AFM24 in Combination with Atezolizumab in Patients with Advanced EGFR-Expressing Solid Tumors: Phase 1/2a Study Design and Rationale

TPS2673

Omar Saavedra[1], Eric Christenson[2], Anthony El-Khoueiry[3], Andrés Cervantes[4], Christa Raab[5], Ulrike Gärtner[5], Kerstin Pietzko[5], Gabriele Hintzen[5], Paulien Ravenstijn[5], Daniela Morales-Espinosa[5], Juanita Lopez[6]

[1]Vall d'Hebron Institute of Oncology, Barcelona, Spain, [2]Johns Hopkins University Hospital, Baltimore, MD, US, [3]University of Southern California, Norris Comprehensive Cancer Center, Los Angeles, CA, US, [4]Hospital Clínico Universitario de Valencia, Valencia, Spain, [5]Affimed GmbH, Heidelberg, Germany, [6]Institute of Cancer Research at the Royal Marsden, Sutton, UK

BACKGROUND

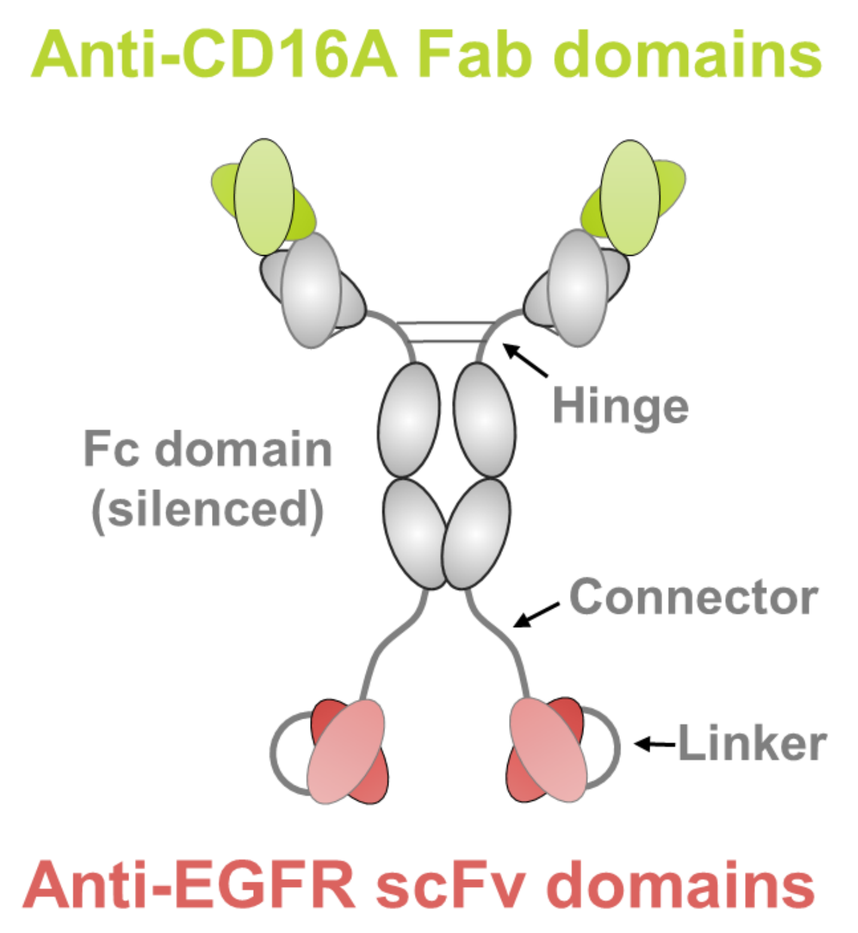
Anti-CD16A Fab domains
Hinge
Fc domain (silenced)
Connector
Linker
Anti-EGFR scFv domains

- Epidermal growth factor receptor (EGFR) is frequently overexpressed on the cell surface of various solid malignancies, and is associated with poor patient prognosis[1]
- AFM24 is a first-in-class, tetravalent, bispecific innate cell engager (ICE®) that targets EGFR+ tumor cells[2]
- AFM24 binds both to EGFR on the surface of tumor cells, and to CD16A on natural killer (NK) cells and macrophages, enhancing antibody-dependent cellular cytotoxicity (ADCC) and antibody-dependent cellular phagocytosis (ADCP), respectively[2]
- Preclinical studies of AFM24 have demonstrated efficacious tumor cell killing independent of EGFR mutational status[3]; an ongoing Phase 1/2a study of AFM24 monotherapy in EGFR+ solid tumors (NCT04259450) has demonstrated a well-managed safety profile[4]
- Atezolizumab is a monoclonal antibody that binds programmed-death ligand-1 (PD-L1), enhancing the adaptive immune response through suppression of the PD-1 immune checkpoint[5]
- Atezolizumab has shown clinical efficacy and a favorable safety profile in solid tumors[6–8]
- The combination of AFM24 with atezolizumab may provide a new treatment modality leveraging both innate and adaptive immune responses to target EGFR+ tumors

OBJECTIVE

To investigate the safety, tolerability and efficacy of AFM24 in combination with atezolizumab for the treatment of patients with advanced EGFR+ solid tumors

METHODS

- An ongoing Phase 1/2a open-label, non-randomized, multicenter, dose escalation (Phase 1) and dose expansion (Phase 2a) study was initiated in November 2021 (NCT05109442)
- Patients will receive combination therapy until disease progression, intolerable toxicity, patient withdrawal of consent, or termination at the Investigator's discretion

Phase 1/2a study design

Phase 1:

- Patients will undergo a safety lead-in phase with AFM24 as a single agent at the dose assigned to the relevant cohort 7 days before receiving the combination therapy
- A standard 3+3 dose escalation design will be used to determine the maximum tolerated dose (MTD) and/or recommended phase 2 dose (RP2D)
- Escalating doses of AFM24 will be given to each cohort as weekly intravenous (IV) infusions; the starting dose and at least two dose escalations are planned based on results from the ongoing AFM24 monotherapy trial; atezolizumab will be given at a fixed dose of 840 mg as a single IV infusion once every two weeks
- MTD will be defined as the highest dose level below the maximum administered dose where the number of patients with DLT is <2 out of 6; RP2D will be defined as any dose level where the MTD has not been met, where clinical, pharmacokinetics (PK), pharmacodynamics (PD) and/or laboratory data indicate that a biologically active dose has been reached
- Safety follow-ups will be carried out at the end of treatment (14 days post-final dose), 30 days, and subsequently every 3 months to accrue long-term data

Phase 2a:

- A Simon two-stage optimal design will be utilized for the malignancies indicated below which meet the inclusion criteria

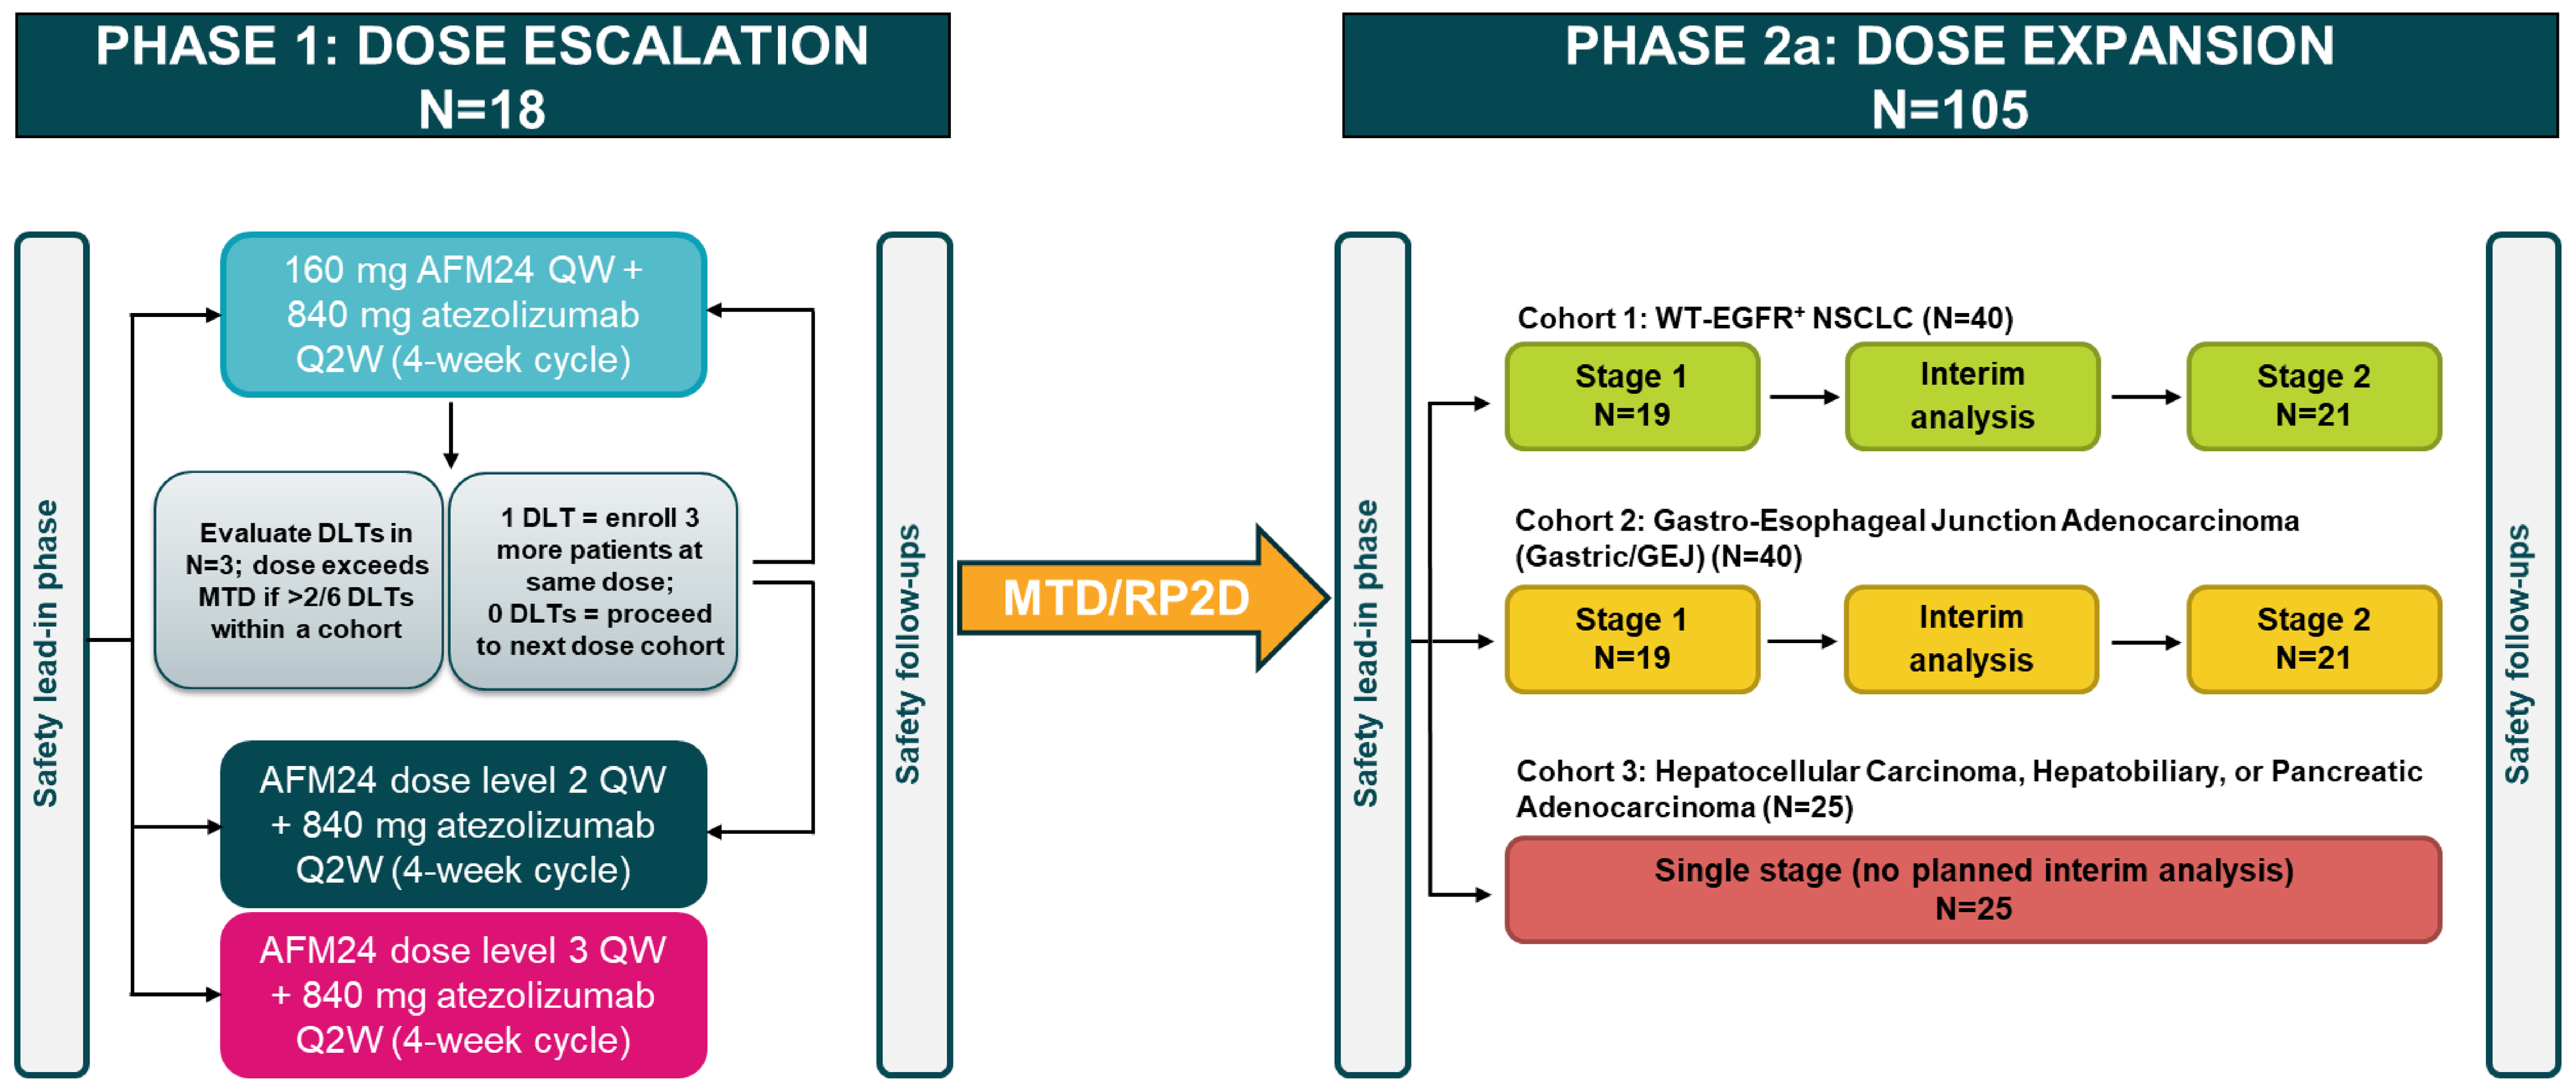
PHASE 1: DOSE ESCALATION
N=18
PHASE 2a: DOSE EXPANSION
N=105
Safety lead-in phase
160 mg AFM24 QW + 840 mg atezolizumab Q2W (4-week cycle)
Evaluate DLTs in N=3; dose exceeds MTD if >2/6 DLTs within a cohort
1 DLT = enroll 3 more patients at same dose; 0 DLTs = proceed to next dose cohort
AFM24 dose level 2 QW + 840 mg atezolizumab Q2W (4-week cycle)
AFM24 dose level 3 QW + 840 mg atezolizumab Q2W (4-week cycle)
Safety follow-ups
MTD/RP2D
Safety lead-in phase
Cohort 1: WT-EGFR+ NSCLC (N=40)
Stage 1 N=19
Interim analysis
Stage 2 N=21
Cohort 2: Gastro-Esophageal Junction Adenocarcinoma (Gastric/GEJ) (N=40)
Stage 1 N=19
Interim analysis
Stage 2 N=21
Cohort 3: Hepatocellular Carcinoma, Hepatobiliary, or Pancreatic Adenocarcinoma (N=25)
Single stage (no planned interim analysis) N=25
Safety follow-ups

DLT, dose-limiting toxicity; EGFR, epidermal growth factor receptor; GEJ, gastro-esophageal junction; MTD, maximum tolerated dose; NSCLC, non-small cell lung cancer; PD1, programmed cell death 1; PDL1, programmed death ligand 1; QW, once weekly; Q2W, once every two weeks; RP2D, recommended Phase 2 dose; WT, wild-type.

STUDY PARAMETERS

Key inclusion criteria

- Patients must be aged 18 or over and have histologically or cytologically confirmed advanced or metastatic EGFR disease (in ≥1% of tumor cells):
 - Advanced or metastatic wild-type EGFR non-small cell lung cancer (NSCLC) with progression following ≥1 prior line of therapy, including a platinum-based doublet in combination or following treatment with an anti-PD-1/ or anti-PD-L1 antibody
 - Advanced, unresectable, or metastatic gastric/gastro-esophageal junction (GEJ) adenocarinoma following ≥1 prior chemotherapy regimen including a platinum and fluoropyrimidine doublet
 - Advanced or metastatic hepatocellular carcinoma, hepatobiliary-, or pancreatic adenocarcinoma: following ≥1 prior line of an approved standard of care therapy for the respective disease type or who is ineligible for standard of care therapy
- Adequate hematological, hepatic, and renal function
- Have an Eastern Cooperative Oncology Group (ECOG) Performance Status (PS) 0 or 1
- Evaluable or measurable disease per Response Evaluation Criteria in Solid Tumors (RECIST v1.1)

Key exclusion criteria

- Untreated or symptomatic central nervous system metastases
- Transfusion of blood products within 14 days of AFM24 treatment
- Clinically significant cardiac disorders
- Treatment with systemic anticancer therapy within four weeks of the first dose of study drug, six weeks for mitomycin C or nitrosoureas, two weeks (or five half-lives) for fluorouracil or small molecule targeted drugs, or two weeks for using traditional Chinese medicine with an anti-tumor indication
- Radiation therapy within four weeks before first dose of study drug, or unresolved (common terminology criteria for adverse events v5.0 >Grade 1) toxicity from previous radiotherapy
- History of any other malignancy known to be active
- Currently active in any other clinical study, or administration of another investigational agent

STUDY ENDPOINTS

Phase 1:

Primary:

- Incidence of dose limiting toxicities (DLTs)

Secondary:

- Overall response rate (ORR) as per Response Evaluation Criteria in Solid Tumors (as per RECIST v1.1) by investigator's assessment
- Incidence of treatment-emergent adverse events (TEAEs) and serious adverse events (SAEs)

Phase 2a:

Primary:

- ORR as per RECIST v1.1 by investigator's assessment

Secondary:

- Progression-free survival (PFS), duration of response (DoR), and the clinical benefit rate as per RECIST v1.1 by investigator's assessment

Secondary endpoints for both phases include TEAEs and SAEs, the PK of AFM24 (maximum plasma concentration (C_{max}), minimum plasma concentration (C_{min}), area under the concentration-time curve over the dose interval (AUC_{tau}), time to C_{max} (T_{max}) and immunogenicity.

KEY POINTS

- This study is a Phase 1/2a trial with the aim of establishing the safety, tolerability and efficacy of AFM24 with atezolizumab in patients with EGFR+ solid tumors
- AFM24 and atezolizumab exhibit well-managed safety profiles in patients with solid tumors[4,5]; atezolizumab monotherapy has exhibited efficacious anti-tumor efficacy in both preclinical and clinical settings[6-8] and, in addition, AFM24 has demonstrated the capability of inducing efficacious killing of EGFR+ tumor cell lines *in vitro*[3]
- A combination of AFM24 with atezolizumab may enhance anti-tumor activity by engaging both the innate and adaptive immune responses to target EGFR+ tumors

REFERENCES

1. Nicholson et al. Eur J Cancer 2001,37(Suppl 4):S9-15; 2. Ellwanger et al. mAbs 2019;11:899–918; 3. Wingert et al. mAbs 2021;13(1):1950264; 4. El-Khoueiry et al. 2022. Poster Presented at AACR, New Orleans, Louisiana, USA; 5. Santini FC et al. Expert Rev. Clin. Pharmacol. 2017;10(9):935-45; 6. Wakelee H et al. J Thorac Oncol. 2016;11:S251–S2; 7. Peters S et al. J Clin Oncol. 2017 JCO2016719476; 8. Rittmeyer A et al. Lancet. 2017;389:255–65.

The research was funded by Affimed GmbH, and editorial assistance was provided by Meridian HealthComms Ltd, funded by Affimed. CR, UG, KP, GH, PR, and DME are employees of Affimed GmbH. AEK has received honoraria and has a consulting/advisory role at Roche. Poster presented at the American Society of Clinical Oncology Annual Meeting (ASCO), Chicago, USA. 3–7 June 2022.

For further information, please contact Dr. Daniela Morales-Espinosa (d.morales-espinosa@affimed.com)