Abstract No
2747

CD30/CD16A TandAb AFM13-Induced Target Cell Lysis By NK-Cells Is Enhanced By CD137 Co-Stimulation And Blocking PD-1

Xing Zhao[1], Narendiran Rajasekaran[1], Uwe Reusch[2], Jens-Peter Marschner[2], Martin Treder[2#], Holbrook Edwin Kohrt[1]
[1]Center for Clinical Sciences Research Stanford, Stanford, CA 94305-5151; [2]Affimed GmbH, Im Neuenheimer Feld 582, 69120 Heidelberg, Germany

Background

The CD30/CD16A bi-specific tetravalent TandAb antibody AFM13 recruits and activates NK-cells by specific binding to CD16A for targeted lysis of CD30⁺ tumor cells[1]. Given promising clinical activity and safety profile of AFM13 and proof-of-mechanism demonstrating dependence on the immune response[2], potential synergy of AFM13 and checkpoint inhibitors was evaluated.

In vitro cytotoxicity assays demonstrated higher potency and efficacy of AFM13 relative to other anti-CD30 antibody formats (CD30/CD16A diabodies, anti-CD30 IgGs)[1]. High potency and efficacy was observed on different $CD30^+$ tumor target cell lines using PBMC or enriched NK-cells as effector cells. To evaluate a potential synergy of AFM13 and checkpoint inhibitors, chromium release assays with effector cells that were cultured for 24 hours together with anti-CD30 and irradiated $CD30^+$ lymphoma tumor cells were performed. Pre-cultured PBMC or enriched NK-cells were used as effector cells at various E:T ratios in 4 hour chromium release assay with ^{51}Cr-labeled $CD30^+$ lymphoma cells in media alone, with AFM13, with checkpoint inhibitors, or with AFM13 in combination with checkpoint inhibitors. Single treatment with AFM13 at suboptimal concentrations (1 pM) induced effector-to-target cell-dependent lysis of $CD30^+$ lymphoma cells up to 40% using enriched NK cells. Immune-modulating antibodies alone mediated substantially lower lysis (<25%). However, the addition of anti-PD-1 or anti-CD137 to AFM13 strongly enhanced specific lysis up to 70%, whereas the addition of anti-CTLA-4 to AFM13 showed no beneficial effect. The most impressive increase of efficacy was observed when AFM13 was applied together with a combination of anti-PD-1 and anti-CD137.

The promising *in vitro* results of the combination of AFM13 with checkpoint inhibitors and anti-CD137 co-stimulation prompted us to assess potential synergy of AFM13 with checkpoint inhibitors in an *in vivo* PDX model.

Methods

Xenografted tumor fragments (8x8mm) derived from a surgical specimen of a newly diagnosed patient with $CD30^+$ lymphoma (including Hodgkin Disease), in Rag2$^{-/-}$IL2Rγ^{null} mice were observed for engraftment and randomized into groups on day 28. Autologous PBMCs were infused on day 28 ($2x10^6$ PBMCs/mouse) intra-peritoneally. Antibody therapy with AFM13 and anti-CTLA-4, anti-CD137, or anti-PD-1 began on day 28 and was continued weekly for a total of three intra-peritoneal injections. Anti-CTLA-4 (Yervoy), anti-CD137 (Urelumab) or anti-PD-1 (Keytruda) were always dosed one day after AFM13. An irrelevant IgG and an irrelevant CD16A-recruiting TandAb (AFM22) were use as a control. At the indicated time points, tumor volumes were determined, and mice were sacrificed for the assessment of tumor-infiltrating lymphocytes and intra-tumoral cytokines.

Results

The efficacy of AFM13 was augmented by each CPI (anti-CTLA-4, anti-PD-1, or anti-CD137) tested, but most impressive and obvious with anti-PD-1. The enhanced antitumor activity of AFM13 in combination with immune-modulating antibodies was associated with higher numbers of tumor-infiltrating NK- and T-cells and augmented release of pro-inflammatory cytokines. Treatment with control IgG or irrelevant CD16A-recruiting TandAbs did not induce an unspecific immune cell activation supporting strict target-dependent NK-cell activation by AFM13.

Conclusion

Engagement of NK-cells using CD30/CD16A TandAbs by binding to CD16A does not completely exploit the efficacy of NK-cells. Combination of AFM13 with immune-modulating anti-CTLA-4, anti-CD137, and anti-PD-1 antibodies not only enhanced the anti-tumor activity of NK-cells but also stimulated infiltration of T-cells and cytokine release in the tumors supporting cross-talk between innate and adaptive immunity. Therefore combination trials with companion intra-tumoral assessment may personalize dual antibody therapy and augment the efficacy of AFM13 and CPIs.

CD30/CD16A TandAb structure and mechanism of action

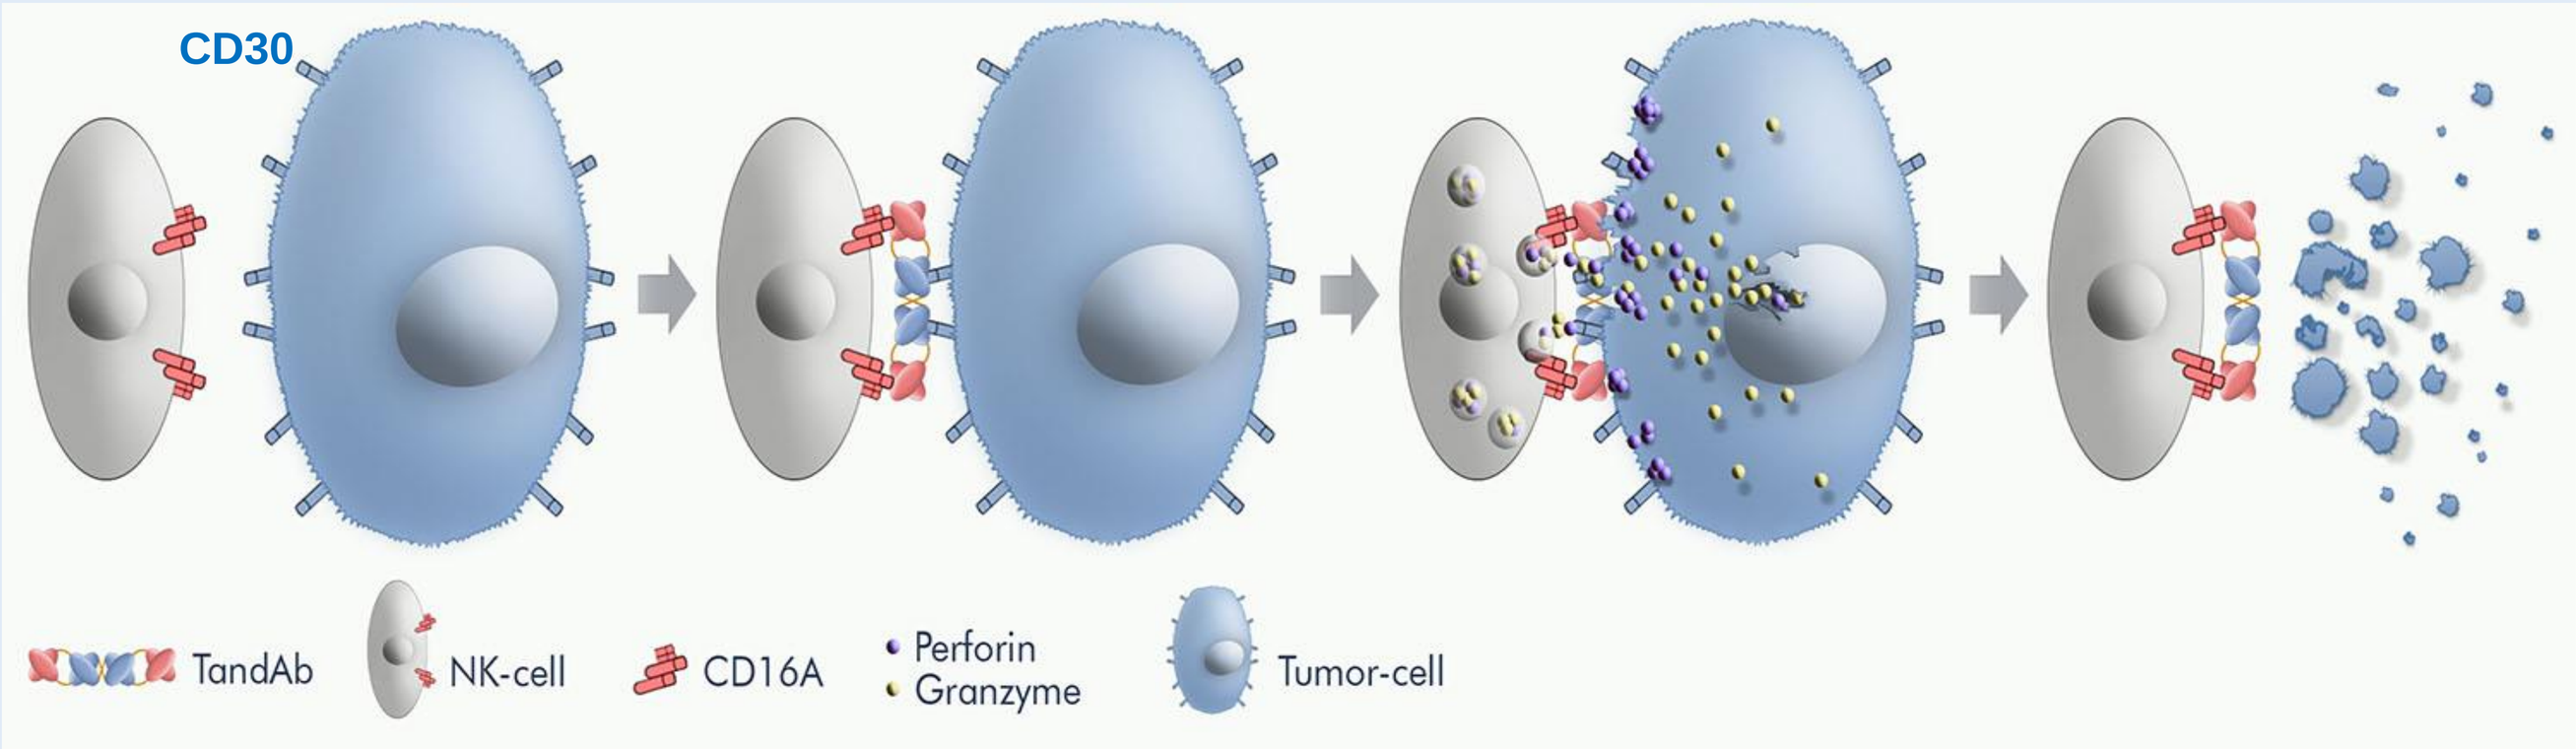
CD30
TandAb
NK-cell
CD16A
Perforin
Granzyme
Tumor-cell

Stage 1
NK-cell with CD16A and tumor cell with CD30 receptors

Stage 2
TandAb locks NK-cell and tumor cell in close proximity and activates NK-cell

Stage 3
NK-cell releases perforin, creating pores in tumor cell membrane for granzyme entry

Stage 4
Granzyme and caspase action trigger apoptosis of tumor cell

Establishment of a Hodgkin Lymphoma PDX model

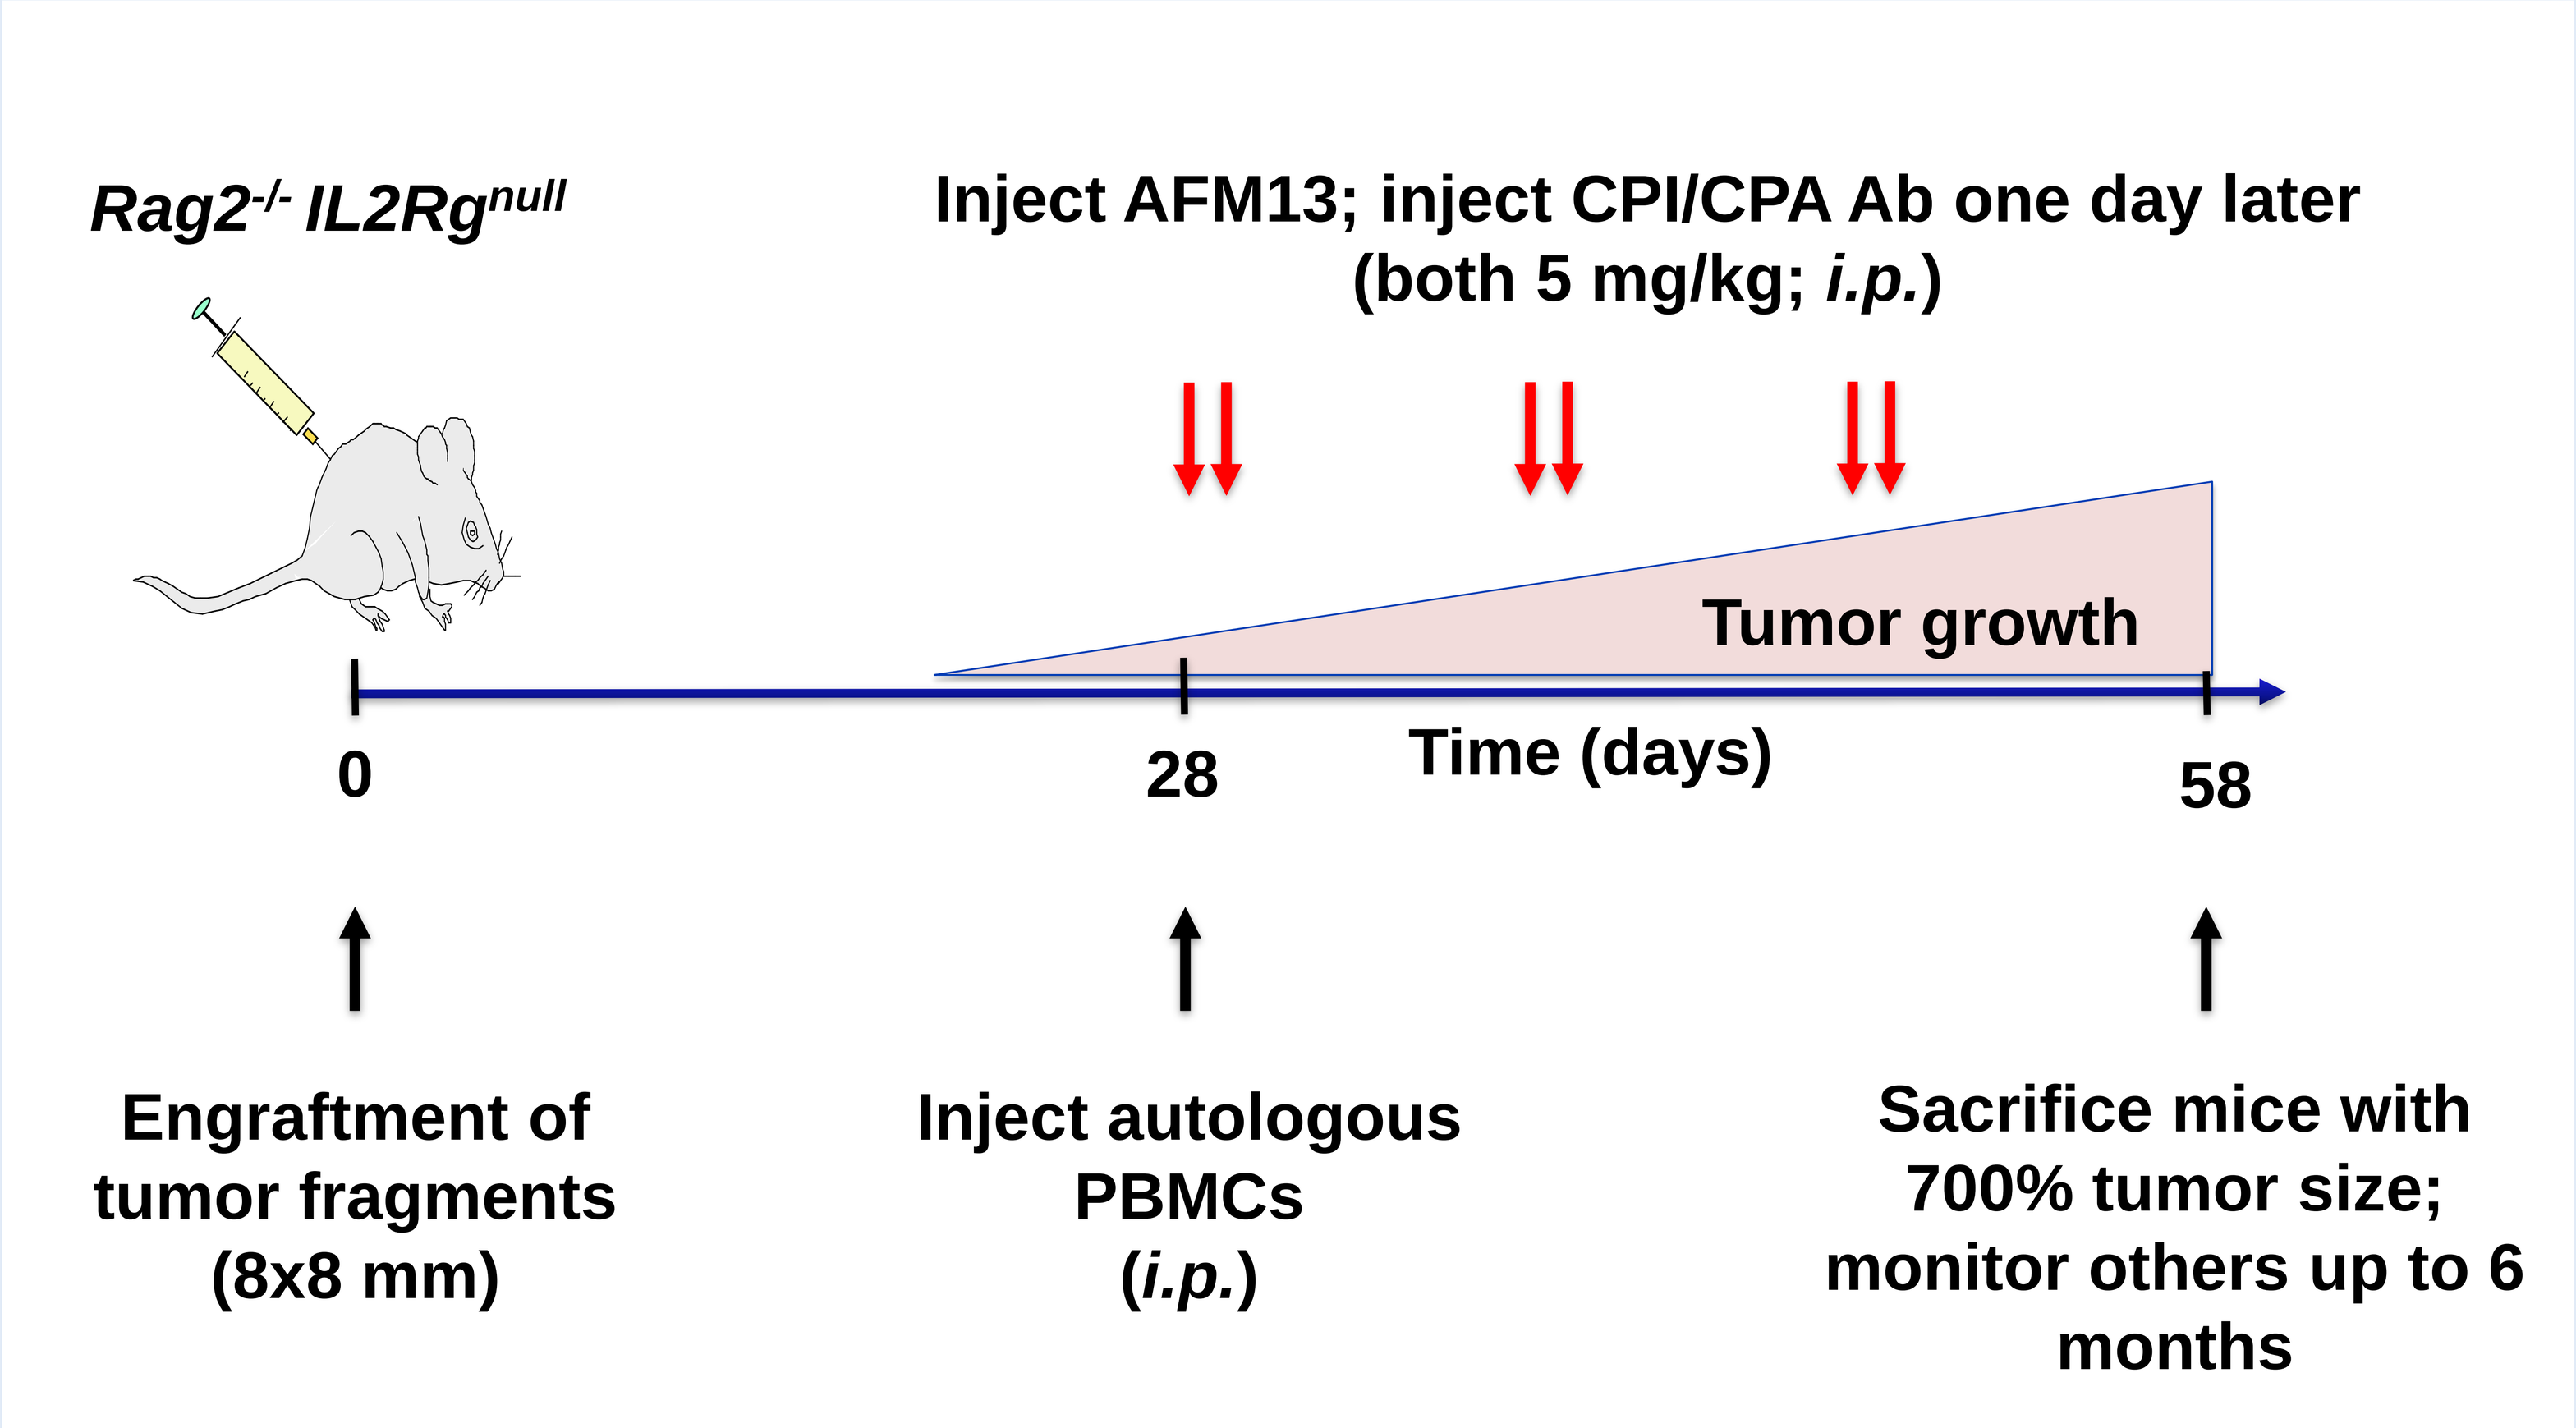
Rag2-/- IL2Rgnull
Inject AFM13; inject CPI/CPA Ab one day later
(both 5 mg/kg; i.p.)
Tumor growth
0
28
Time (days)
58
Engraftment of tumor fragments (8x8 mm)
Inject autologous PBMCs (i.p.)
Sacrifice mice with 700% tumor size; monitor others up to 6 months

- Therapy with AFM13 and anti-CTLA-4, anti-CD137, or anti-PD-1 began on day 28 and continued weekly for a total of three intraperitoneal injections.
- As a control, mice were treated with irrelevant IgG or an irrelevant TandAb (AFM22).
- Tumor volumes, tumor infiltrating lymphocytes and cytokines were assessed at the indicated time points.

AFM13 combined with CD137 co-stimulation or blocking PD-1 results in PDX tumor regression

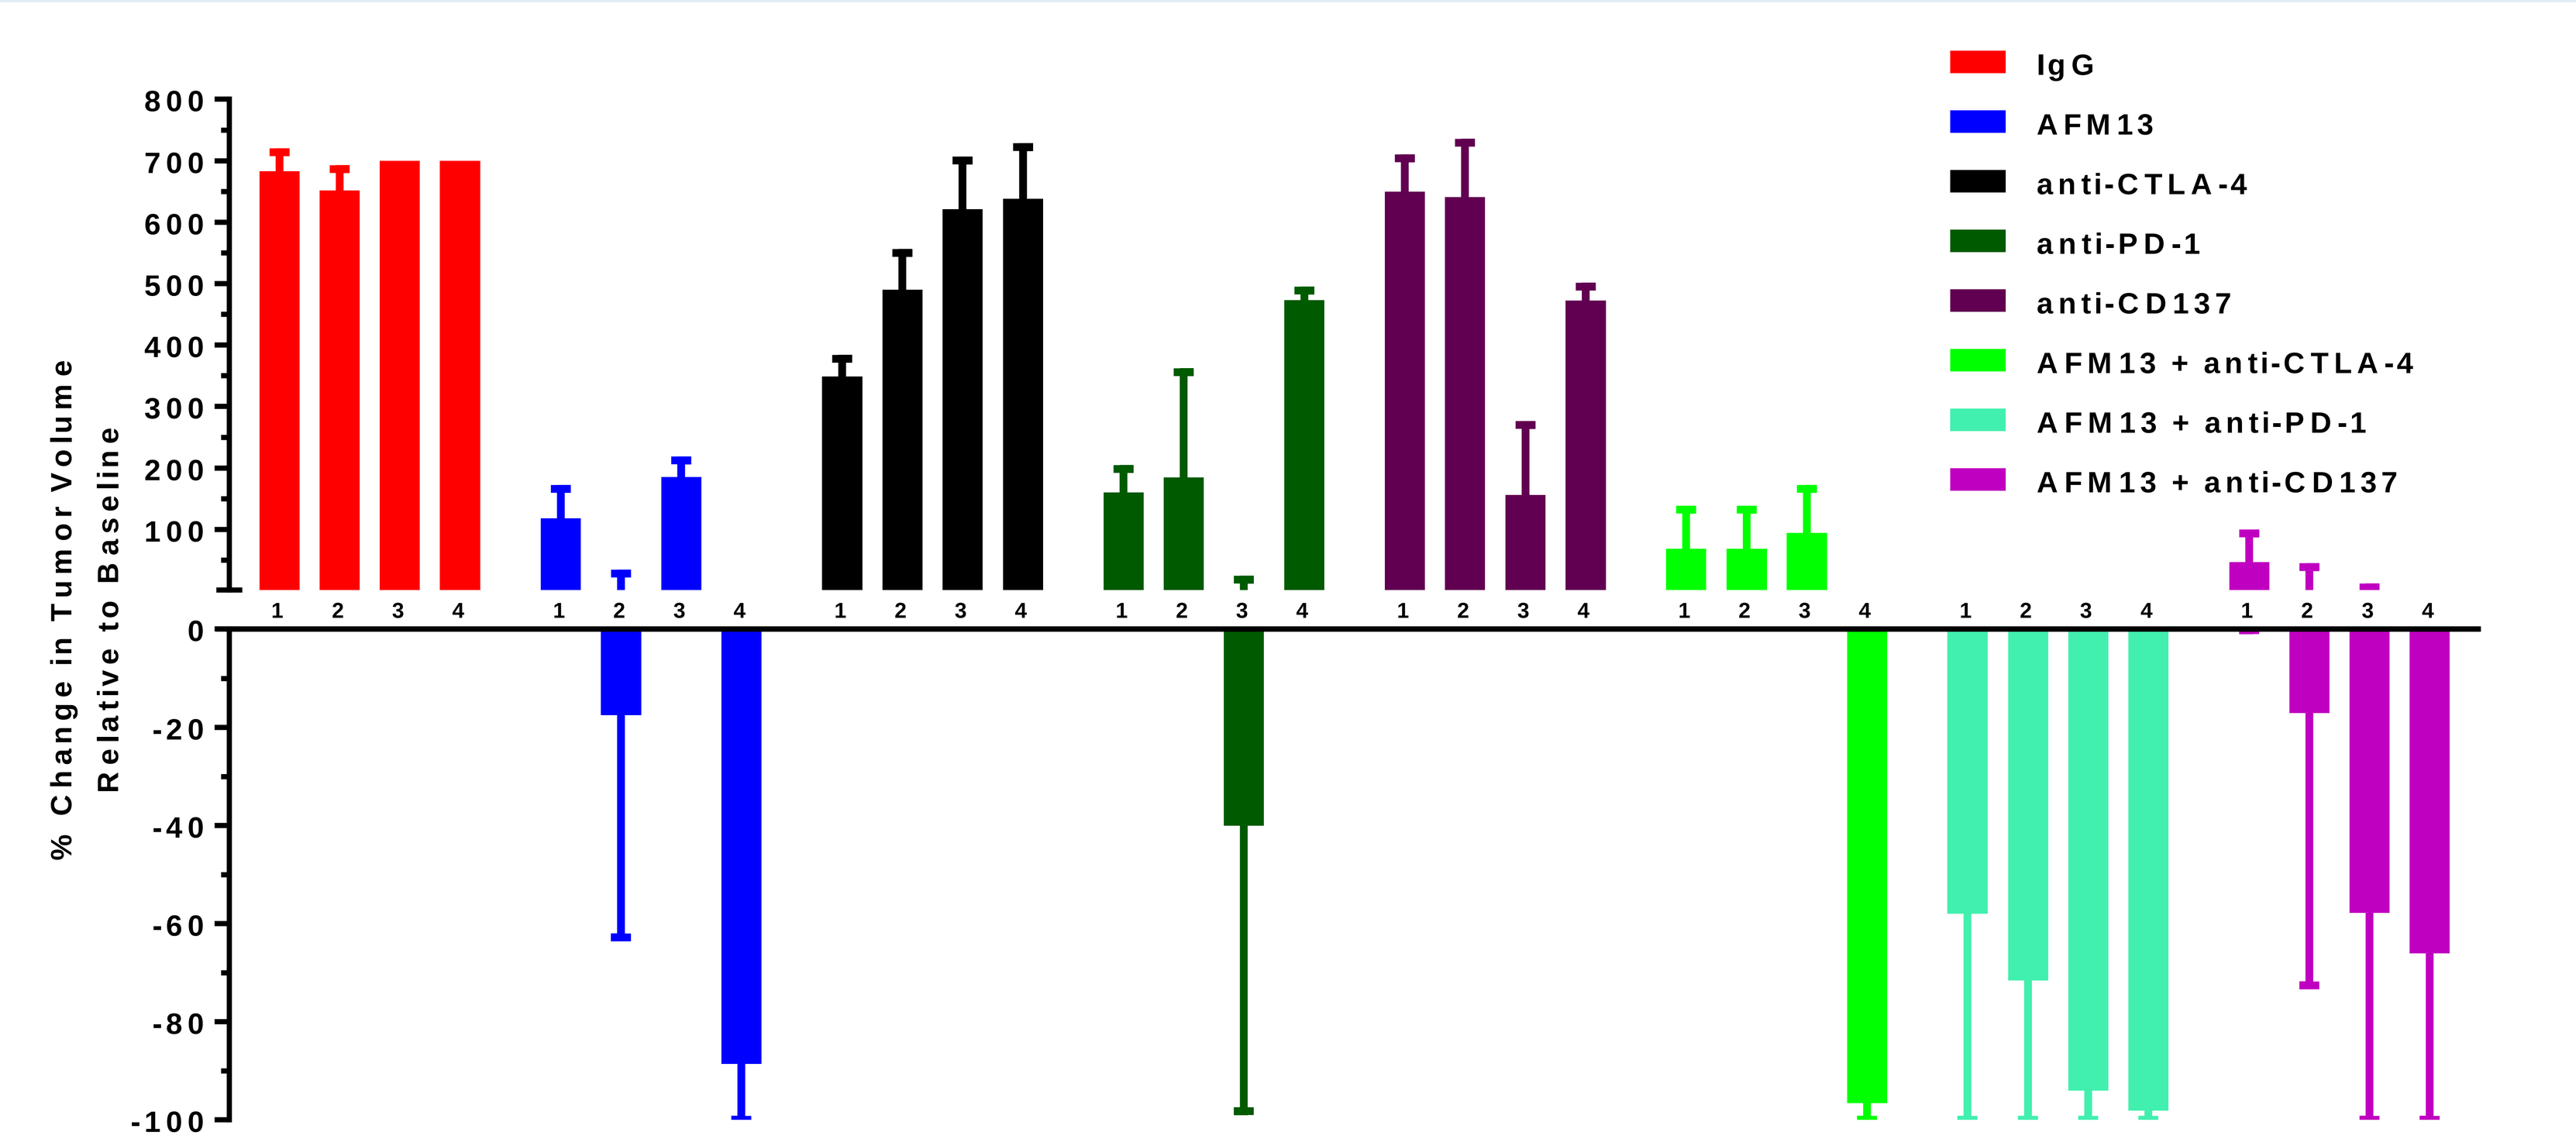
% Change in Tumor Volume Relative to Baseline
IgG
AFM13
anti-CTLA-4
anti-PD-1
anti-CD137
AFM13 + anti-CTLA-4
AFM13 + anti-PD-1
AFM13 + anti-CD137

- AFM13 reduced tumor growth in all four independent HL PDX studies (1-4).
- The effect of the immune-modulating antibodies on tumor growth was weaker than AFM13 and strongest for anti-PD-1.
- AFM13 efficacy was augmented by each combination tested, but most impressive in all four independent studies with anti-PD-1.

AFM13 in combination with anti-PD-1 reduces PDX tumor growth

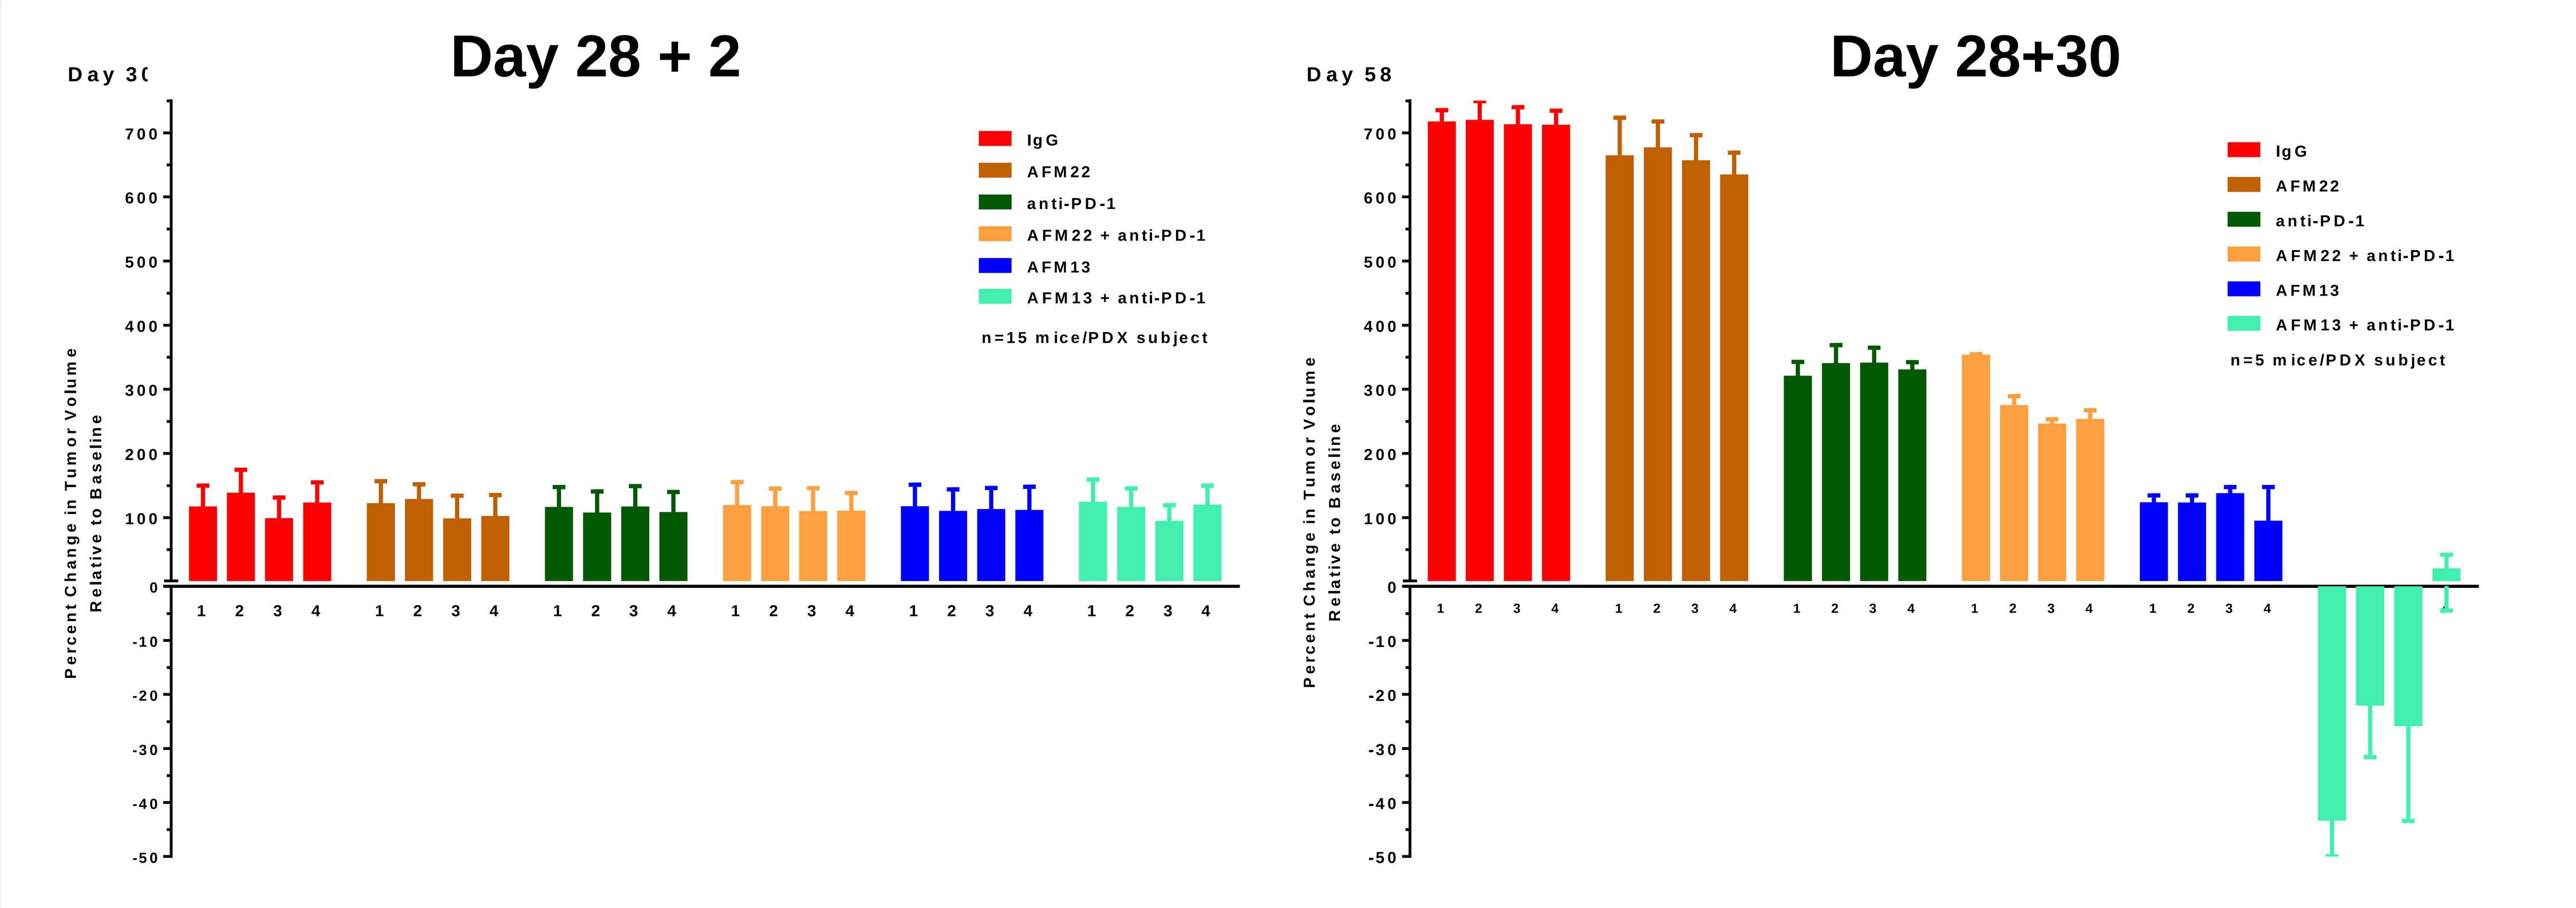
Day 28 + 2
Day 28+30
Day 58
Percent Change in Tumor Volume Relative to Baseline
IgG
AFM22
anti-PD-1
AFM22 + anti-PD-1
AFM13
AFM13 + anti-PD-1
n=15 mice/PDX subject
n=5 mice/PDX subject

- Tumor growth control was investigated in four independent *in vivo* PDX studies on day 30 and day 58.
- AFM13 reduced tumor growth in all four PDX experiments (1-4) on day 58.
- The inhibitory effect on tumor growth of the anti-PD-1 antibody alone was weaker than that of AFM13 alone, but augmented AFM13 efficacy.
- IgG and an irrelevant CD16A TandAb (AFM22) did not affect tumor growth.
- In 3/4 cases, the combination of AFM13 with anti-PD-1 induced tumor shrinkage.

Combination of AFM13 with anti-PD-1 enhances cross-talk between NK- and T-cells

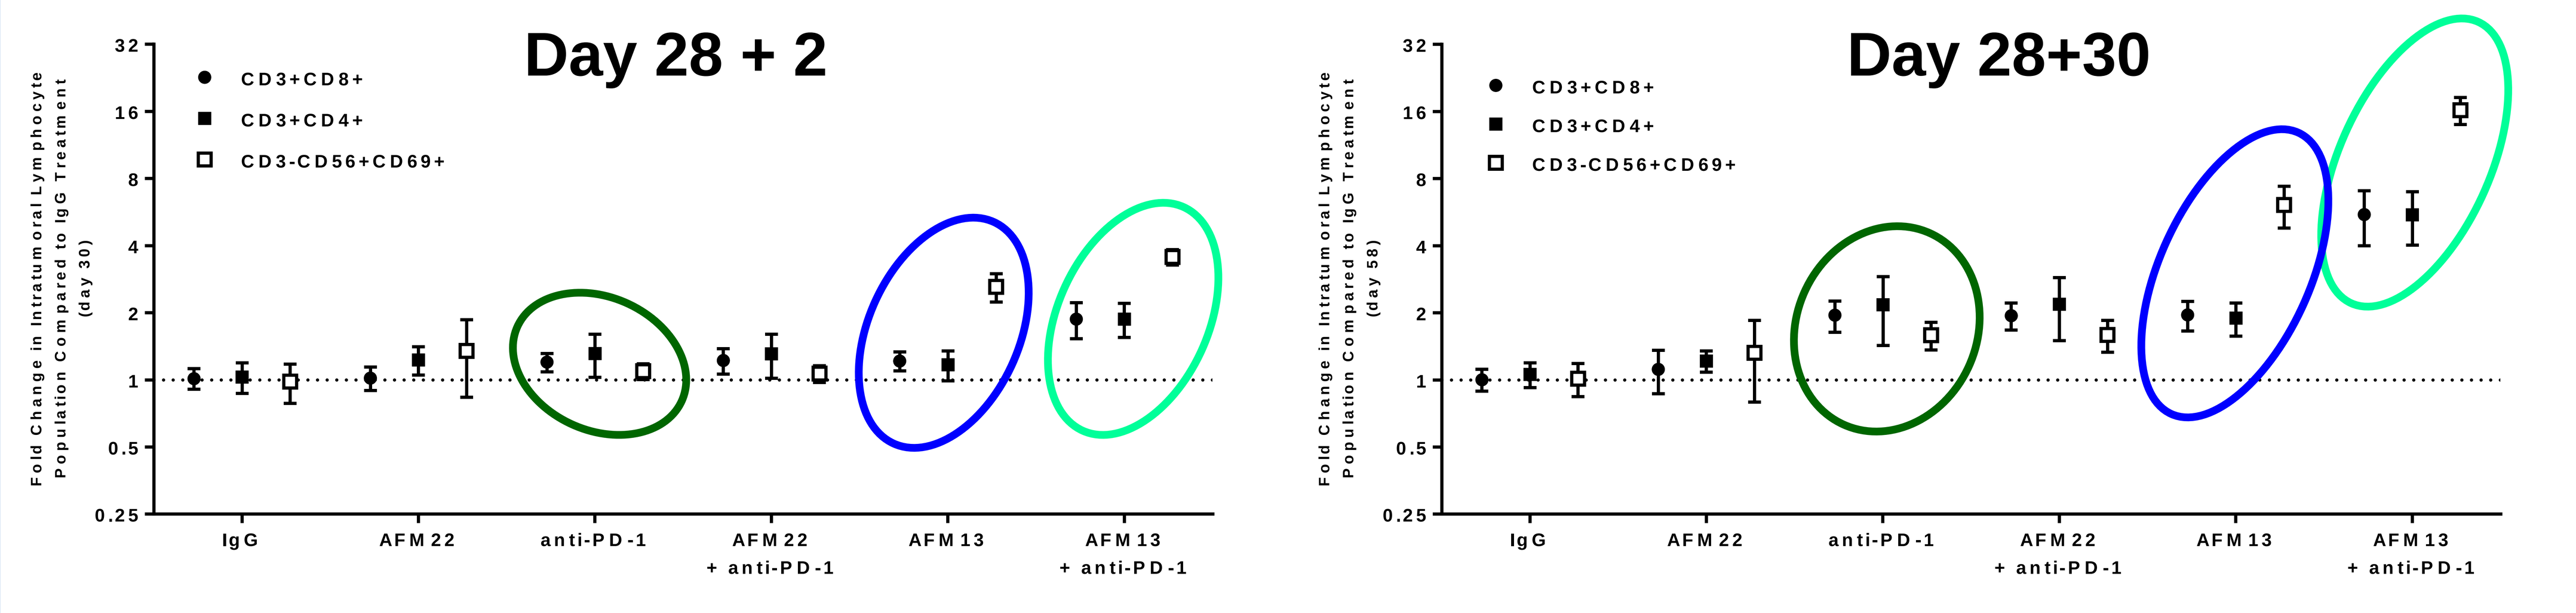
Day 28 + 2
Day 28+30
Fold Change in Intratumoral Lymphocyte Population Compared to IgG Treatment (day 30)
Fold Change in Intratumoral Lymphocyte Population Compared to IgG Treatment (day 58)
CD3+CD8+
CD3+CD4+
CD3-CD56+CD69+
IgG
AFM22
anti-PD-1
AFM22 + anti-PD-1
AFM13
AFM13 + anti-PD-1

- Treatment with AFM13 alone initially stimulates NK-cell infiltration into tumors, after 58 days infiltration of both NK-cells and T-cells.
- Combining AFM13 with anti-PD-1 potentiates not only infiltration of NK-cells ($CD3^-CD56^+CD69^+$), but also migration of $CD4^+$ & $CD8^+$ T-cells into tumors.
- These findings support AFM13-stimulated cross-talk between innate and adaptive immunity that is further augmented by anti-PD-1.
- An irrelevant CD16A-recruiting TandAb (AFM22) and/or anti-PD-1 did not substantially enhance the number of infiltrating T- or NK-cells.

Intra-tumoral cytokine assessment on day 58

- Treatment with AFM13 or anti-PD-1 induces intra-tumoral release of IFN-γ, TNF-α and IL-2.
- The combination therapy with AFM13 and anti-PD-1 increases the release.
- Treatment with irrelevant CD16A TandAb (AFM22) did not induce substantial intra-tumoral cytokine release

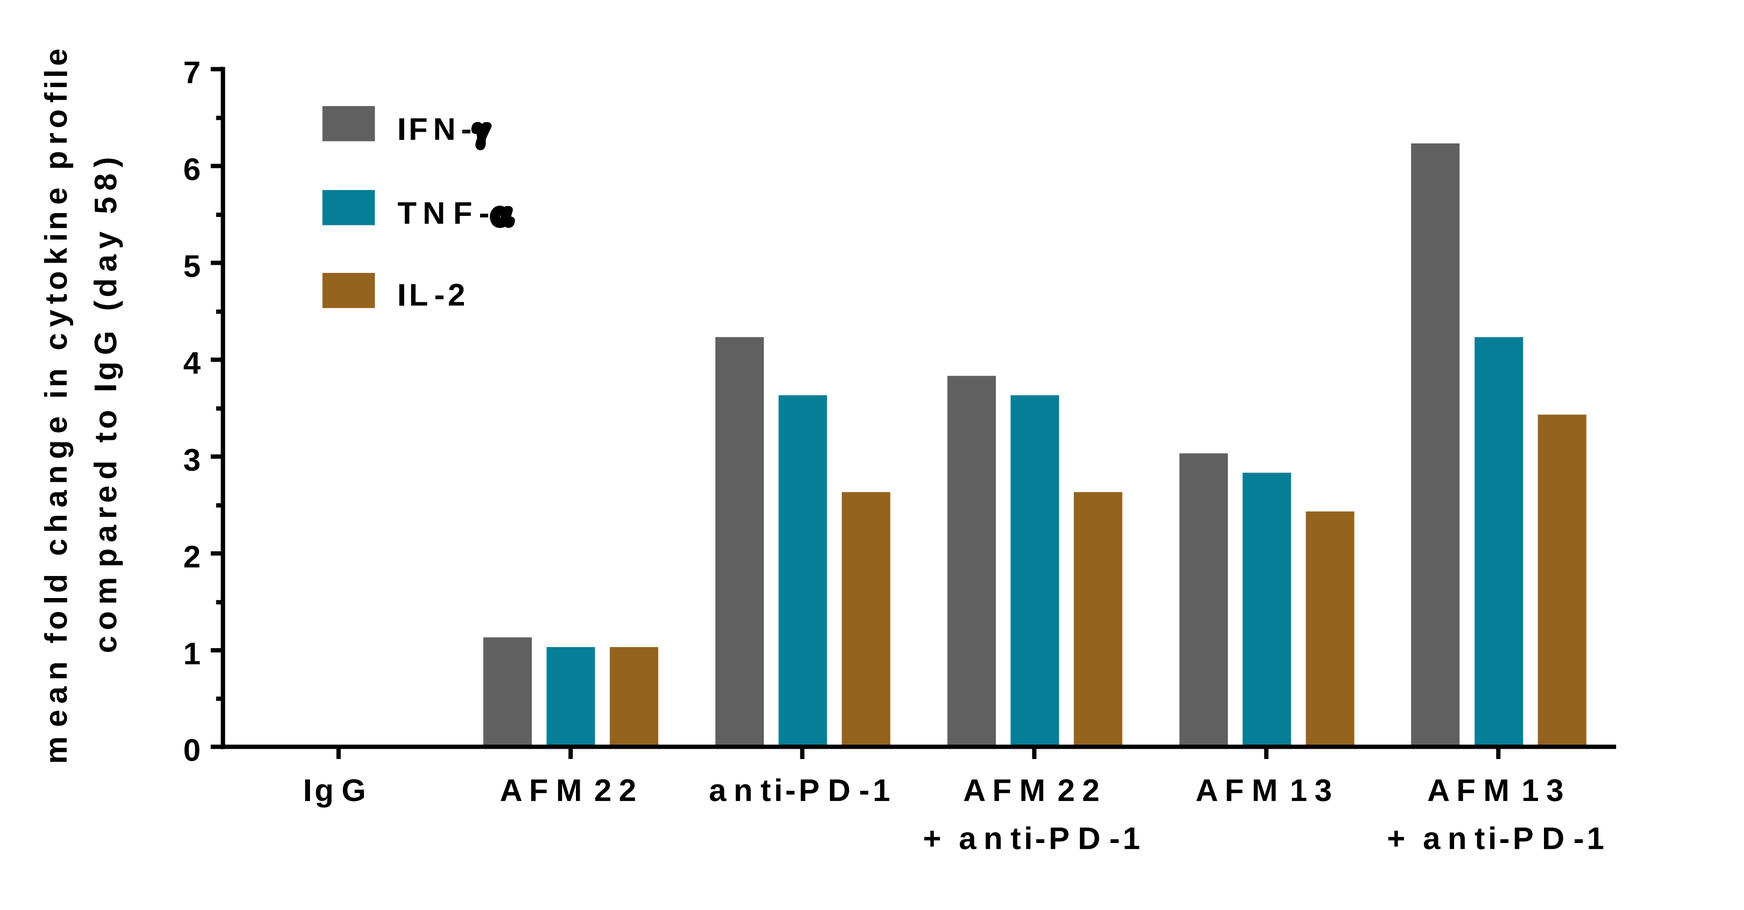
mean fold change in cytokine profile compared to IgG (day 58)
IFN-γ
TNF-α
IL-2
IgG
AFM22
anti-PD-1
AFM22 + anti-PD-1
AFM13
AFM13 + anti-PD-1

Corresponding author

References: 1. Reusch, U. *et al.* A novel tetravalent bispecific TandAb (CD30/CD16A) efficiently recruits NK cells for the lysis of CD30⁺ tumor cells. Mabs 6(3): 728–739 (2014).
2. Rothe, A. *et al.* A phase I study of the bispecific anti-CD30/CD16A antibody construct AFM13 in patients with relapsed or refractory Hodgkin lymphoma. Blood 125, 4024–4032 (2015).