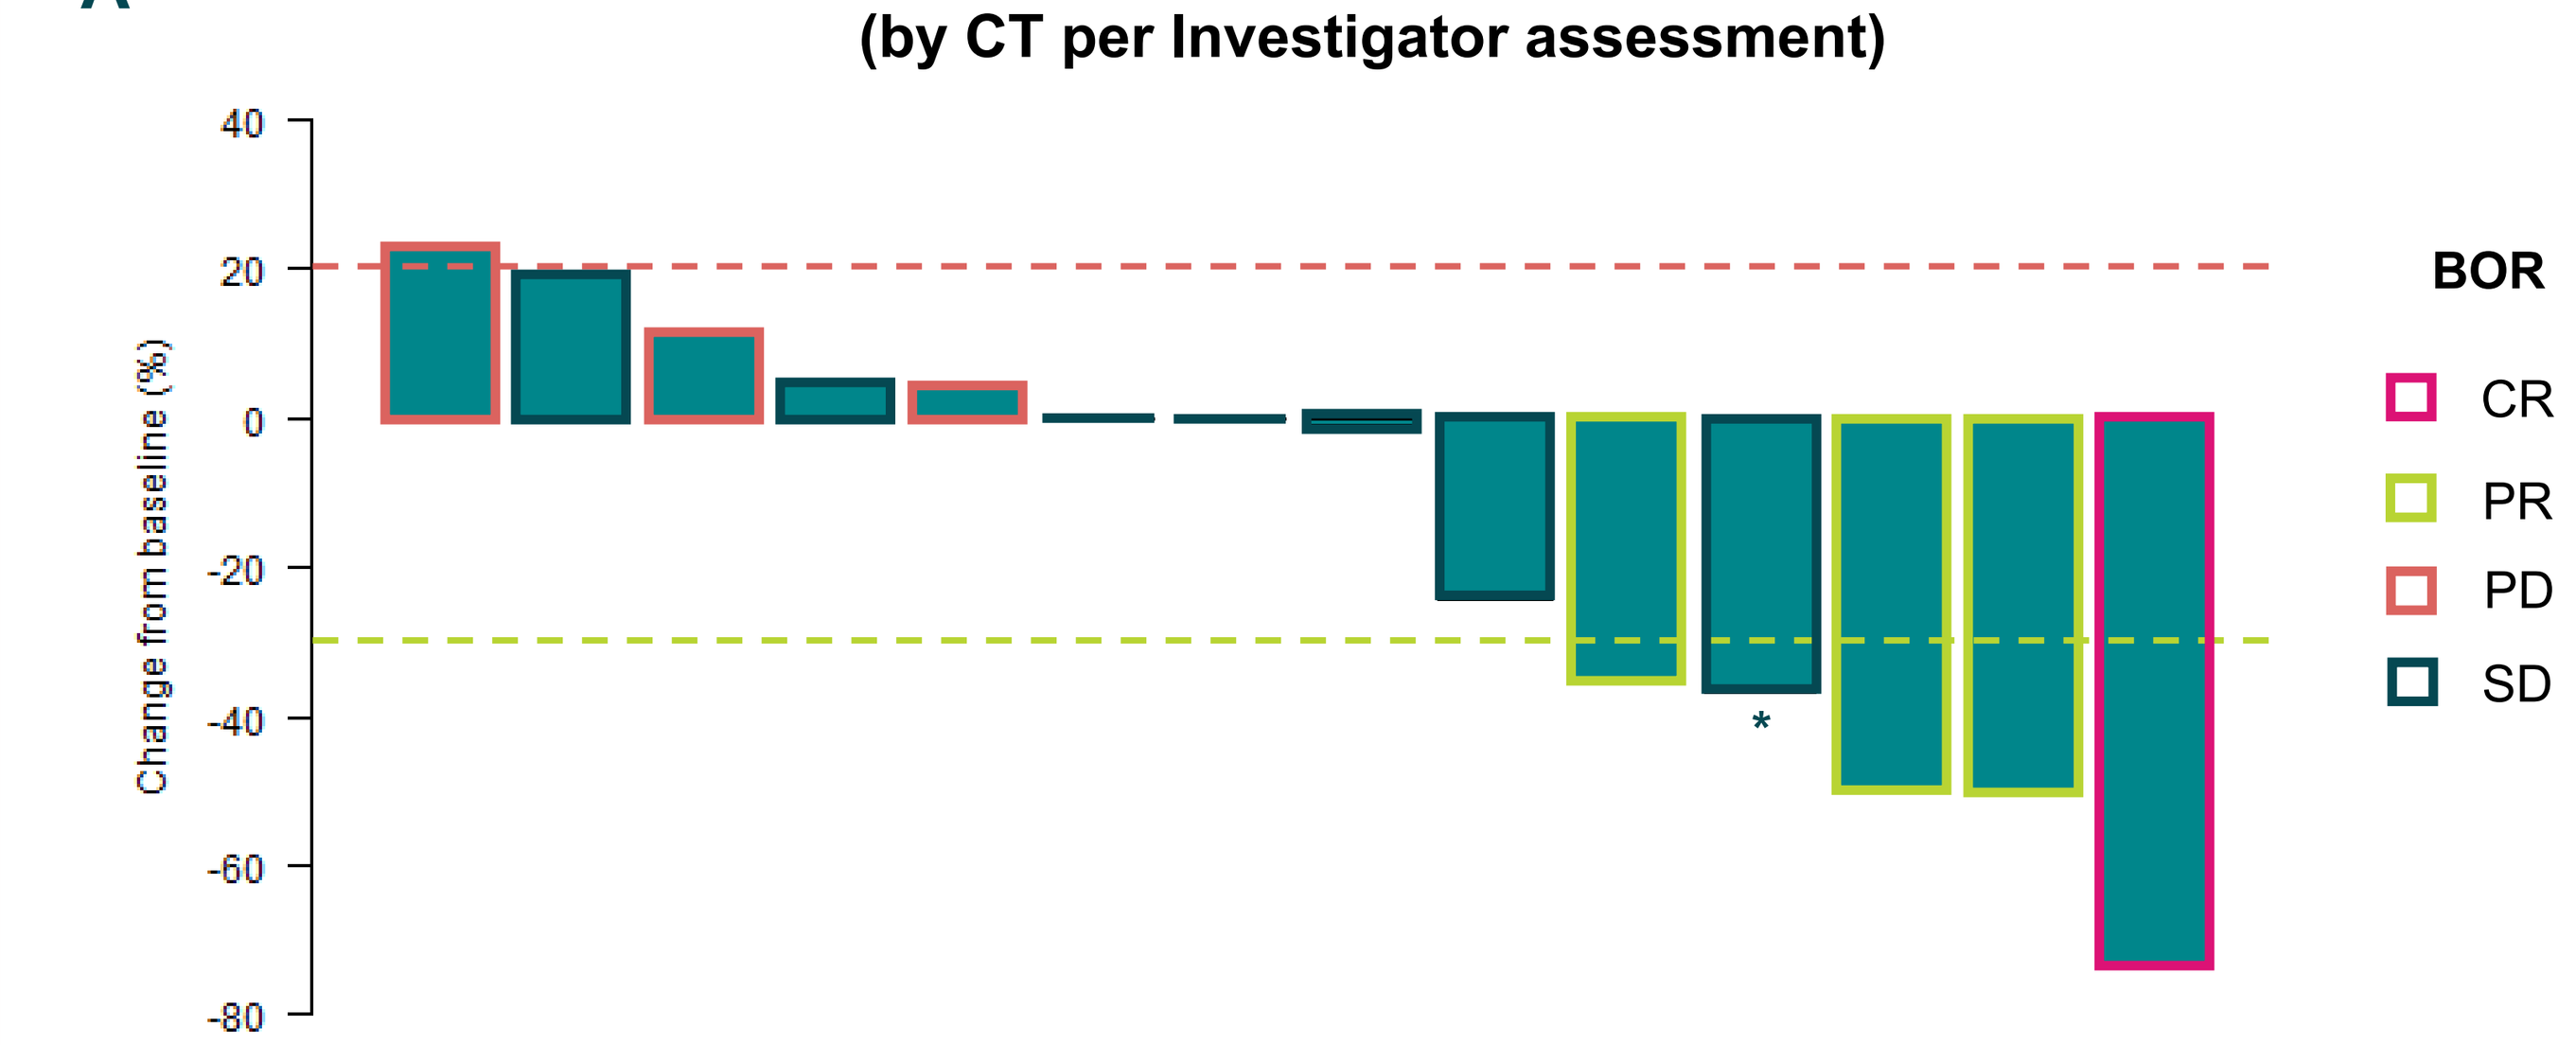
Change from baseline (%)
40
20
0
-20
-40
-60
-80
*
BOR
CR
PR
PD
SD

Seven out of 15 evaluable patients exhibited tumor shrinkage following AFM24 plus atezolizumab therapy**

B **Percentage change from baseline in target lesions over time (by CT per Investigator assessment)**

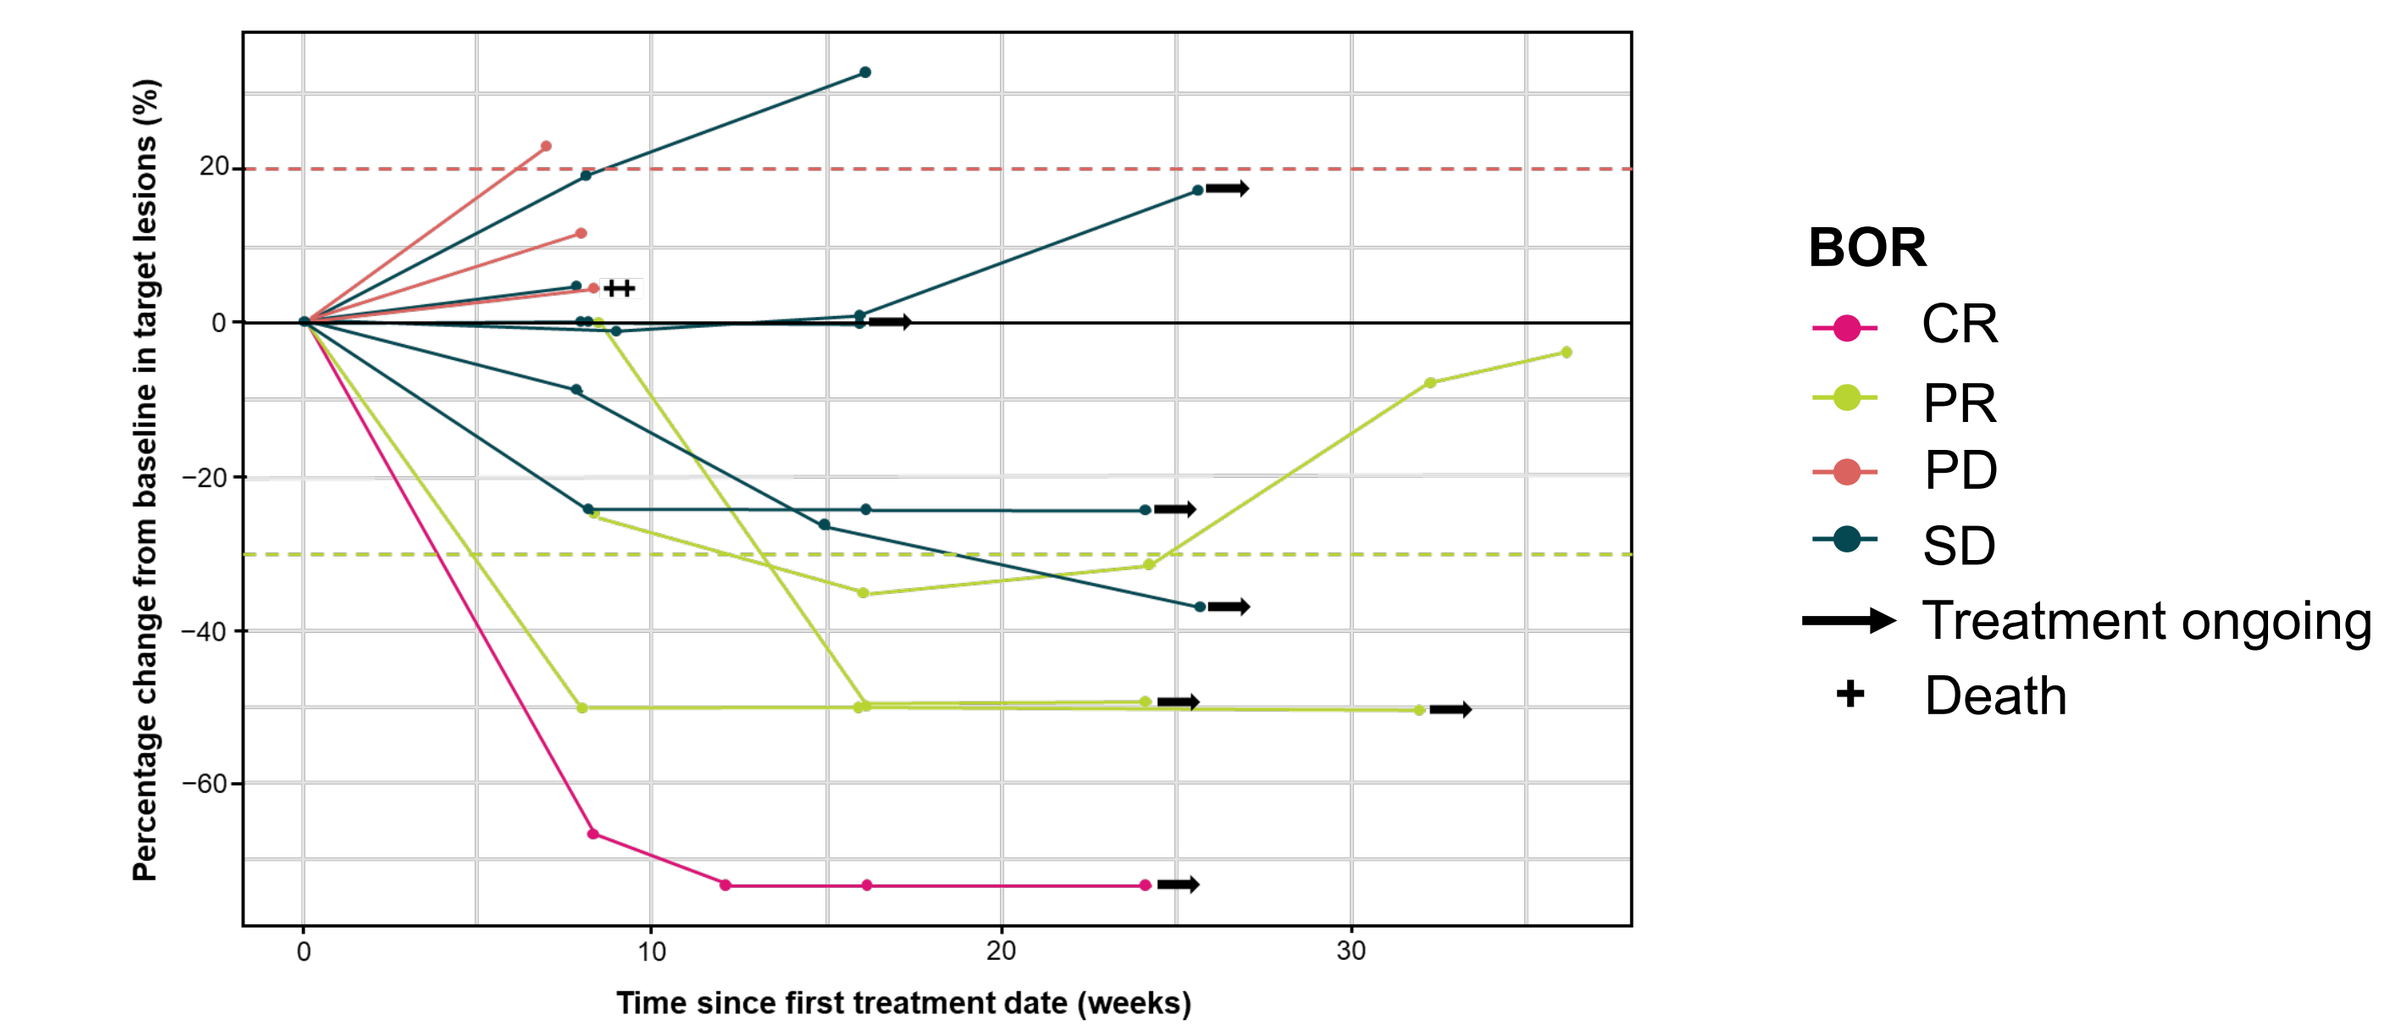
Percentage change from baseline in target lesions (%)
20
0
−20
−40
−60
0 10 20 30
Time since first treatment date (weeks)
BOR
CR
PR
PD
SD
Treatment ongoing
Death

In several patients, a deepening of response occurred after 10 weeks (2.5 cycles) on AFM24 plus atezolizumab therapy

Each bar/line represents one patient. A. Tumor diameter was measured at baseline and at subsequent tumor assessments by CT scan, and greatest change in diameter presented as percentage change from baseline. B. Target lesion diameter was measured at baseline and at subsequent tumor assessments following combination therapy by CT scan, and change in diameter over time presented as percentage change from baseline. *This patient exhibited a shrinkage in target lesion on the same day as new lesions were observed; their best response prior to this progressive disease was SD. **One patient (no. 16 on the swimmer plot) had their scan prior to the assessment period and consequently was excluded from these analyses.

BOR, best overall response. CR, complete response; CT, computerized tomography; PD, progressive disease; PR, partial response; SD, stable disease.

CONCLUSIONS

- **AFM24 in combination with atezolizumab exhibited promising clinical activity in a heavily pre-treated cohort of patients with *EGFR*-WT NSCLC; the DCR was 73.3%, with one patient exhibiting a CR, three patients exhibiting a PR and seven patients had SD**
- **All patients who responded were resistant to prior checkpoint inhibitor therapy, supporting the hypothesis that combining AFM24 with atezolizumab may provide an alternative strategy to overcome resistance to existing therapies for EGFR-expressing tumors**
- **The safety profile was tolerable and well managed; the majority of AFM24 TRAEs were mild to moderate**
- **The study is ongoing, with the *EGFR*-WT NSCLC expansion cohort currently enrolling up to a total of 40 patients**

The authors would like to thank all patients, carers, investigators and site staff for their invaluable contributions to the study.
Poster presented at the American Society of Clinical Oncology (ASCO) Annual Meeting, Chicago, USA, May 31st to June 4th, 2024. Medical writing support provided by Meridian HealthComms, Macclesfield, UK.
For more information, please contact d.morales-espinosa@affimed.com

REFERENCES
1. Ellwanger K, et al. mAbs 2019;11:899–918 | 2. Wingert S, et al. mAbs 2021;13(1):1950264 | 3. Ayati A, et al. Bioorg Chem 2020;99:103811 | 4. Zubair T and Bandyopadhyay D. Int J Mol Sci 2023;24(3):2651 | 5. Santini F and Rudin C. Expert Rev Clin Pharmacol 2017;10(9):935–945 | 6. El-Khoueiry AB, et al. Ann Oncol 2022;33(7):5889.