Abstract No
2763

STANFORD SCHOOL OF MEDICINE
Stanford University Medical Center

In Vitro and *In Vivo* Characterization of CD19/CD3 Tandab AFM11 and CD19/CD16A Tandab AFM12 Targeting NHL and ALL

Xing Zhao[1], Narendiran Rajasekaran[1], Uwe Reusch[2], Michael Weichel[2], Kristina Ellwanger[2], Jens-Peter Marschner[2], Martin Treder[2#], Holbrook Edwin Kohrt[1]

[1]Center for Clinical Sciences Research Stanford, Stanford, CA 94305-5151; [2]Affimed GmbH, Im Neuenheimer Feld 582, 69120 Heidelberg, Germany

affimed

Background

CD19 is expressed by B cells from early development through differentiation into plasma cells, and represents a validated target for the development of therapeutic antibodies to treat B cell malignancies such as Non-Hodgkin-Lymphoma (NHL) and acute lymphoblastic leukemia (ALL). Different CD19-targeting T-cell engagers are investigated in clinical studies for the treatment of NHL or ALL, including Affimed's AFM11[1], a bi-specific CD19/CD3 TandAb antibody, which is currently investigated in a phase I dose escalation study. Indeed, Affimed's bi-specific tetravalent platform comprises not only T-cell engaging TandAbs with two binding sites for CD3, but also NK-cell recruiting TandAbs with two binding sites for CD16A[2]. In the present study, Affimed's AFM11, was characterized and compared in vitro and in vivo with the CD19/CD16A TandAb AFM12.

Methods

Analogous to the CD19/CD3 TandAb AFM11, a bi-specific tetravalent TandAb AFM12 was constructed with two binding sites for CD19 and two sites for CD16A. Both TandAbs were characterized for their biophysical properties, binding affinities to CD19+ tumor target cells and to their respective effector cells by flow cytometry. Kinetics and dose-response characteristics were evaluated in in vitro cytotoxicity assays. Potency and efficacy of both TandAbs were compared on different CD19+ tumor target cell lines using primary human effector cells. To compare the efficacy of AFM11 and AFM12 a patient-derived tumor xenograft model was developed.

Results

AFM12 mediated efficacious target cell lysis with a very fast on-set in vitro. Lysis induced by AFM11 was less efficacious (lower specific lysis than AFM12) but reproducibly more potent (lower EC50 value). In addition to the potency and efficacy of AFM11 and AFM12, different aspects of safety, such as effector cell activation in the presence and absence of target cells were investigated and will be described.

Conclusion

Affimed's CD19/CD3 and CD19/CD16A TandAbs with identical anti-CD19 tumor-targeting domains but different effector cell-recruiting domains represent interesting molecules to study T-cell- or NK-cell-based immunotherapeutic approaches. The comparison of AFM11 and AFM12 demonstrated that AFM12-mediated lysis was fast and efficacious, and AFM11 showed higher potency. In summary, the NK-cell recruiting TandAb AFM12 represents an alternative to T-cell recruiting molecules, as it may offer a different side effect profile, comparable to that of AFM13, the first NK-cell TandAb in clinical development.

Molecular Structure

AFM11

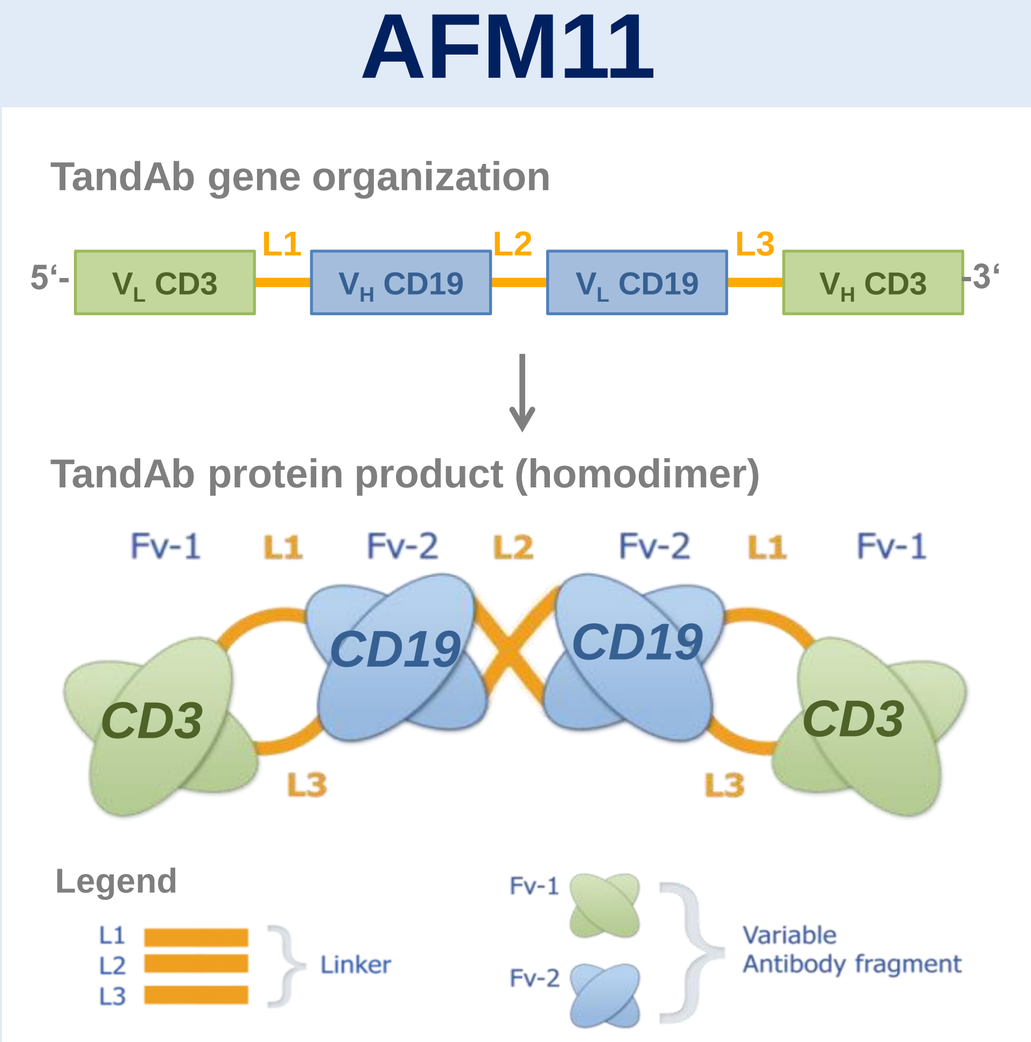
TandAb gene organization
5'- V_L CD3 L1 V_H CD19 L2 V_L CD19 L3 V_H CD3 -3'
TandAb protein product (homodimer)
Fv-1 L1 Fv-2 L2 Fv-2 L1 Fv-1
CD3 CD19 CD19 CD3
L3 L3
Legend
L1 L2 L3 Linker
Fv-1 Fv-2 Variable Antibody fragment

AFM12

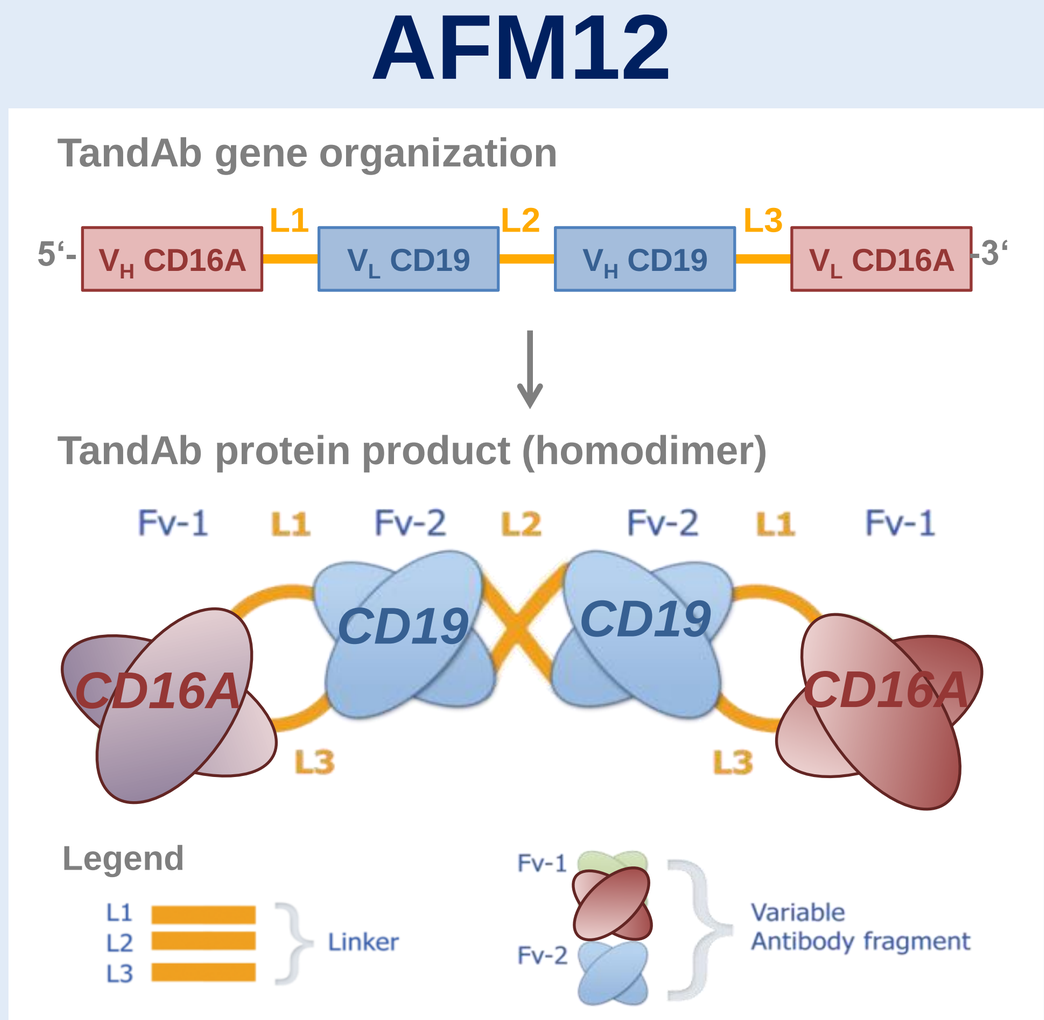
TandAb gene organization
5'- V_H CD16A L1 V_L CD19 L2 V_H CD19 L3 V_L CD16A -3'
TandAb protein product (homodimer)
Fv-1 L1 Fv-2 L2 Fv-2 L1 Fv-1
CD16A CD19 CD19 CD16A
L3 L3
Legend
L1 L2 L3 Linker
Fv-1 Fv-2 Variable Antibody fragment

Biophysical characterization of AFM11 and AFM12

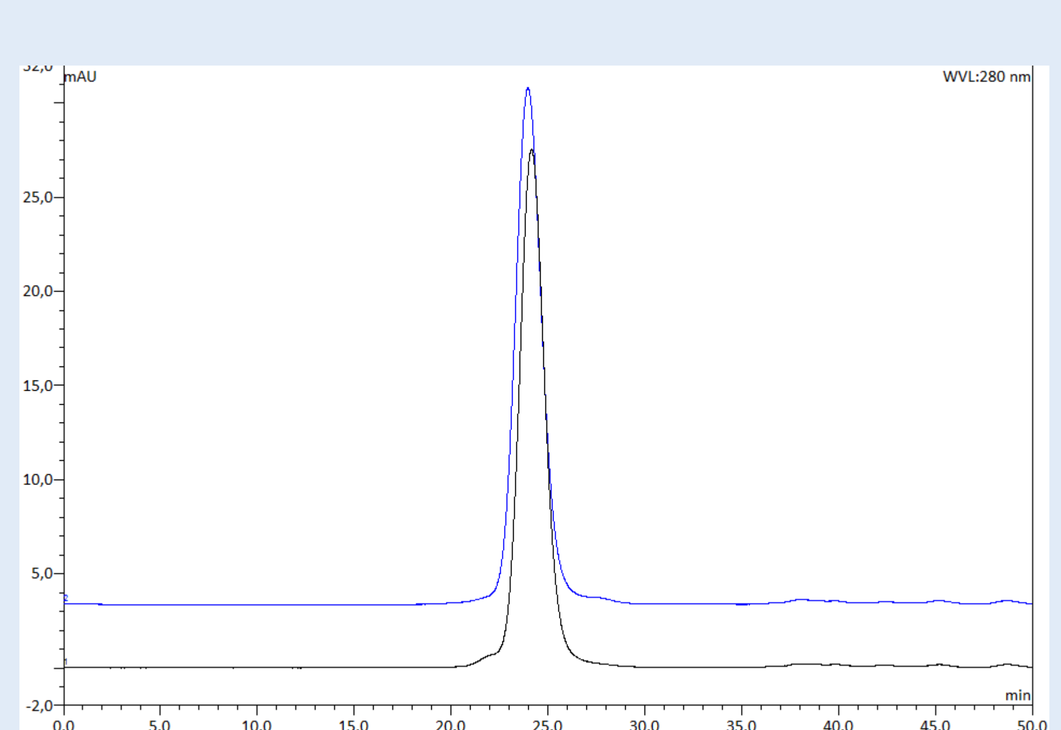

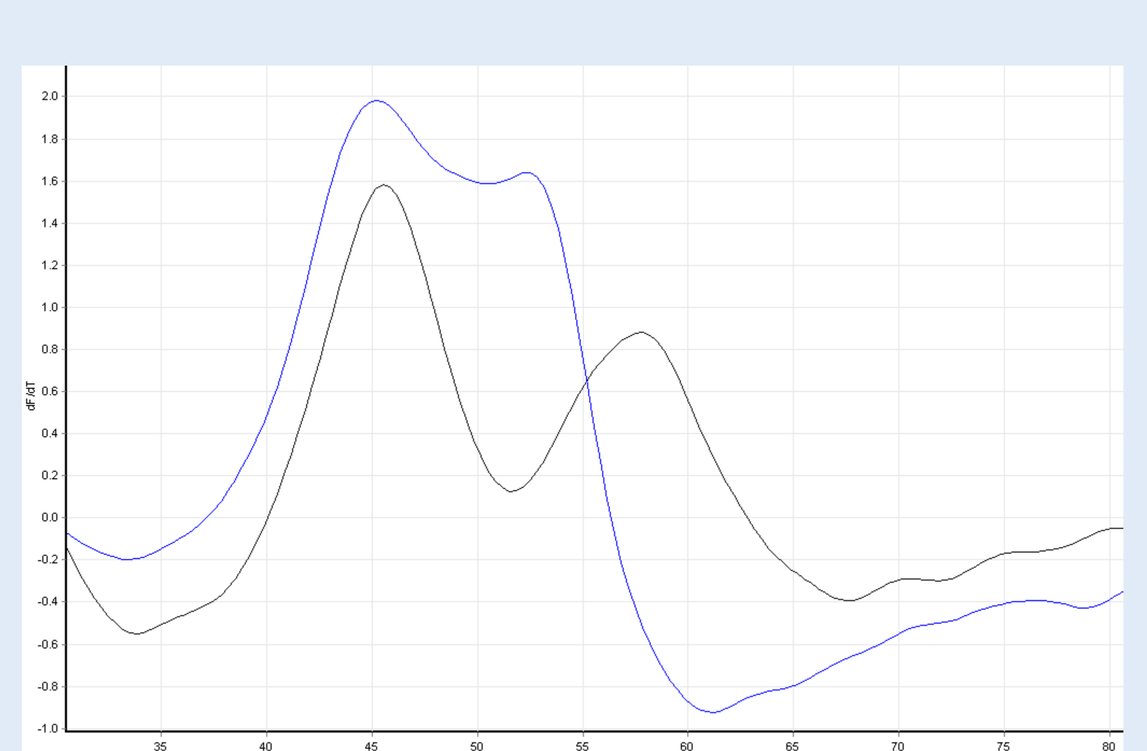

- Left: comparable dimer contents of ≥95% (SE-HPLC) achieved by two-step process.
- Right: both TandAbs exhibit comparable primary thermal transitions at approx. 45 °C (DSF).

In vitro binding to effector and target cells

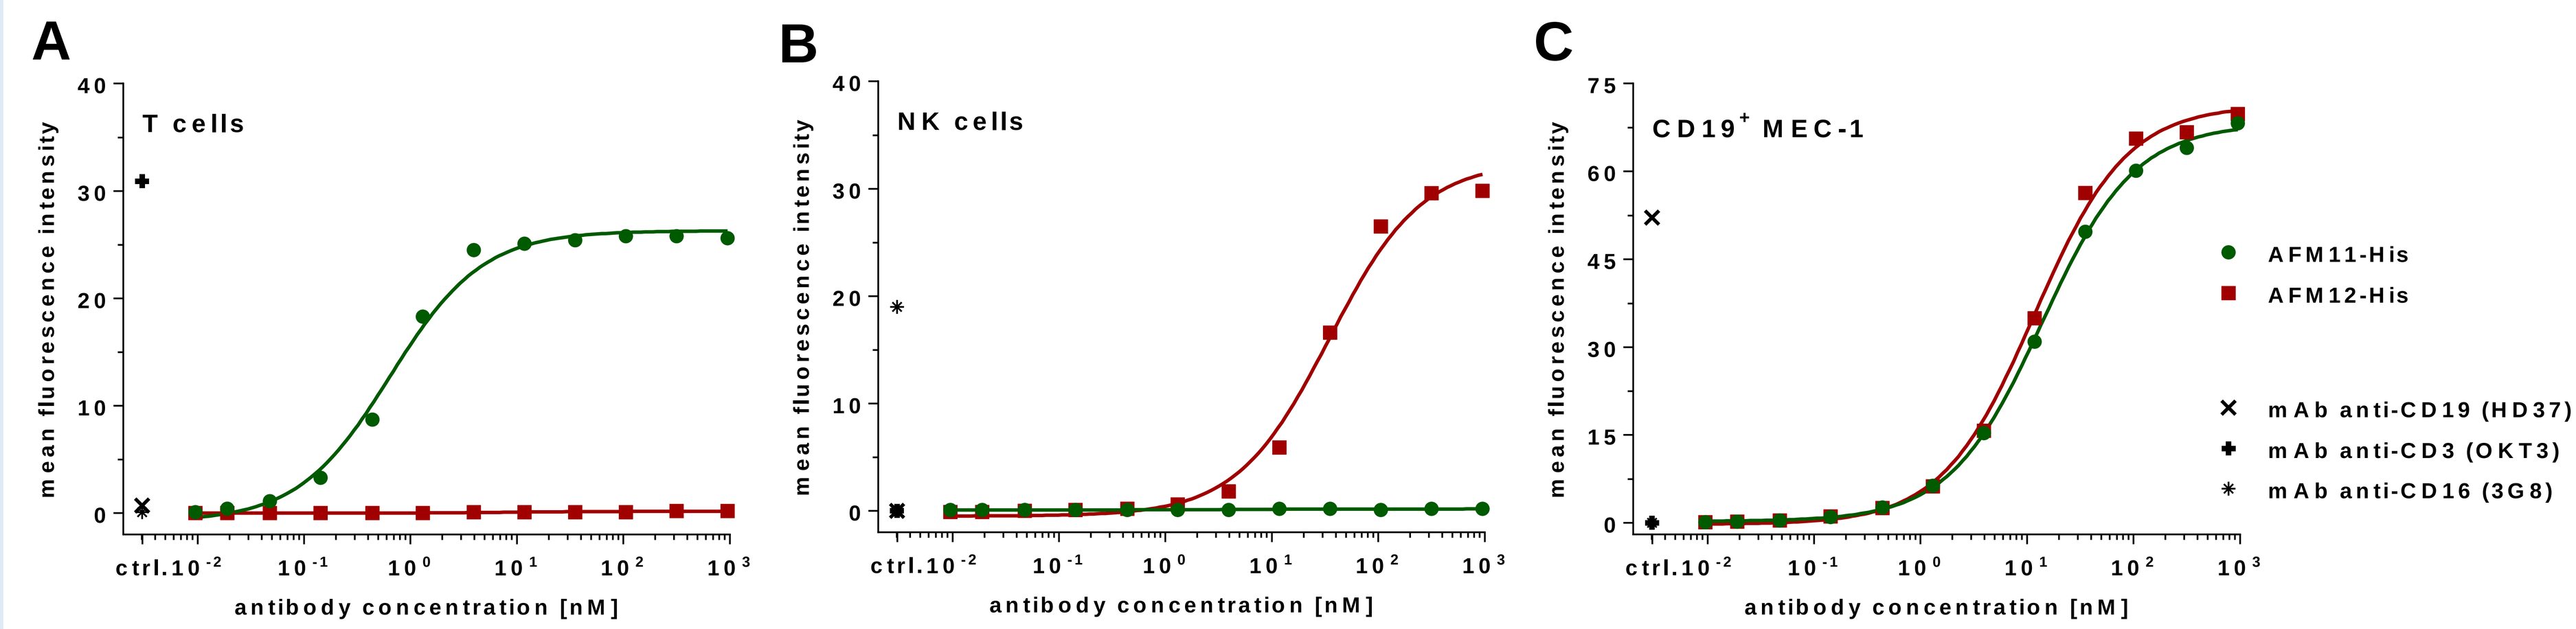
A
T cells
B
NK cells
C
CD19+ MEC-1
mean fluorescence intensity
antibody concentration [nM]
AFM11-His
AFM12-His
mAb anti-CD19 (HD37)
mAb anti-CD3 (OKT3)
mAb anti-CD16 (3G8)

Apparent affinities were determined by incubation of serial dilutions of His-tagged AFM11 and AFM12 with T-cells (**A**), NK-cells (**B**), and CD19⁺ MEC-1 cells (**C**). Cell-bound TandAbs were detected by staining with mAb anti-His followed by FITC-conjugated goat anti-mouse IgG and subsequent flow cytometric analysis. Mean fluorescence intensities were used to calculate K_D values by non-linear regression (hyperbolic function). Values determined in the presented experiment are indicated.

K_D value	primary T-cells	primary NK-cells	MEC-1	Raji	NALM-6
AFM11-His	0.7 nM	no binding	14 nM	18 nM	4 nM
AFM12-His	no binding	36 nM	12 nM	14 nM	5 nM

- High affinity binding of AFM11 to CD3⁺ T-cells in the sub-nanomolar range
- High affinity binding of AFM12 to CD16A⁺ NK-cells in the nanomolar range
- Similar binding affinity of AFM11 and AFM12 to CD19⁺ tumor cell lines

In vitro potency and efficacy in cytotoxicity assays

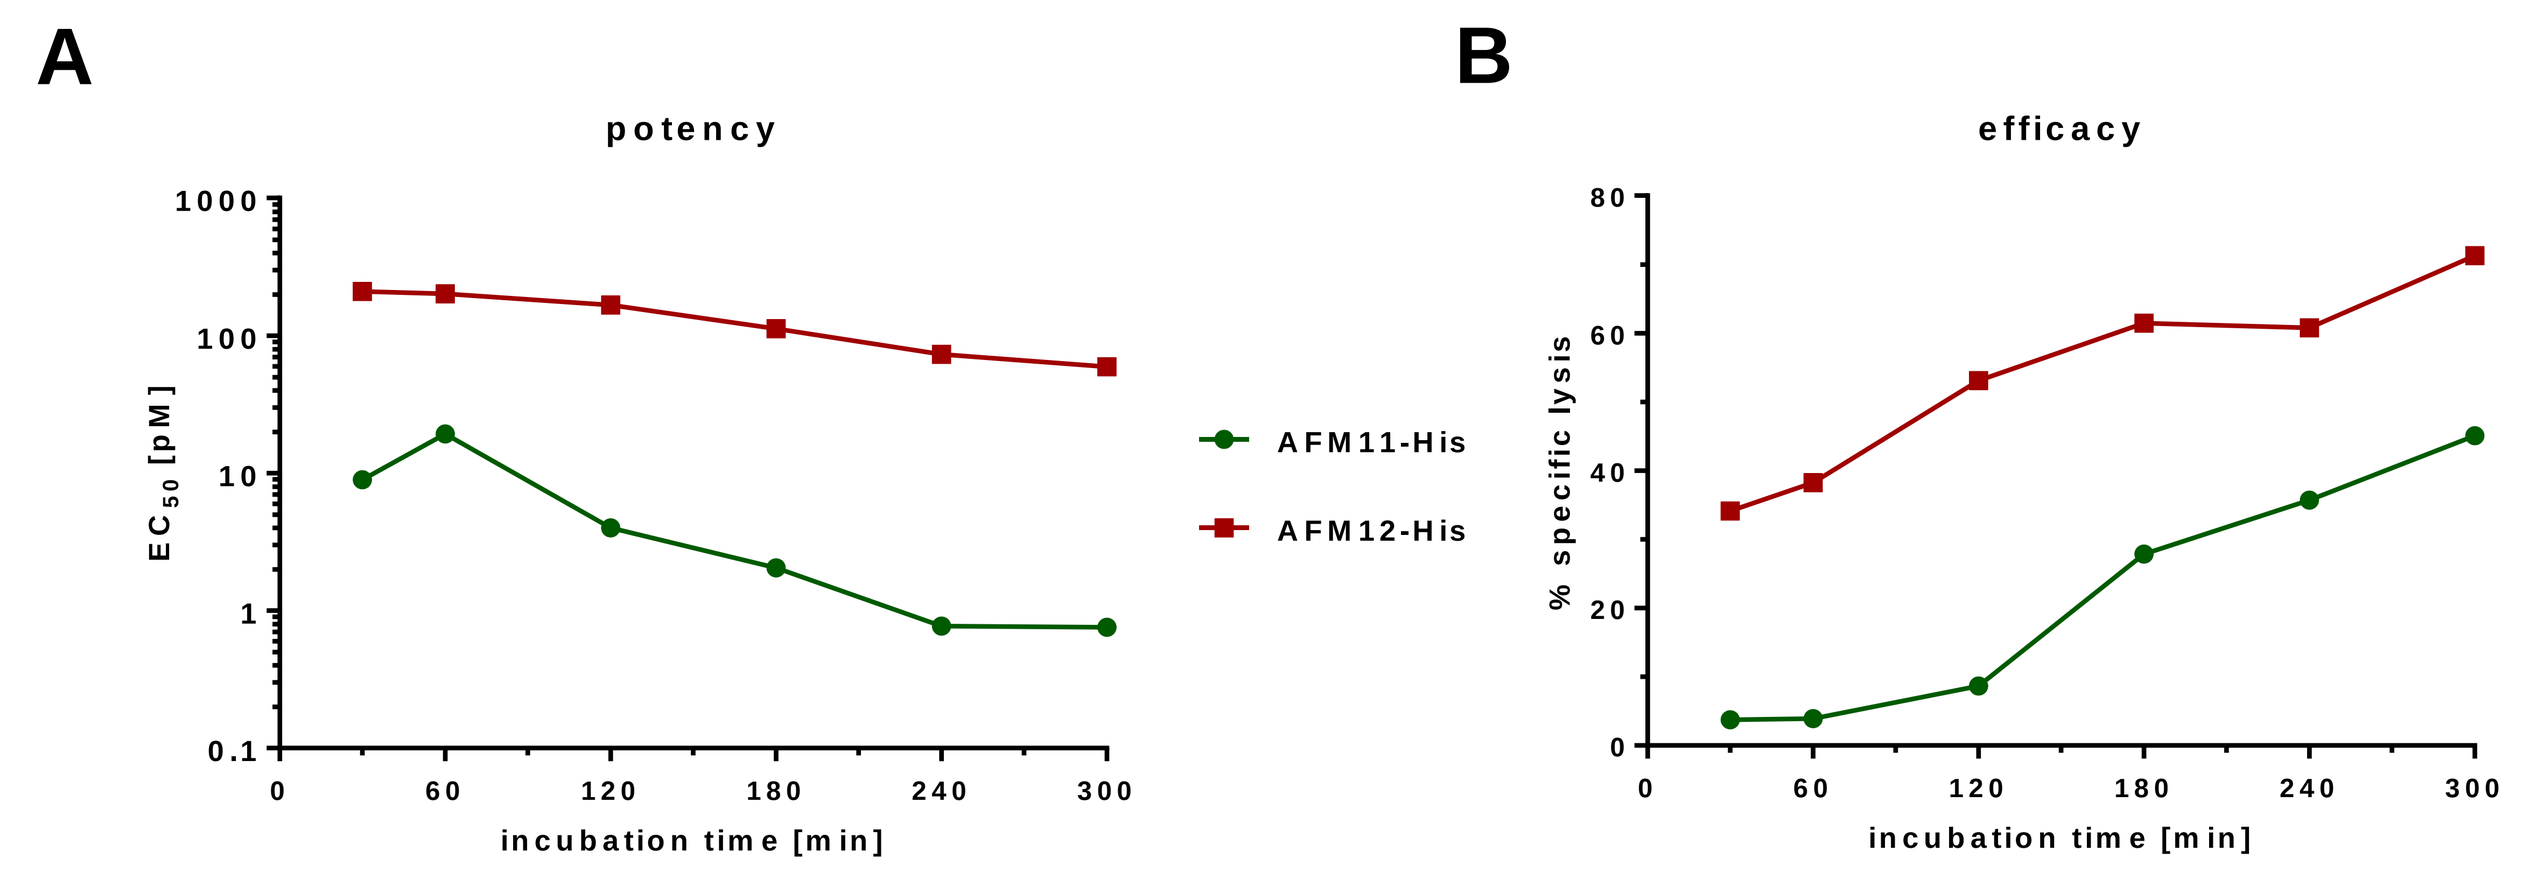
A
potency
EC_{50} [pM]
incubation time [min]
AFM11-His
AFM12-His
B
efficacy
% specific lysis
incubation time [min]

Kinetics of AFM11- and AFM12-mediated target cell lysis was assessed in a calcein-release assay on MEC-1 target cells and PBMC from a healthy volunteer as effector cells at an E:T ratio of 50:1. Dose-dependent target cells was measured for His-tagged AFM11 (AFM11-His) and AFM12 (AFM12-His) or without antibodies between 30 min and 5 hours incubation. EC_{50} values (potency, **A**) and percentage of TandAb-mediated lysis (efficacy, **B**) were determined at each time point and plotted.

- Faster on-set of AFM12-mediated lysis when compared to AFM11-induced cytotoxicity
- Higher efficacy for AFM12 and higher potency for AFM11 at each tested time point

AFM11- and AFM12-mediated cytotoxicity towards primary B-CLL cells

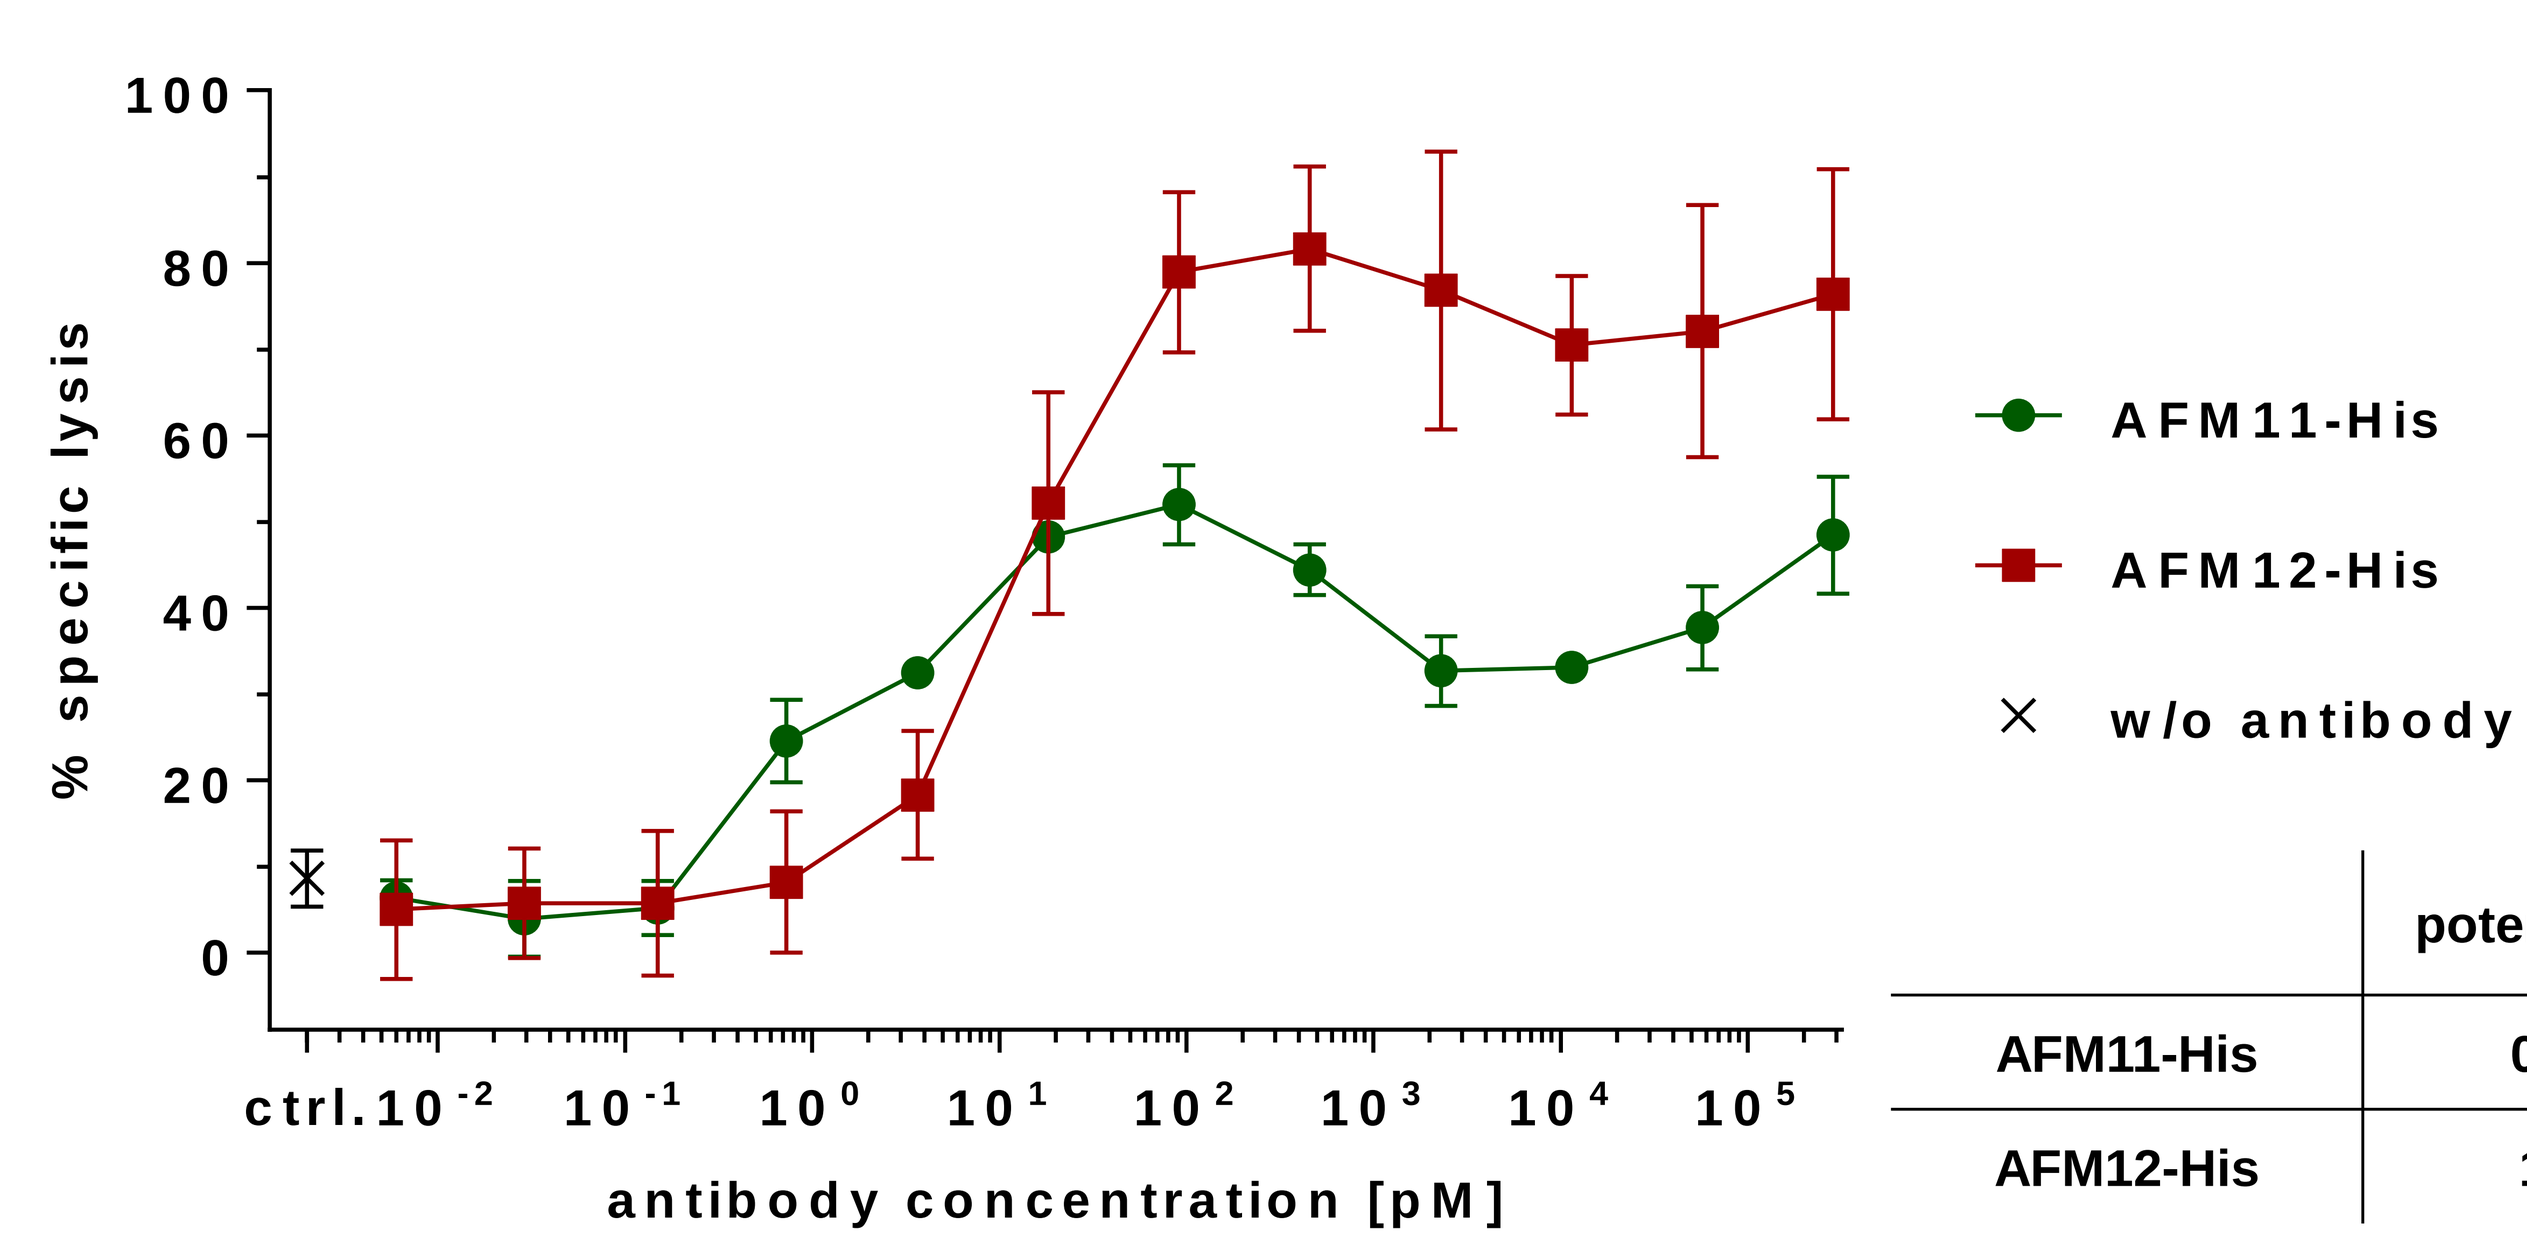
% specific lysis
antibody concentration [pM]
AFM11-His
AFM12-His
w/o antibody

Primary PBMC from a patient with B-CLL (>95% CD19⁺ cells) were used as target cells in a calcein-release assay with PBMC from a healthy volunteer as effector cells at an E:T ratio of 50:1. After 4 h incubation in the presence of increasing concentrations of AFM11-His and AFM12-His, fluorescent calcein released from lysed target cells was used to calculate specific lysis. EC_{50} values (potency) and percentage of TandAb-mediated target cell lysis (efficacy) were determined by non-linear regression/sigmoidal dose-response.

	potency / EC_{50}	efficacy / TandAb-mediated specific lysis
AFM11-His	0.8 pM	38%
AFM12-His	11 pM	70%

- Potency and efficacy of AFM11- and AFM12-mediated lysis of primary CLL cells is similar to CD19⁺ cell lines

Induction of proliferation and T- and NK-cell activation by AFM11 and AFM12 in PBMC cultures

- Strong, dose-dependent proliferation in 5 days PBMC cultures was induced by AFM11, but not by AFM12.
- AFM11 induces dose-dependent induction of activation markers CD25 and CD69 on T-cells, but also on NK-cells in 24 hours cultures of unfractionated PBMC.
- Consistent with the proliferation assay, AFM12 induced only strong NK-cell activation (CD69 expression), but no or only minimal activation of T-cells in 24 hours cultures.

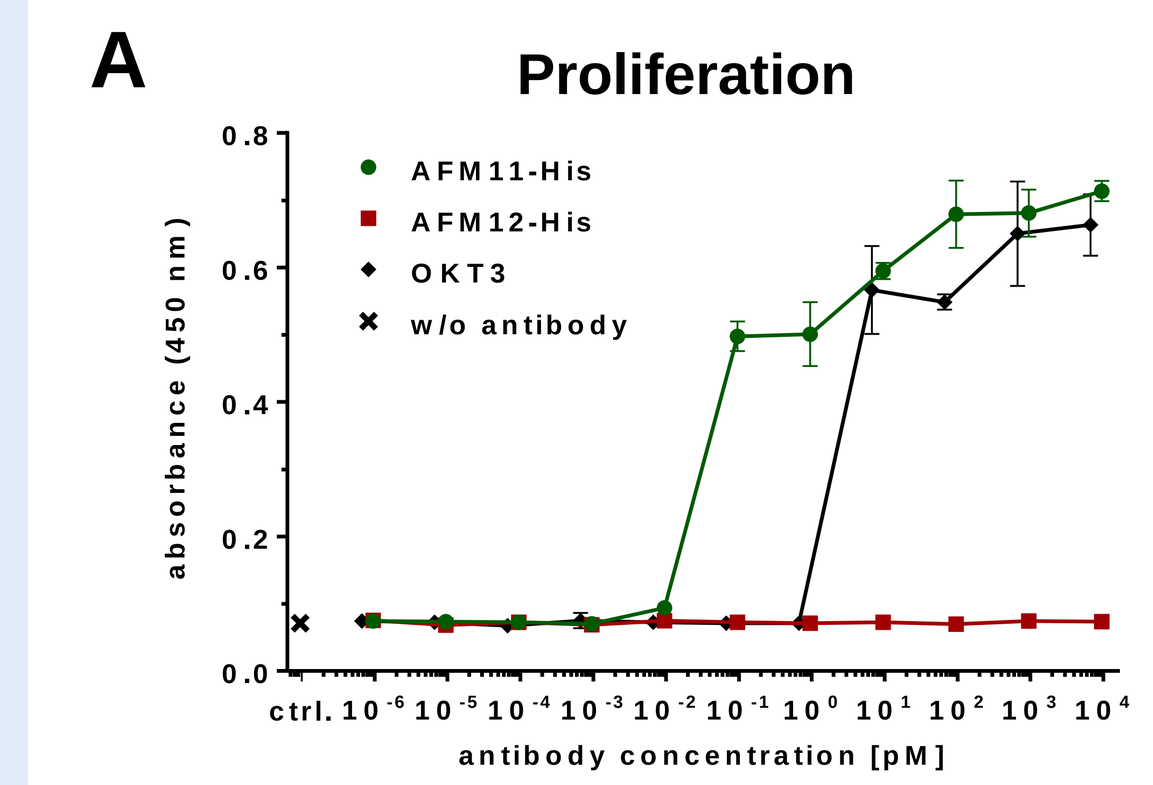
A
Proliferation
absorbance (450 nm)
antibody concentration [pM]
AFM11-His
AFM12-His
OKT3
w/o antibody

PBMC from healthy volunteers were cultured in the presence of increasing concentrations of AFM11, AFM12, and mAb anti-CD3 OKT3 for 4 days and then pulsed O/N with BrdU. Proliferation was quantified using a BrdU incorporation ELISA (**A**). T- and NK-cell activation was assessed in unfractionated PBMC cultured for 24 hours in the presence of AFM11, AFM12, and, as a control, with CD3/CD28 activator beads. Percentage of CD25⁺ and CD69⁺ T- and NK-cells was determined by flow cytometry (**B**).

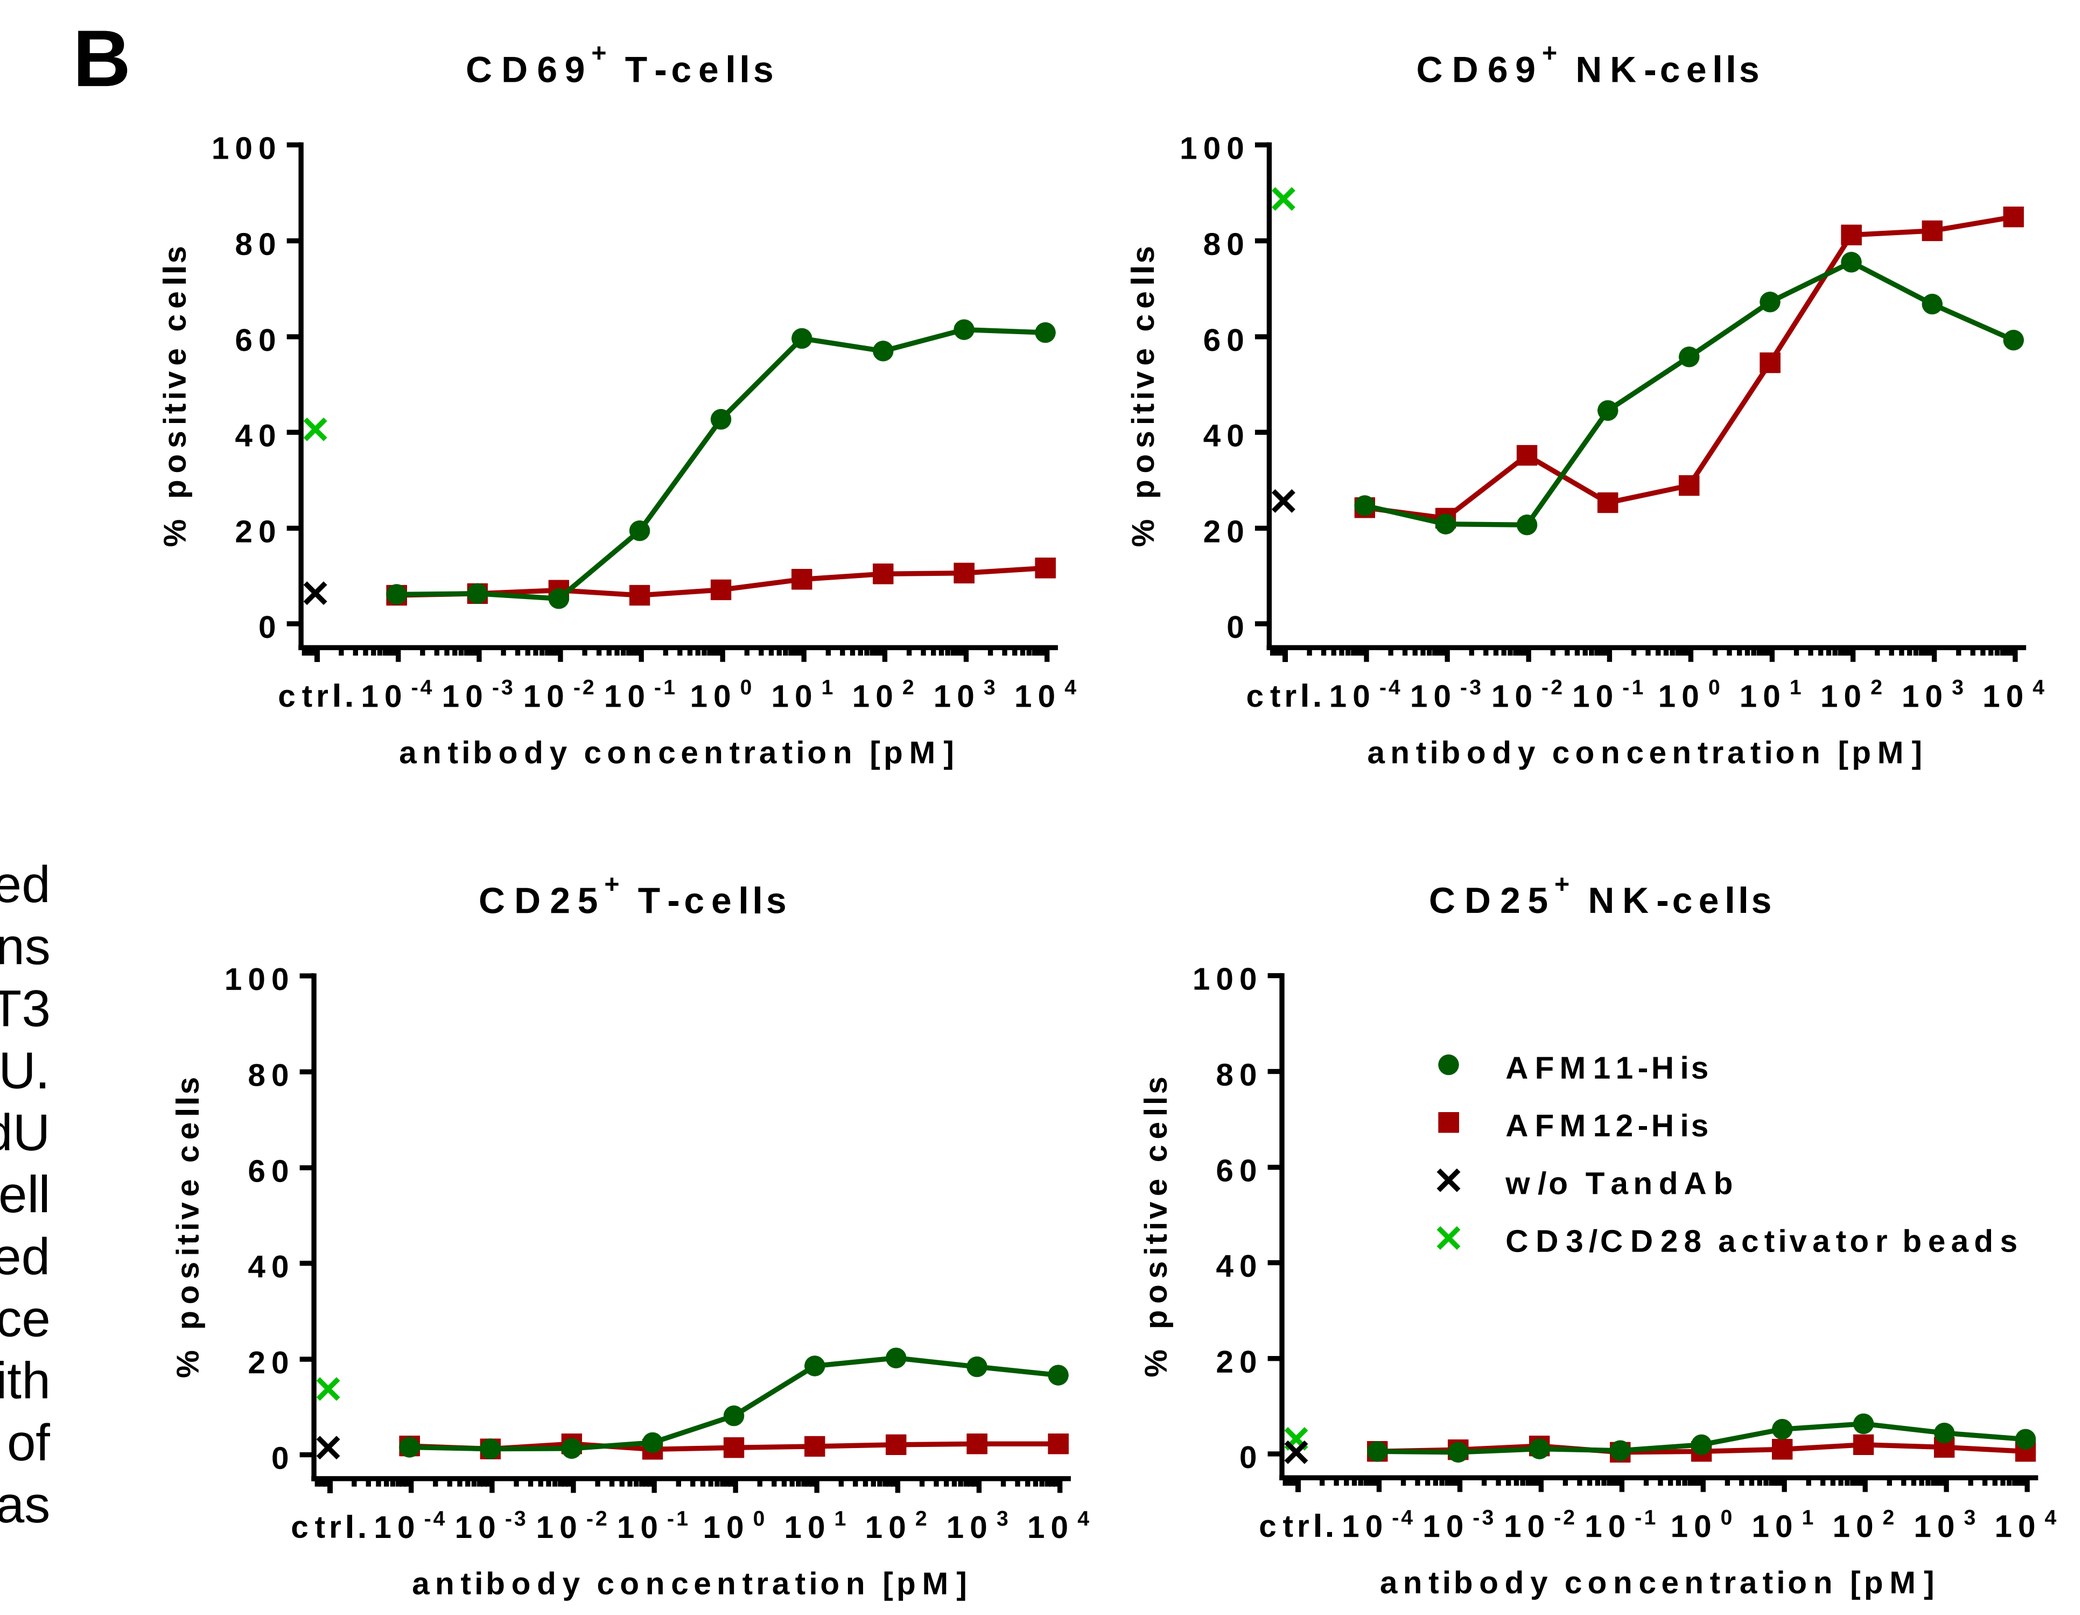
B
CD69+ T-cells
CD69+ NK-cells
CD25+ T-cells
CD25+ NK-cells
% positive cells
antibody concentration [pM]
AFM11-His
AFM12-His
w/o TandAb
CD3/CD28 activator beads

AFM11- and AFM12-mediated inhibition of tumor growth in an ALL PDX model

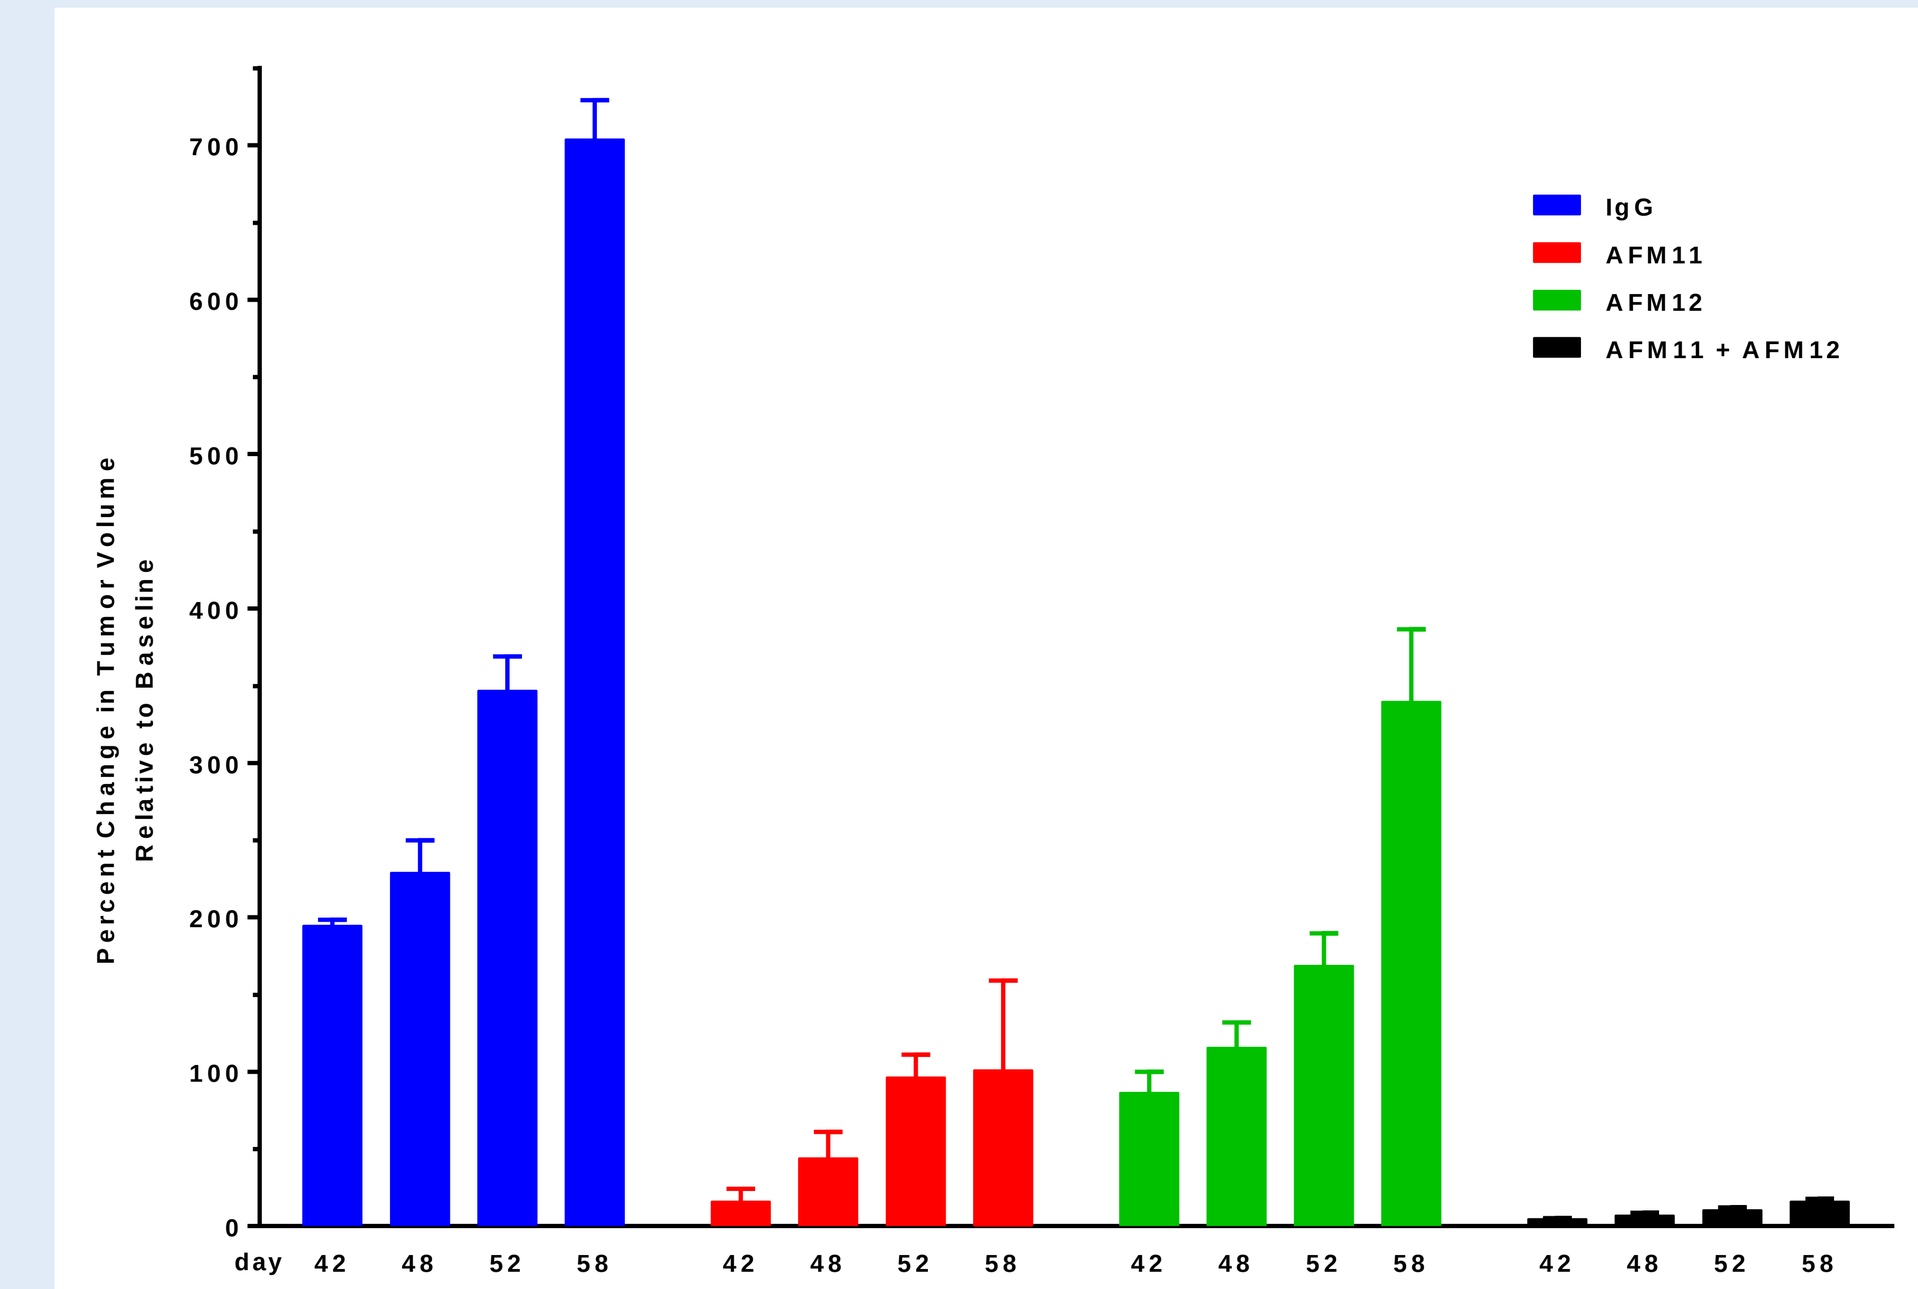
Percent Change in Tumor Volume Relative to Baseline
day 42 48 52 58
IgG
AFM11
AFM12
AFM11 + AFM12

- AFM11 and AFM12 reduced the tumor growth on day 58.
- The combination of AFM11 with AFM12 resulted in enhanced tumor growth control.

Infiltration of T- and/or NK-cells into tumors

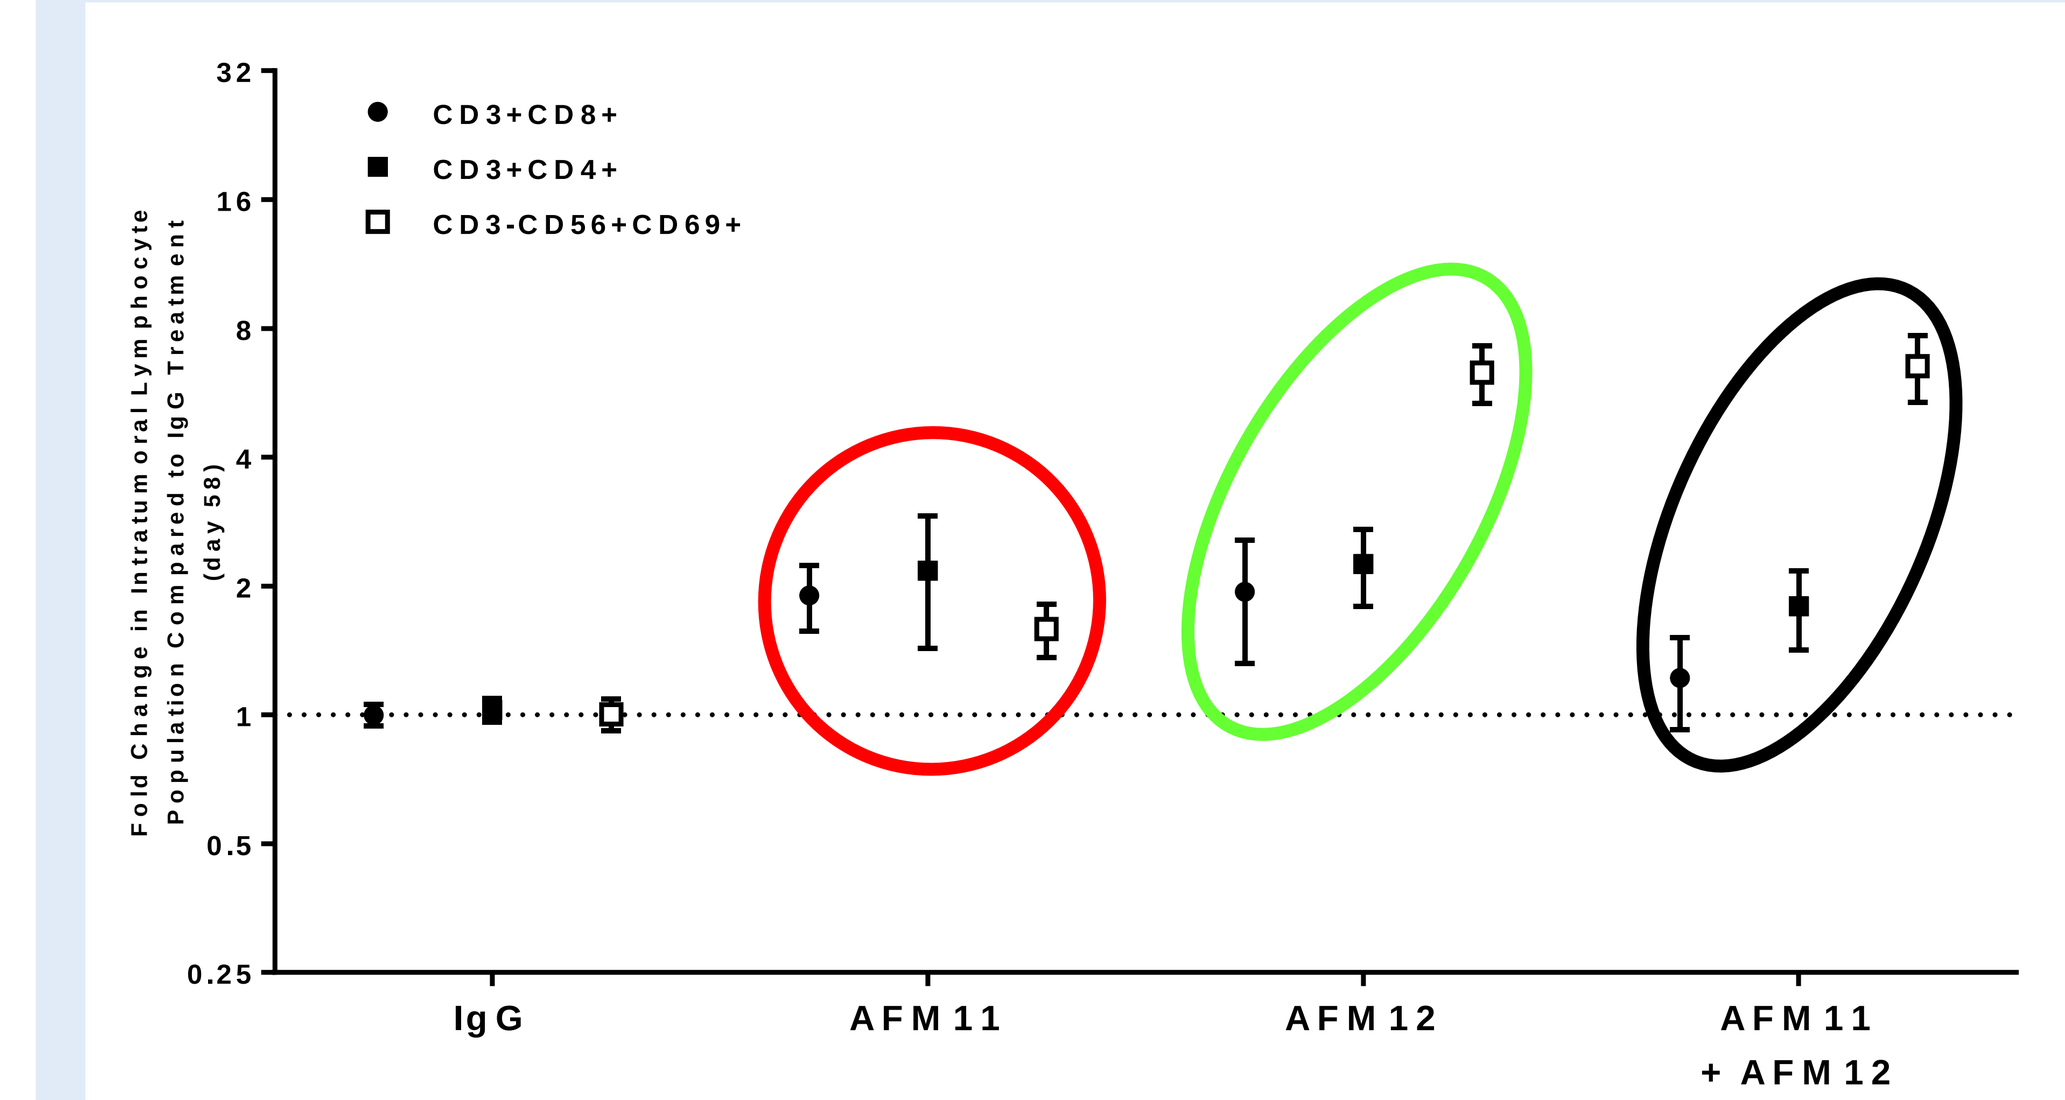
Fold Change in Intratumoral Lymphocyte Population Compared to IgG Treatment (day 58)
CD3+CD8+
CD3+CD4+
CD3-CD56+CD69+
IgG
AFM11
AFM12
AFM11 + AFM12

- Treatment with AFM11 alone induces T-cell infiltration into tumors, whereas AFM12 induces both NK- and T-cell infiltration.

Anti-PD-1 augments inhibition of tumor growth mediated by AFM11 and AFM12

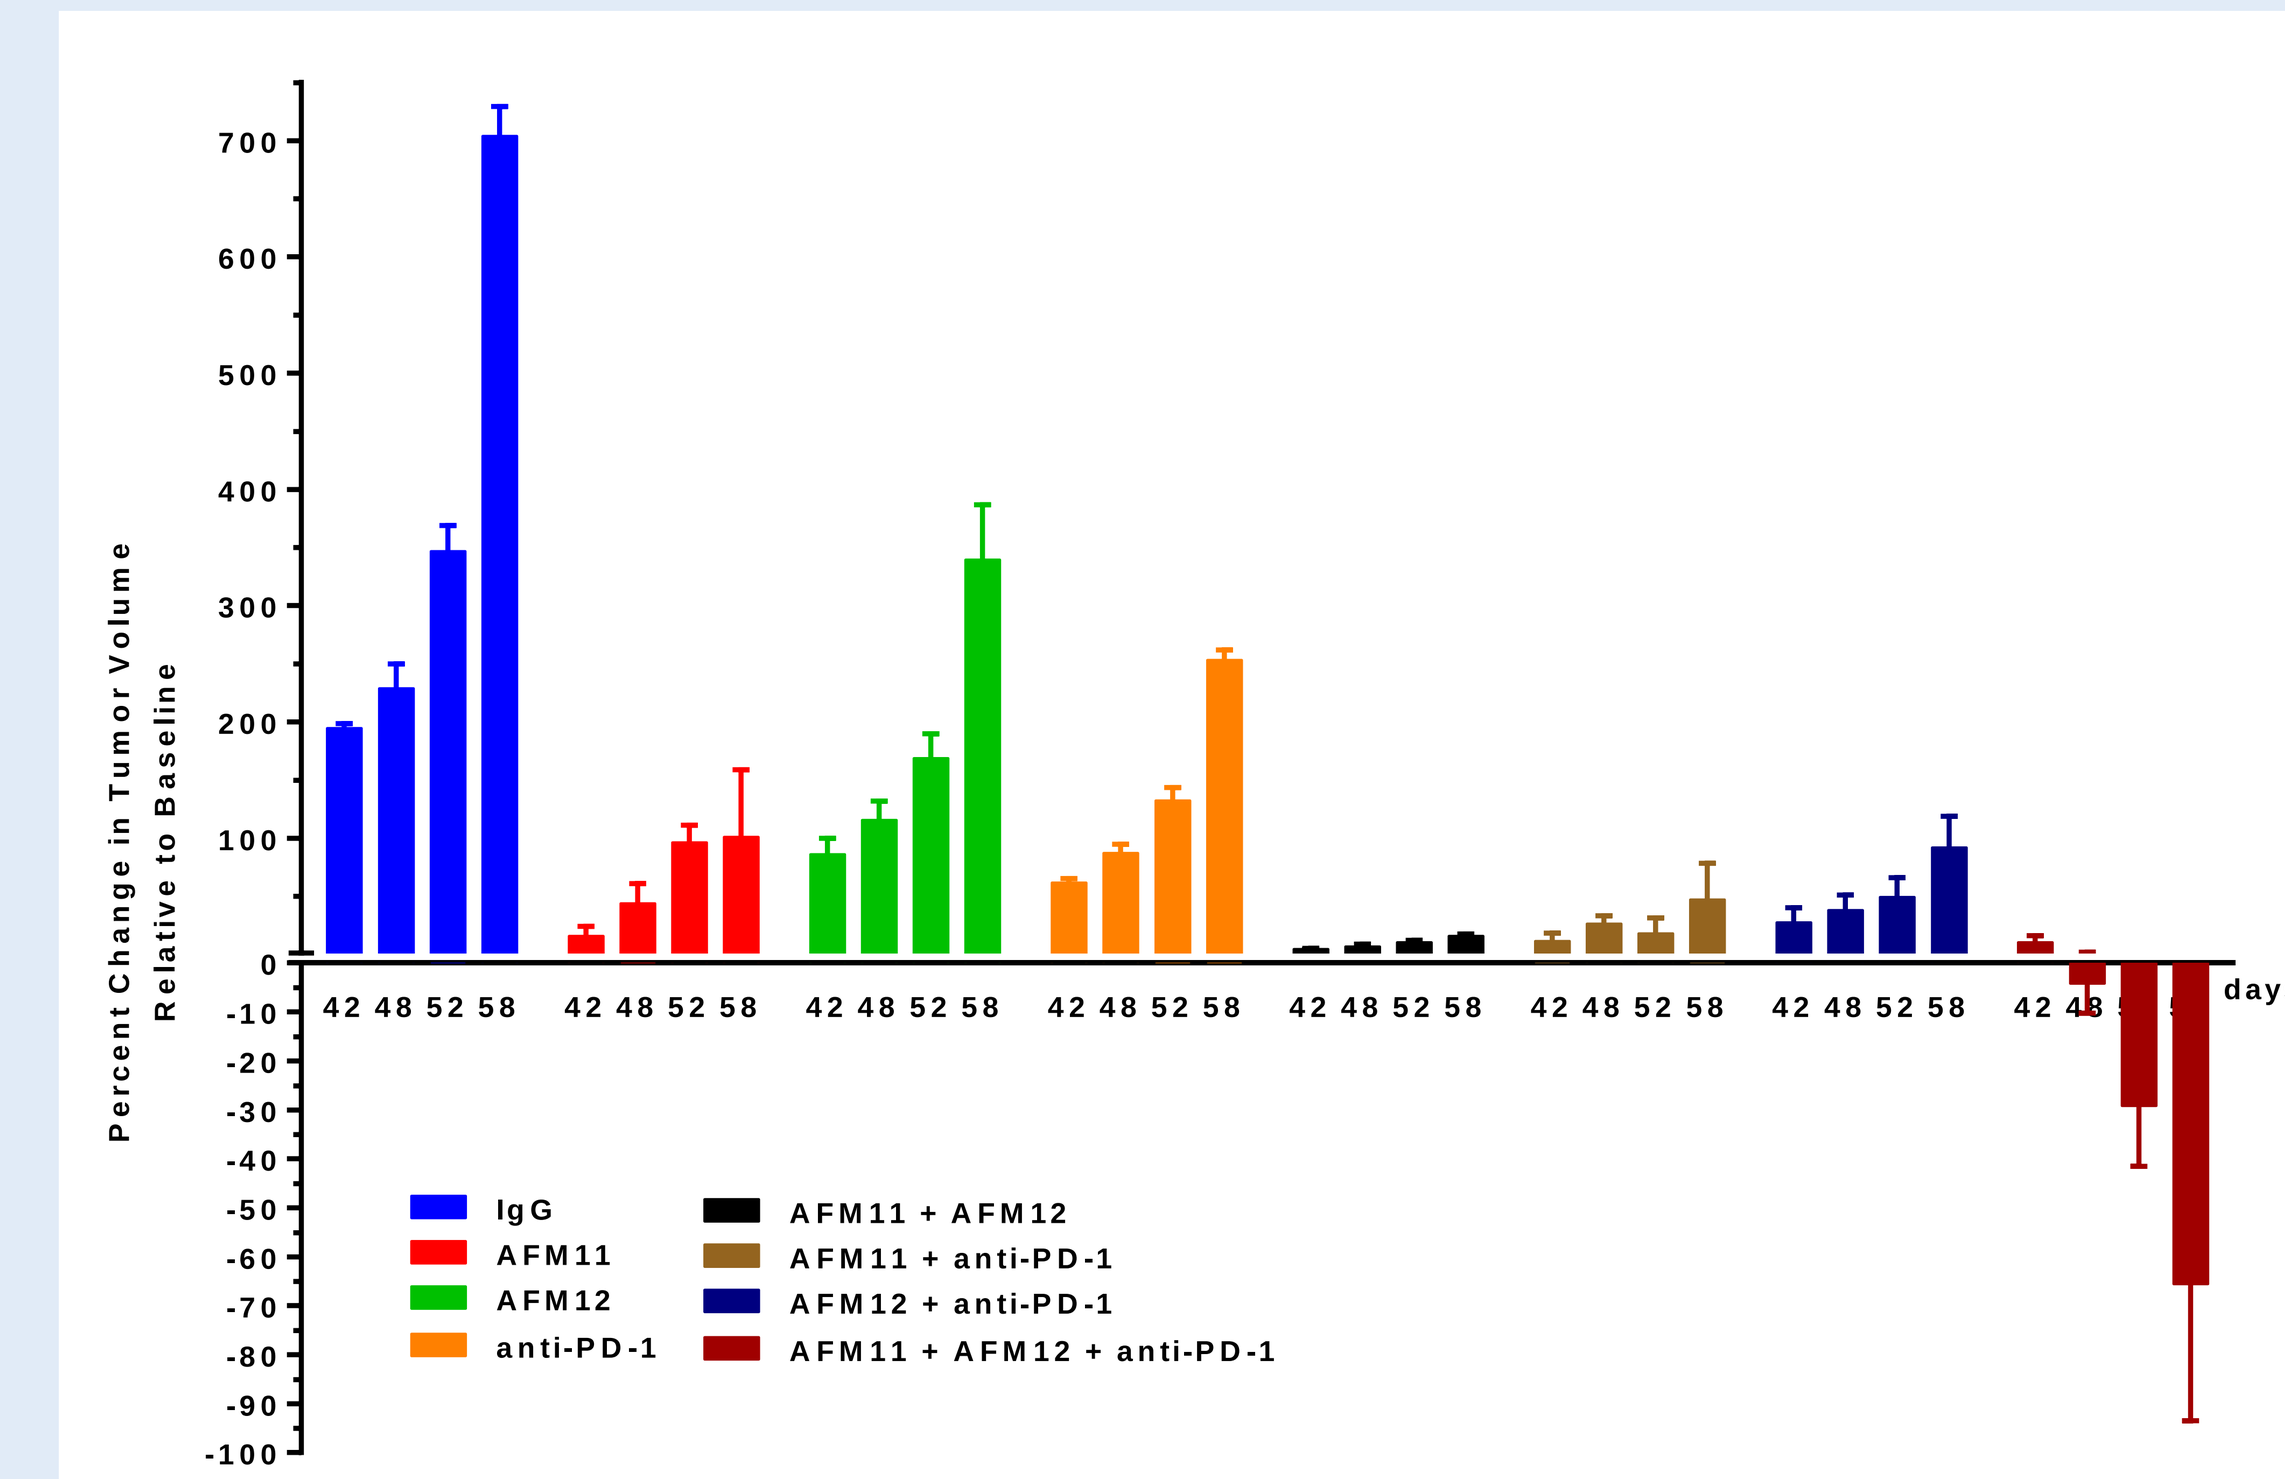
Percent Change in Tumor Volume Relative to Baseline
days
IgG
AFM11
AFM12
anti-PD-1
AFM11 + AFM12
AFM11 + anti-PD-1
AFM12 + anti-PD-1
AFM11 + AFM12 + anti-PD-1

- Anti-PD-1 inhibited tumor growth with similar efficacy as AFM12 in an ALL PDX model, whereas tumor growth control by AFM11 was slightly more efficacious.
- Combination of AFM11 or AFM12 with anti-PD-1 substantially augments tumor growth control in both combinations.
- A triple therapy with AFM11, AFM12, and anti-PD-1 not only prevented tumor growth but induced shrinkage of tumors.

Corresponding author

References: 1. Reusch, U. *et al*. A tetravalent bispecific TandAb (CD19/CD3), AFM11, efficiently recruits T cells for potent lysis of CD19⁺ tumor cells. Mabs 7(3): 584-604 (2015).
2. Reusch, U. *et al*. A novel tetravalent bispecific TandAb (CD30/CD16A) efficiently recruits NK cells for the lysis of CD30⁺ tumor cells. Mabs 6(3): 728–739 (2014).